Prediction of Human Effective Dose and Pharmacokinetics-Pharmacodynamics using Preclinical Data

Sunyoung Lim, Seokhyun Hong, Yunju Kang, Dongjin Hong, Seung Hyun Jung, Yu-Yon Kim, Taehun Song, Young Gil Ahn, Sang Hyun Lee

Hanmi Research Center, Hanmi Pharmaceutical Co. Ltd., Republic of Korea

ISSX JSSX HONOLULU, HAWAII 2024

Introduction

HM97662 is a potent dual inhibitor of histone methyltransferase enhancer of zeste homolog 1 (EZH1) and EZH2, which are subunits of polycomb repressive complexes 2 (PRC2). EZH1/2 have a function in tumorigenesis as oncogenes, therefore, targeting PRC2 for cancer therapies has been developed in clinics. EZH1 is involved in cellular metabolism, kinase regulation, and cytokine-mediated signaling. Also, EZH1 also plays a compensatory role in maintaining tri-metylation of H3K27 and directly binds to chromatin, modulating is condendation. EZH2 is often overexpressed in diverse solid tumors and hematological malignancies, and associated with metastatic progression, suggesting an oncogenic function. Dual inhibition of EZH1 and EZH2 could give more effective than EZH2 inhibition alone in blocking PRC2 function as an anti-cancer therapy. The antitumor activities of HM97662 were evaluated in HT-1376 bladder cancer (ARID1A mut/ SMARCD2 mut/ KDM6A mut) cell subcutaneous xenograft models as target-associated cancers. Therefore, we aim to confirm translational PK/PD ralationship to predict human PK and support clinical trial design.

Objectives

The overall objective of this study is to assess the predictability of preclinical data regarding human PK, and define the PK/PD relationship in HT-1376 xenograft mouse model to determination of effective doses.

Figure 1. Strategy of Human PK & Effective dose prediction

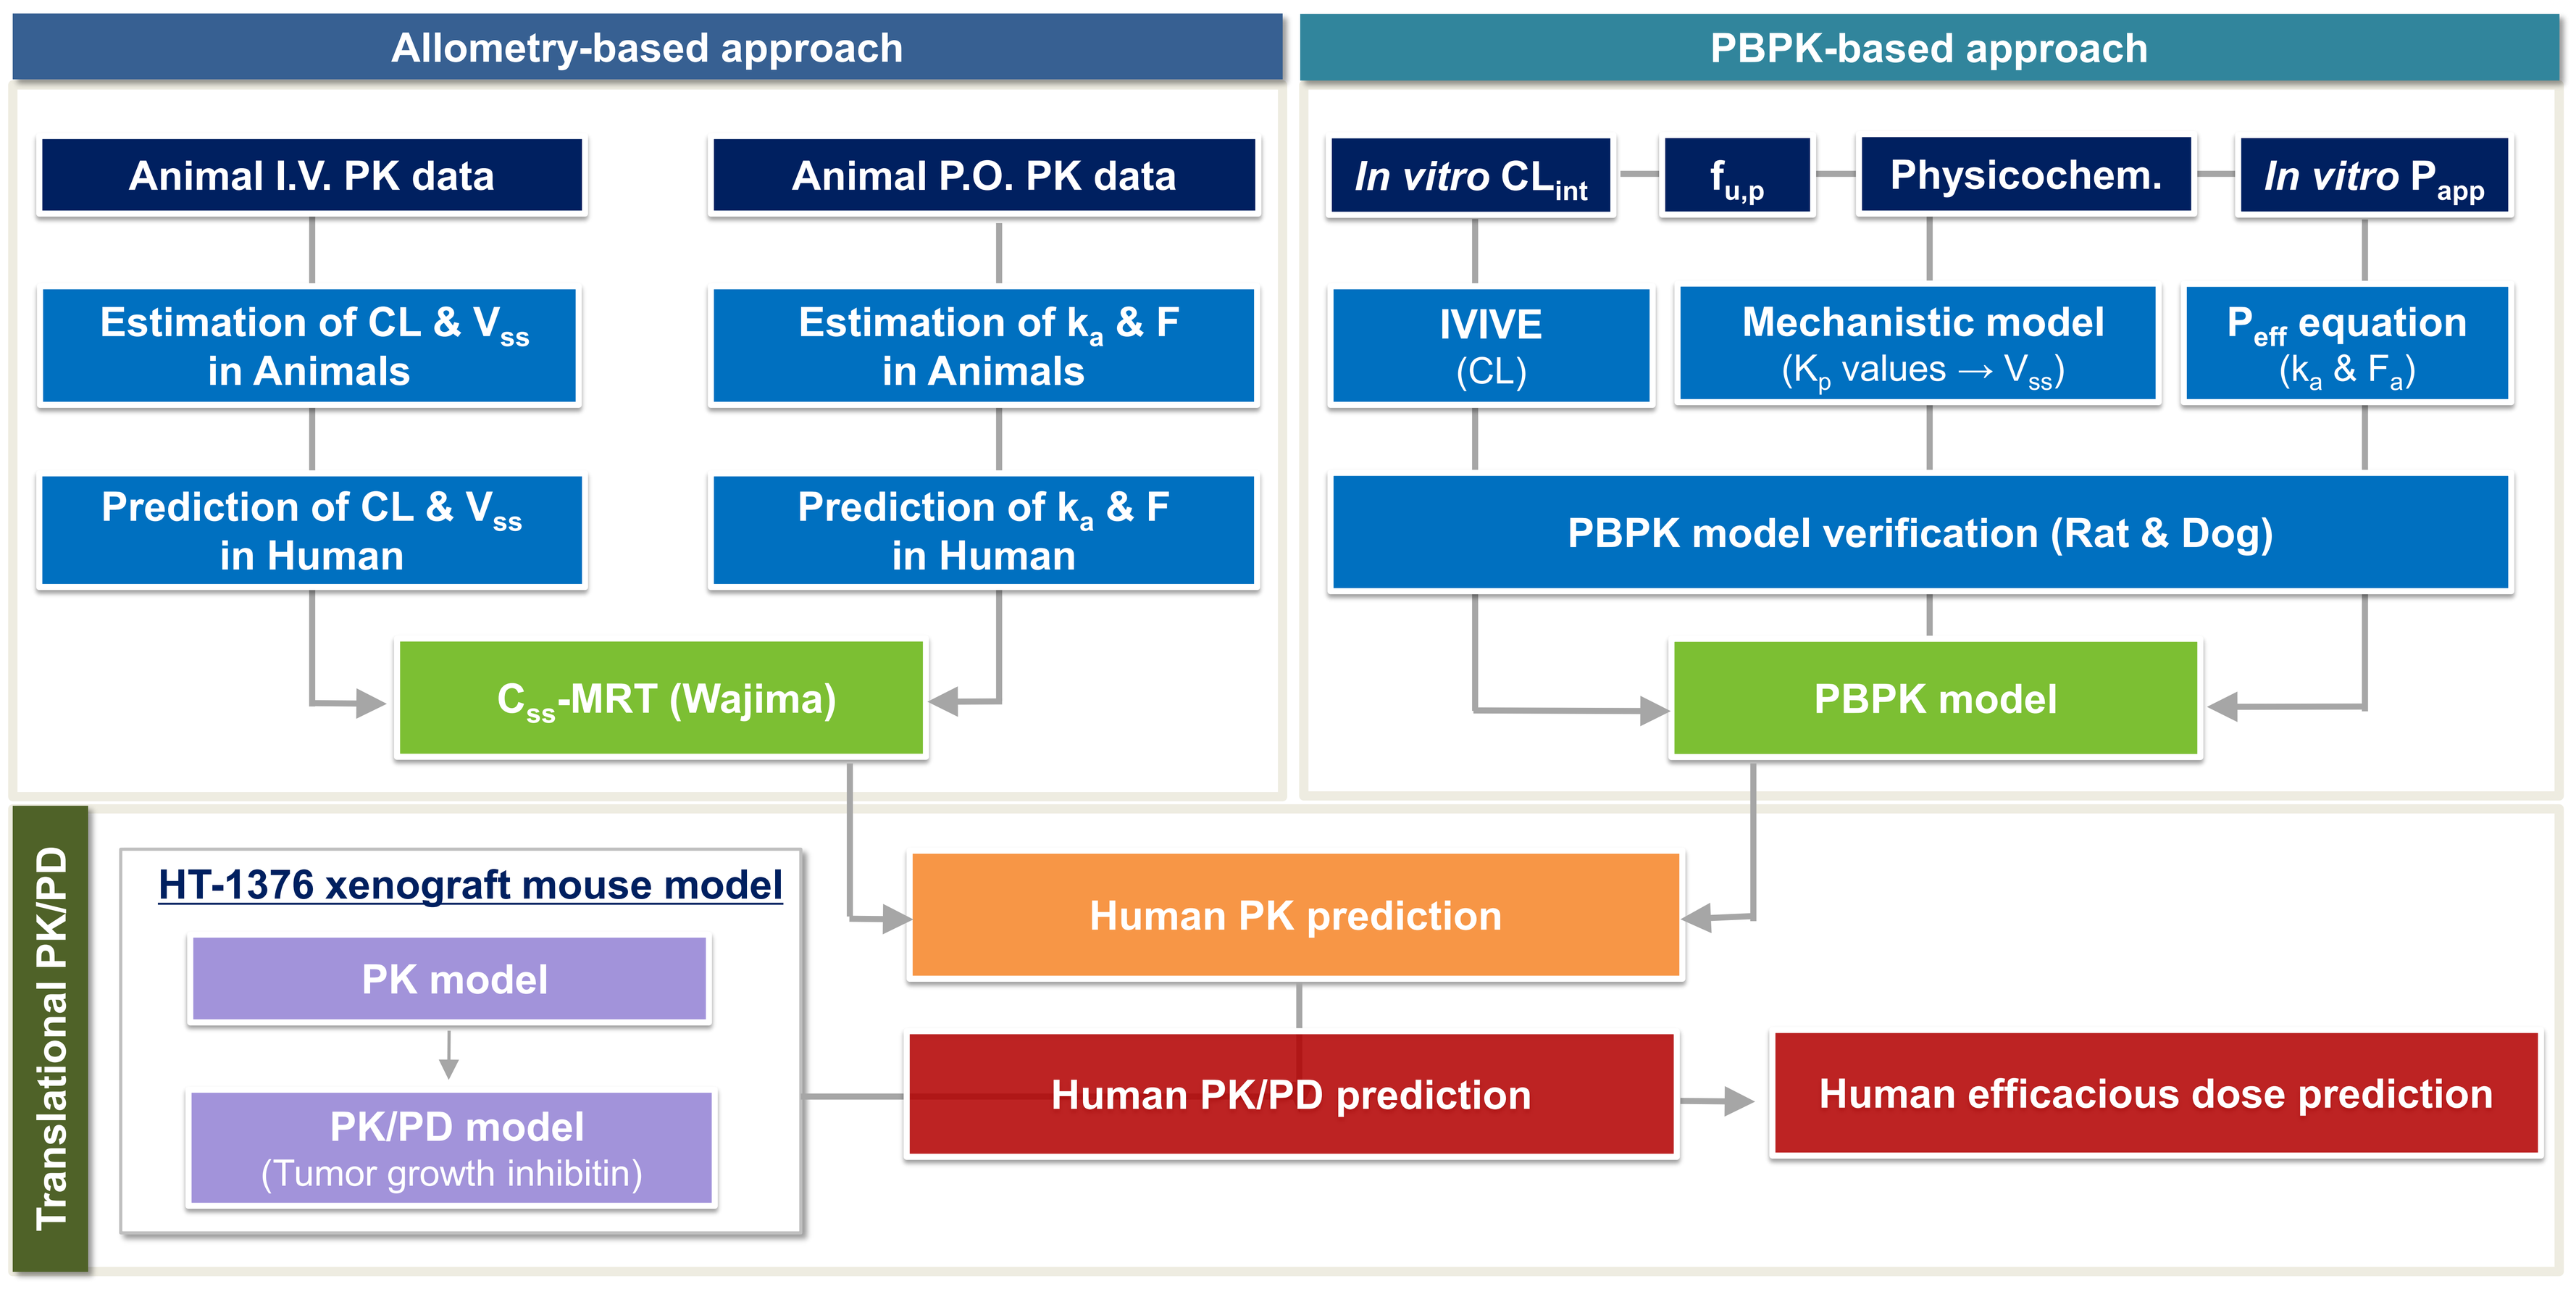
Allometry-based approach
PBPK-based approach
Animal I.V. PK data
Animal P.O. PK data
Estimation of CL & V_{ss} in Animals
Estimation of k_a & F in Animals
Prediction of CL & V_{ss} in Human
Prediction of k_a & F in Human
C_{ss}-MRT (Wajima)
In vitro CL_{int}
$f_{u,p}$
Physicochem.
In vitro P_{app}
IVIVE (CL)
Mechanistic model (K_p values → V_{ss})
P_{eff} equation (k_a & F_a)
PBPK model verification (Rat & Dog)
PBPK model
Translational PK/PD
HT-1376 xenograft mouse model
PK model
PK/PD model (Tumor growth inhibitin)
Human PK prediction
Human PK/PD prediction
Human efficacious dose prediction

Methods

- ***In vivo* PK studies**
 - Animal PK studies : Male CD-1 (ICR) mice, SD rats and beagle dogs were administered single doses of HM97662 via intravenous (I.V.) or oral (P.O.) route. These preclinical data were used to predict the human PK of HM97662.
- ***In vitro* PK studies**
 - Permeability in MDCKII cells : The apparent permeability coefficient (P_{app}) was determined in MDCKII cells monolayers. This value was utilized in bottom-up prediction of human oral absorption rate constant (k_a) and fraction absorbed (F_a).
 - Metabolic stability in hepatocytes : The metabolic stability was investigated using mouse, rat, dog and human hepatocytes (0.7-1.0 $\times 10^6$ cells/mL) for 90 min incubation. The calculated *in vitro* intrinsic clearance (CL_{int}) values were used to predict the human clearance by *in vitro-in vivo* extrapolation (IVIVE) approach.
 - Plasma protein binding (PPB) : The unbound fraction of HM97662 in mouse, rat, dog and human plasma were determined using rapid equilibrium dialysis (RED) with LC-MS/MS. The unbound fractions in plasma ($f_{u,p}$) were used in human PK prediction and correction of effective concentration between mouse and human.
- ***In vivo* PK/PD study**
 - Female BALB/c nude mice were implanted with human bladder cancer cells (HT-1376). In the PK study, HM97662 was orally administered to HT-1376 tumor bearing mice once daily at doses of 5, 10, 20 and 40 mg/kg during 14 days. The blood samples were collected at the day 1 and day 14. In the efficacy study, HM97662 was treated once daily via oral gavage for 28 consecutive days at doses of 5, 10, 20 and 40 mg/kg.

Modeling & Simulation Process

- **Human PK prediction**
 - Allometry-based approach : The human CL and V_{ss} were predicted using *in vivo* animal PK data by fraction unbound intercept correction method (FCIM) and Øie-Tozer method, respectively.[1, 2] The oral absorption rate constant (k_a) and bioavailability (F) in human was set to average of animal k_a (calculated by method of residuals) and F. The human PK profile was predicted using C_{ss}-MRT (Wajima) method.[3]
 - PBPK-based approach : The bottom-up approaches were applied in PBPK-based human PK prediction. The human CL was predicted using hepatocyte CL_{int} by $f_{u,liver}$ correction method [4], and V_{ss} was predicted using physicochemical properties (logP and pKa) and $f_{u,p}$ by mechanistic distribution model [2] with K_p scalar. These prediction approaches were verified by animal CL and V_{ss} prediction. The oral k_a and fraction absorbed (F_a) were calculated using *in vitro* P_{app} in MDCKII cells.[5, 6]
- **PK/PD modeling in HT-1376 xenograft model**
 - The PK/PD modeling was conducted in a sequential manner. The two compartment PK model with saturable absorption rate and non-linear bioavailability was used to describe the PK profiles in HT-1376 xenograft mouse modell. The signal distribution model (SDM)[7] with transit compartment was used for reflecting delayed anti-tumor effect and combined exponential and linear tumor growth model [8] was utilized to describe tumor growth in vehicle control group.
- **Translational human PK/PD model & human efficacious dose prediction**
 - Translational human PK/PD models were developed by combining each predicted human PK model (Allometry-based or PBPK-based model) and PD model for HT-1376 xenograft mouse with correction of difference in PPB between human and mouse (KC_{50} corrected). Using these translational human PK/PD models, the optimal human efficacious dose was determined to dose required to achieve ≥ 90% tumor growth inhibition (TGI).[9]

Figure 2. PK/PD model structure in HT-1376 xenograft mouse model

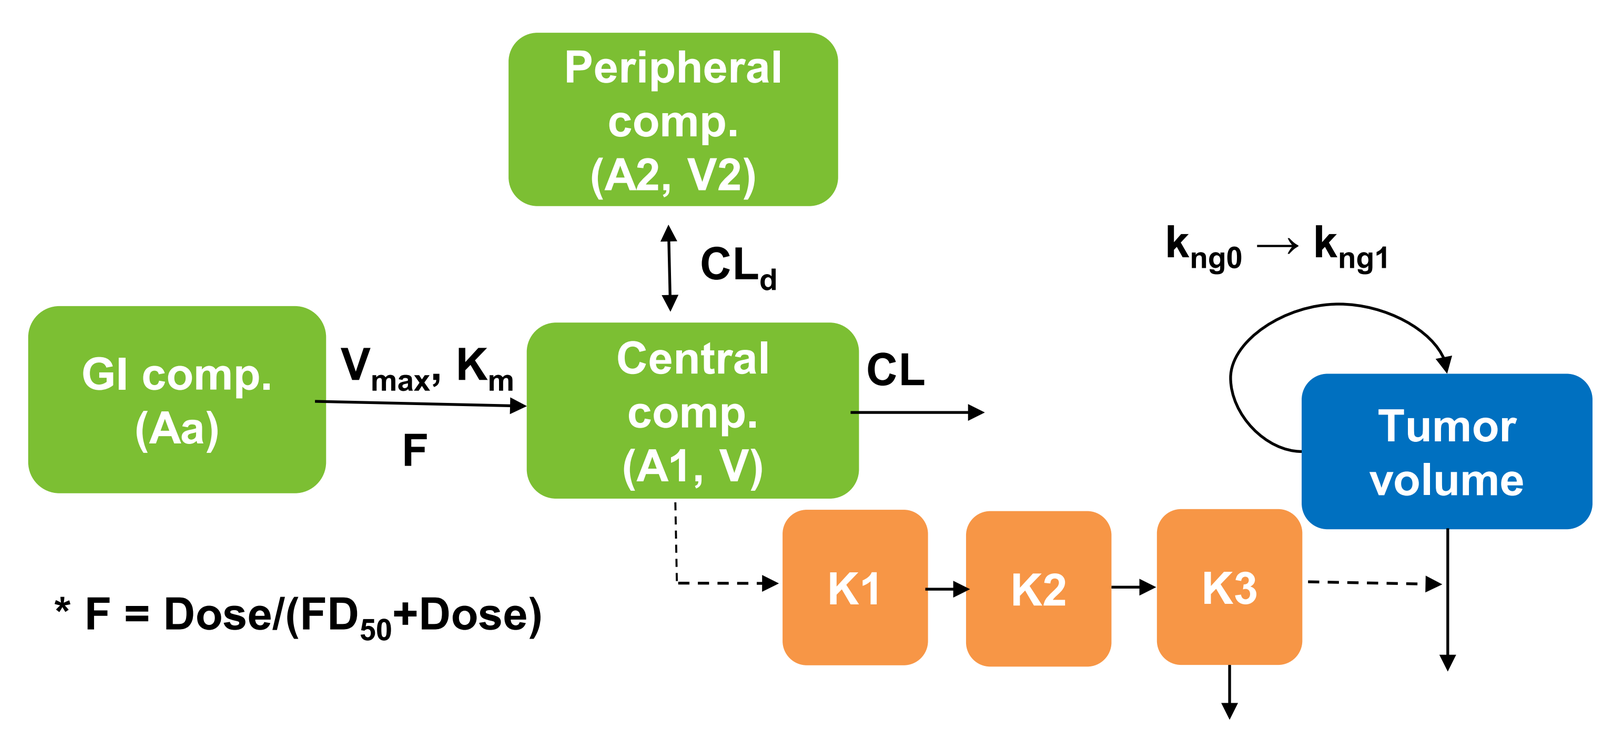
Peripheral comp. (A2, V2)
CL_d
GI comp. (Aa)
V_{max}, K_m
F
Central comp. (A1, V)
CL
$k_{ng0} \rightarrow k_{ng1}$
Tumor volume
K1
K2
K3

* F = Dose/(FD_{50}+Dose)

$$\frac{dK1}{dt} = \left(\frac{K_{max} \cdot C}{KC_{50} + C} - K1\right)/\tau \qquad \frac{dK2}{dt} = (K1 - K2)/\tau$$

$$\frac{dK3}{dt} = (K2 - K3)/\tau \qquad \frac{dTV}{dt} = \frac{k_{ng0} \cdot TV}{\left(1 + \left(\frac{k_{ng0}}{k_{ng1}} \cdot TV\right)^{20}\right)^{1/20}} - K3 \cdot TV$$

- All model analysis and simulations were performed using Phoenix WinNonlin® 8.1 (Certara, USA) and Berkeley Madonna® 10.6.1 (Berkeley Madonna, USA).

Results

Figure 3. Approaches for Human PK prediction

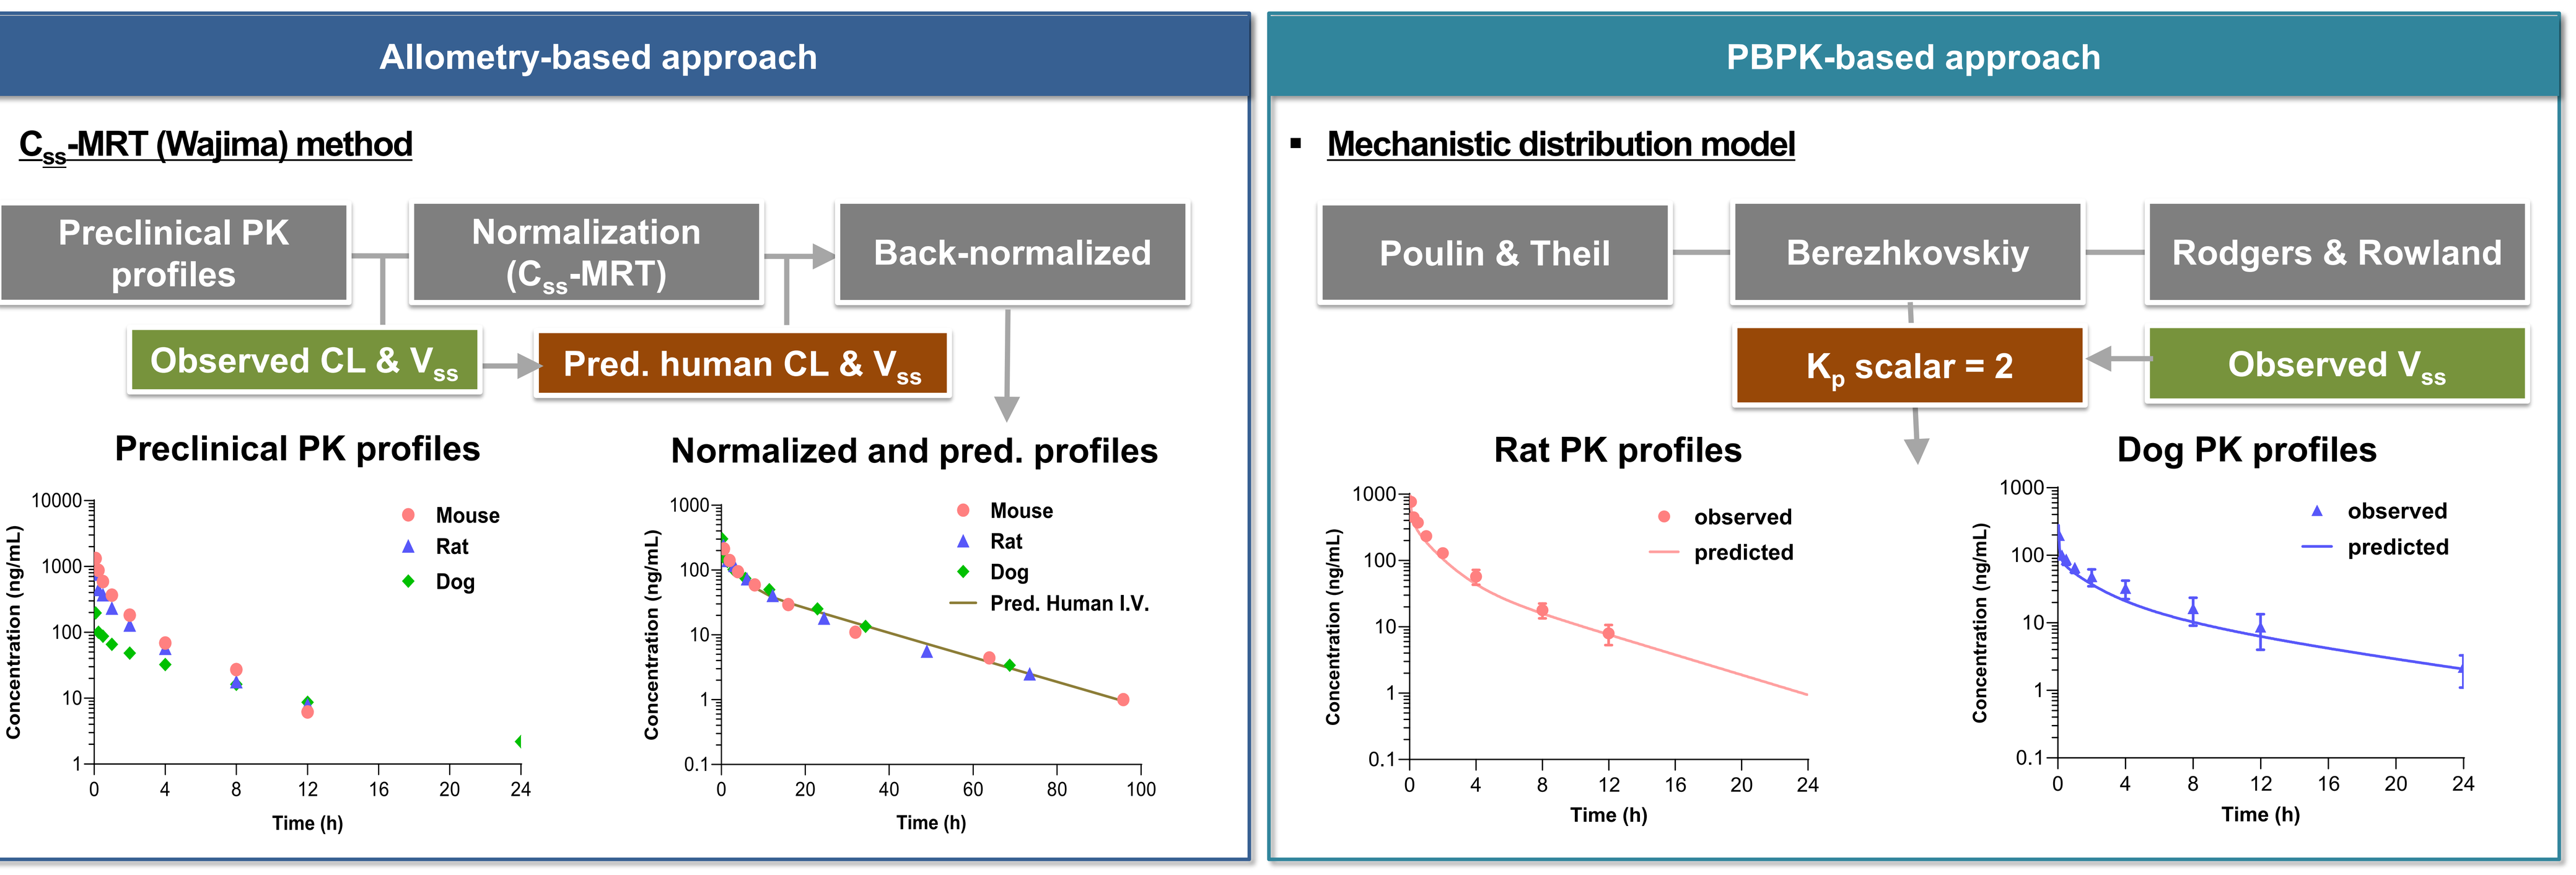
Allometry-based approach
C_{ss}-MRT (Wajima) method
Preclinical PK profiles
Normalization (C_{ss}-MRT)
Back-normalized
Observed CL & V_{ss}
Pred. human CL & V_{ss}
Preclinical PK profiles
Normalized and pred. profiles
Mouse
Rat
Dog
Pred. Human I.V.
Concentration (ng/mL)
Time (h)
PBPK-based approach
Mechanistic distribution model
Poulin & Theil
Berezhkovskiy
Rodgers & Rowland
K_p scalar = 2
Observed V_{ss}
Rat PK profiles
Dog PK profiles
observed
predicted

1. Human PK prediction

Predicted		Allometry		PBPK	
Predicted Primary PK parameters	CL (L/h/kg)	0.52		0.70	
	V_{ss} (L/kg)	9.13		6.42	
	Absorption rate constant ; k_a (1/h)	1.18		1.17	
	BA	0.55 (F)		0.95 (F_a)	
Predicted of PK parameters (50 mg, oral)	Day	C1D1	C1D8	C1D1	C1D8
	AUC_τ (ng·h/mL)	576.6	753.5	354.8	399.1
	C_{max} (ng/mL)	62.4	74.2	67.4	71.3
	Half-life (h)	13.0	-	8.6	-
Observed		PK parameters (50 mg, oral)			
		C1D1		C1D8	
AUC_τ (ng·h/mL)		358.1 ± 192.4		417.0 ± 251.8	
C_{max} (ng/mL)		60.9 ± 24.3		71.6 ± 49.3	
Half-life (h)		11.0 ± 3.4		-	

Figure 4. Observed Human PK profiles (HM-EZHI-101 Cohort#1 50 mg, oral, QD)

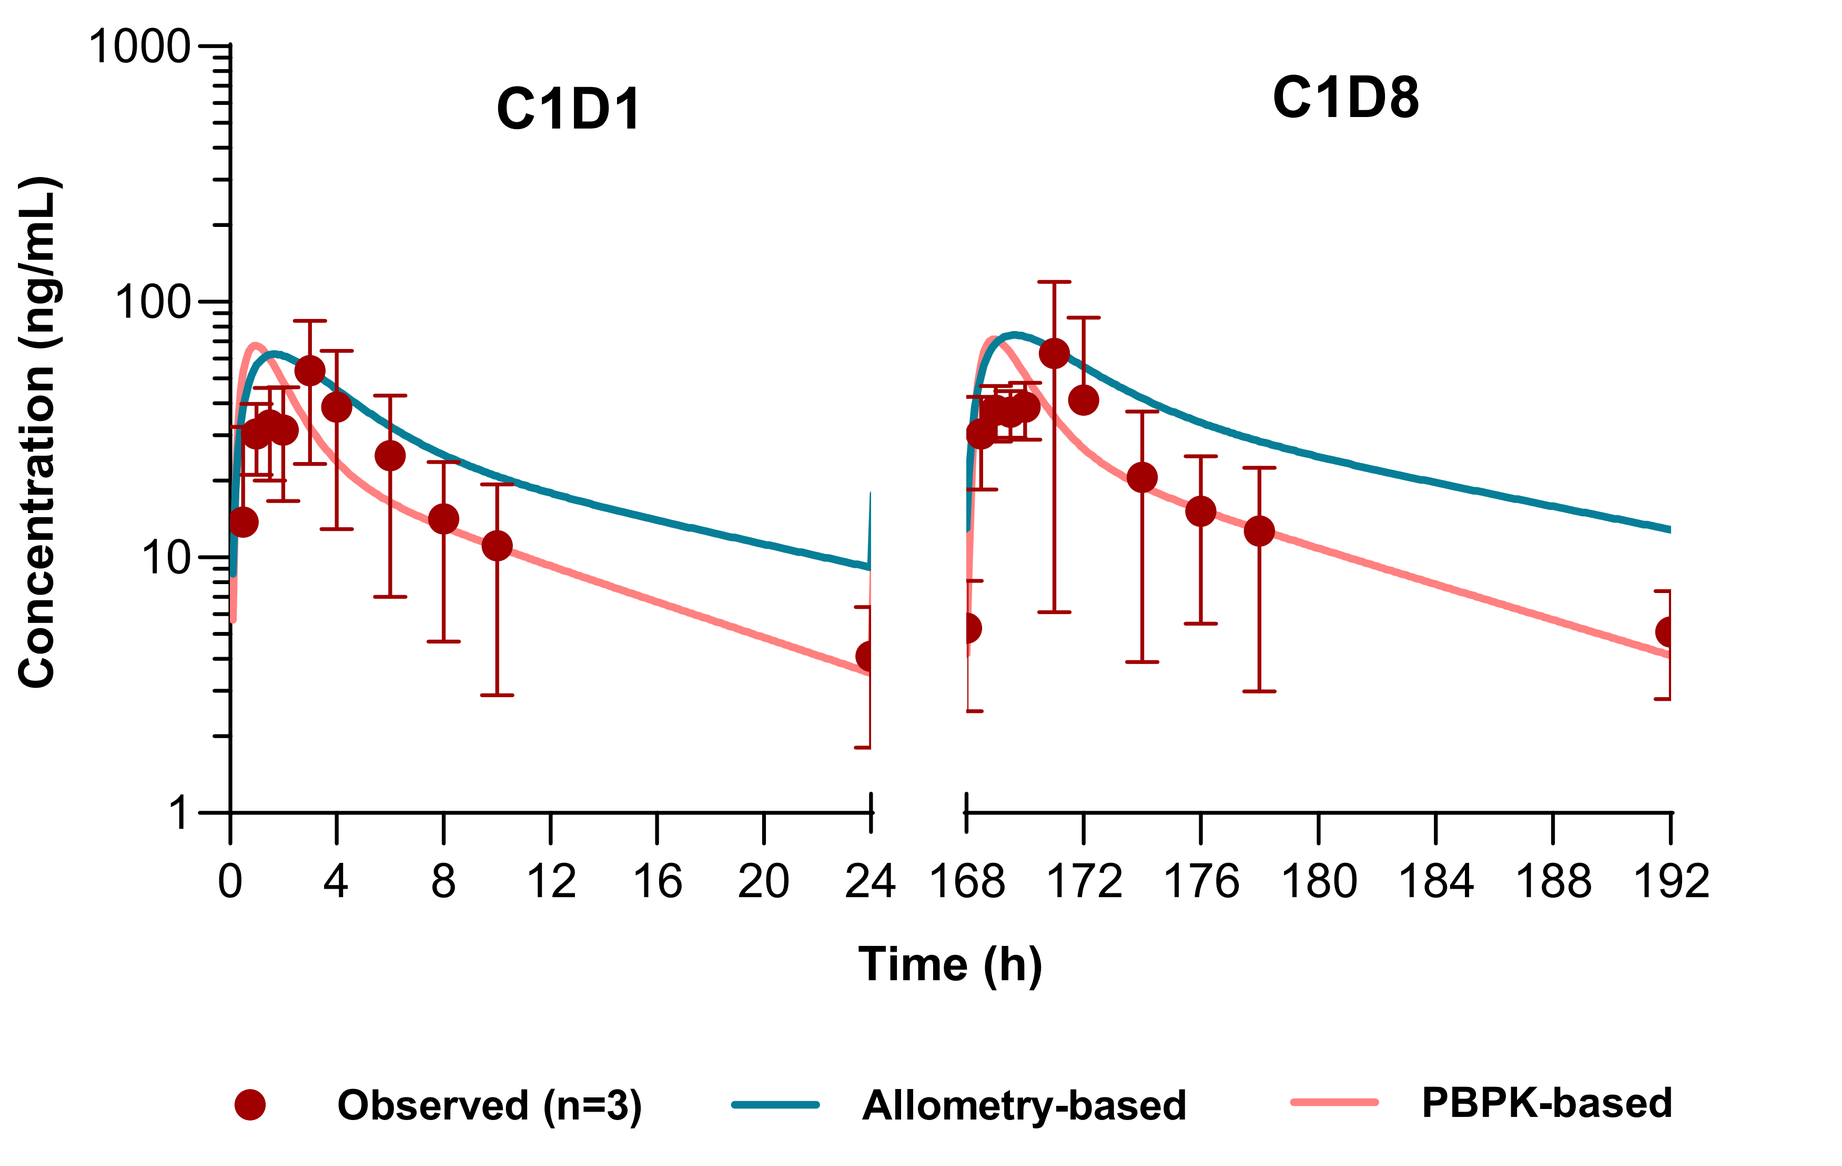
C1D1
C1D8
Concentration (ng/mL)
Time (h)
Observed (n=3)
Allometry-based
PBPK-based

- Allometric scaling of *in vivo* CL and V_{ss} data obtained from mouse, rat and dog PK studies showed a reasonable extrapolation of both parameters to human. Also, applying PBPK modeling using bottom-up method was subsequently used to predict the human PK profile. The HM97662 exposure was measured at C1D1 and C1D8 in clinical Cohort#1 treated with 50 mg QD and was compared with the exposure predicted from the human PK parameters obtained using the allometric and PBPK approaches. The predicted C_{max} and AUC_T values of HM97662 were similar (within 1.9-fold) between allometry and PBPK modeling, and all predicted values were within 1.8-fold of observed results, suggesting that these models are adequate for predicting human PK profiles.

2. PK/PD modeling in HT-1376 xenograft models

Model Parameters		Value	SE (%)
PK	CL (L/h/kg)	2.193	5.2
	CL_d (L/h/kg)	0.920	33.9
	V (L/kg)	2.686	10.8
	V2 (L/kg)	2.261	20.7
	V_{max} (mg/kg/h)	10.162	7.0
	K_m (mg/kg)	5.892	32.4
	FD_{50} (mg/kg)	5.089	10.7
PD	K_{ng0} (1/h)	0.004	3.3
	K_{ng1} (mm^3/h)	2.098	1.5
	K_{max} (1/h)	0.012	10.5
	KC_{50} (ng/mL)	176.047	13.2
	T0 (mm^3)	126.953	4.4
	τ (h)	99.853	14.0

Figure 5. Comparison of observed and predicted PK and Efficacy profiles in HT-1376 xenograft models

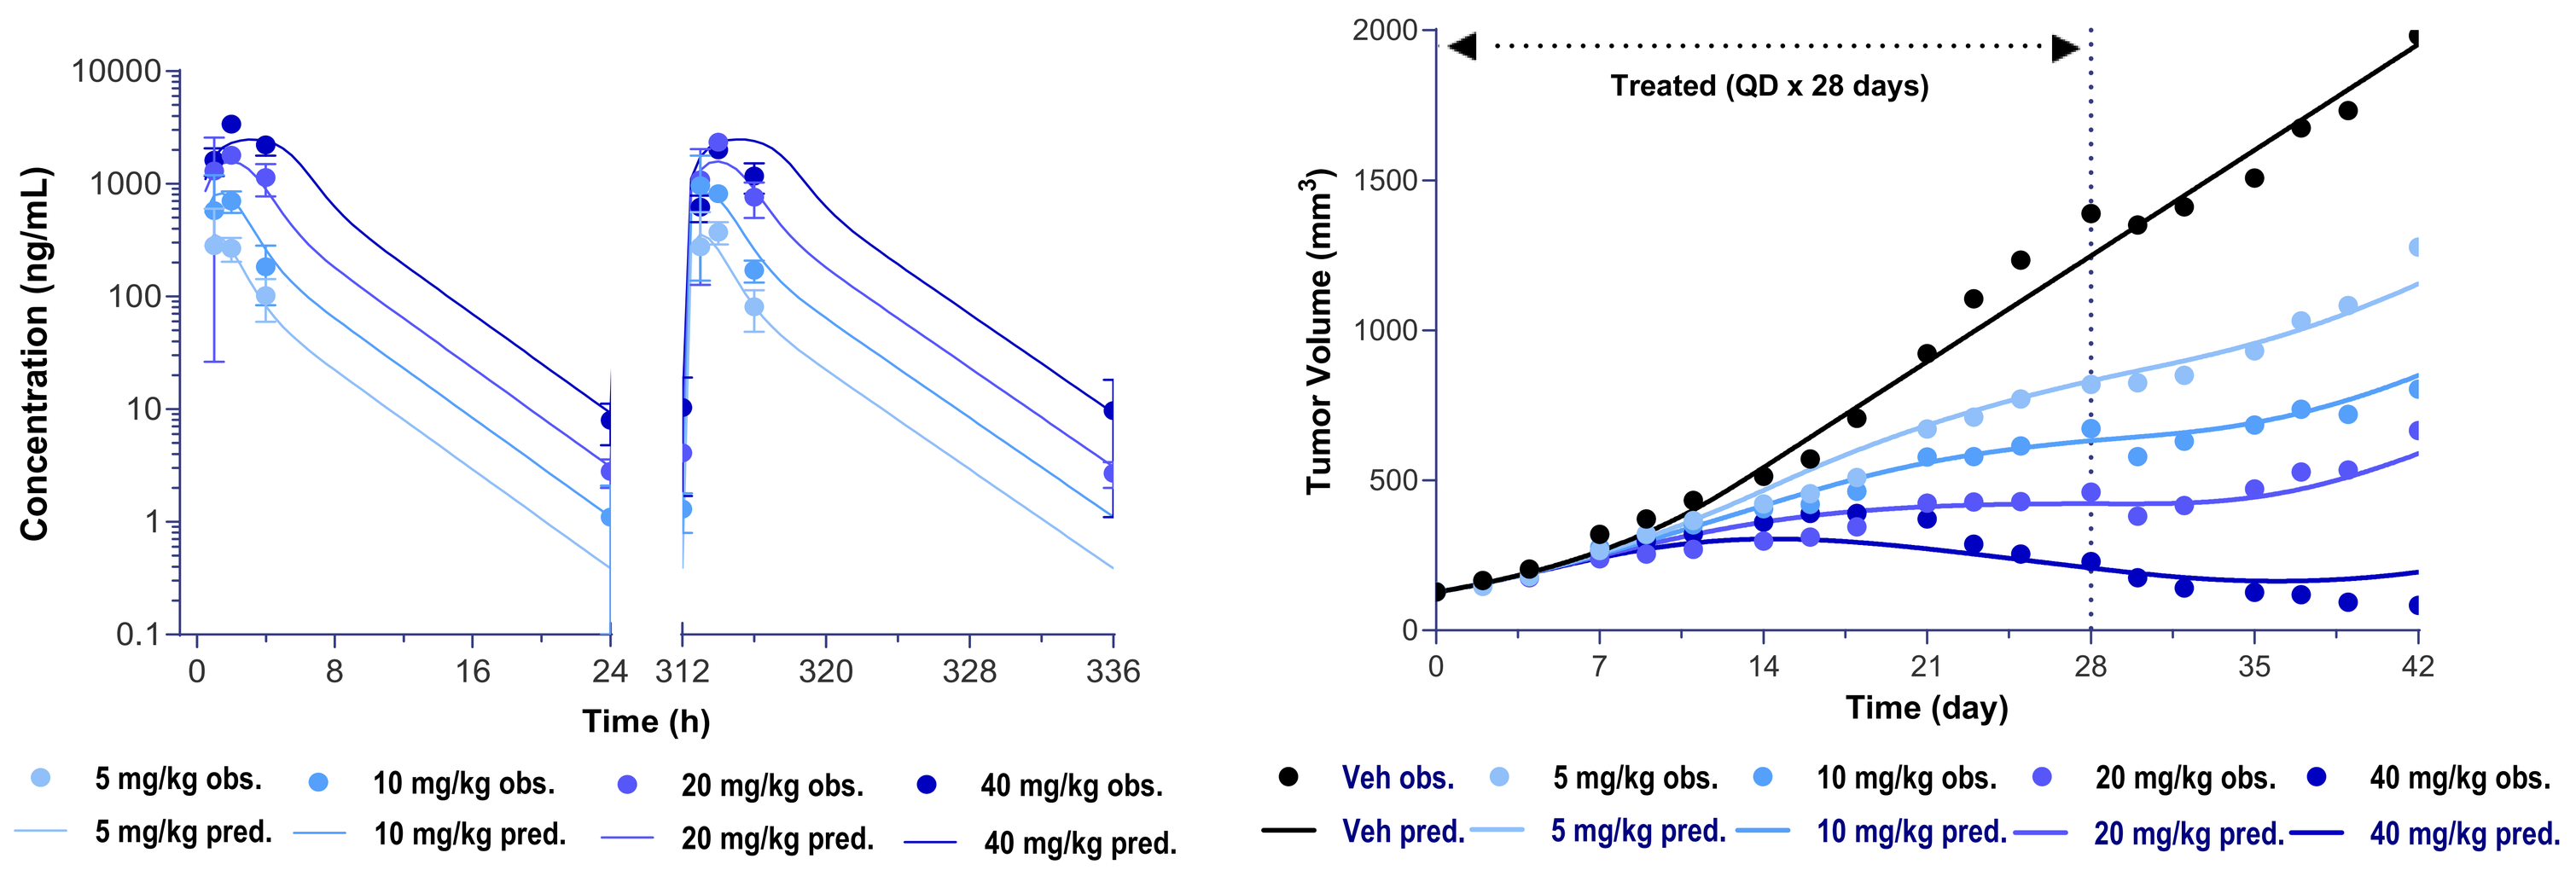
Concentration (ng/mL)
Time (h)
5 mg/kg obs.
10 mg/kg obs.
20 mg/kg obs.
40 mg/kg obs.
5 mg/kg pred.
10 mg/kg pred.
20 mg/kg pred.
40 mg/kg pred.
Treated (QD x 28 days)
Tumor Volume (mm^3)
Time (day)
Veh obs.
Veh pred.

- A two-compartment PK model with saturable absorption and non-linear bioavailability well described the overall PK profiles of HM97662 in HT-1376 xenograft models. The combined exponential and linear tumor growth model was able to reflect the HT-1376 tumor growth data (vehicle control) and delayed tumor growth inhibition of HM97662 as characteristics of epigenetic anti-cancer drugs was adequately described by signal distribution model (SDM) with three transit compartments.

3. Translational Human PK/PD & efficacious dose prediction

Figure 6. Simulated Human PK/PD profiles

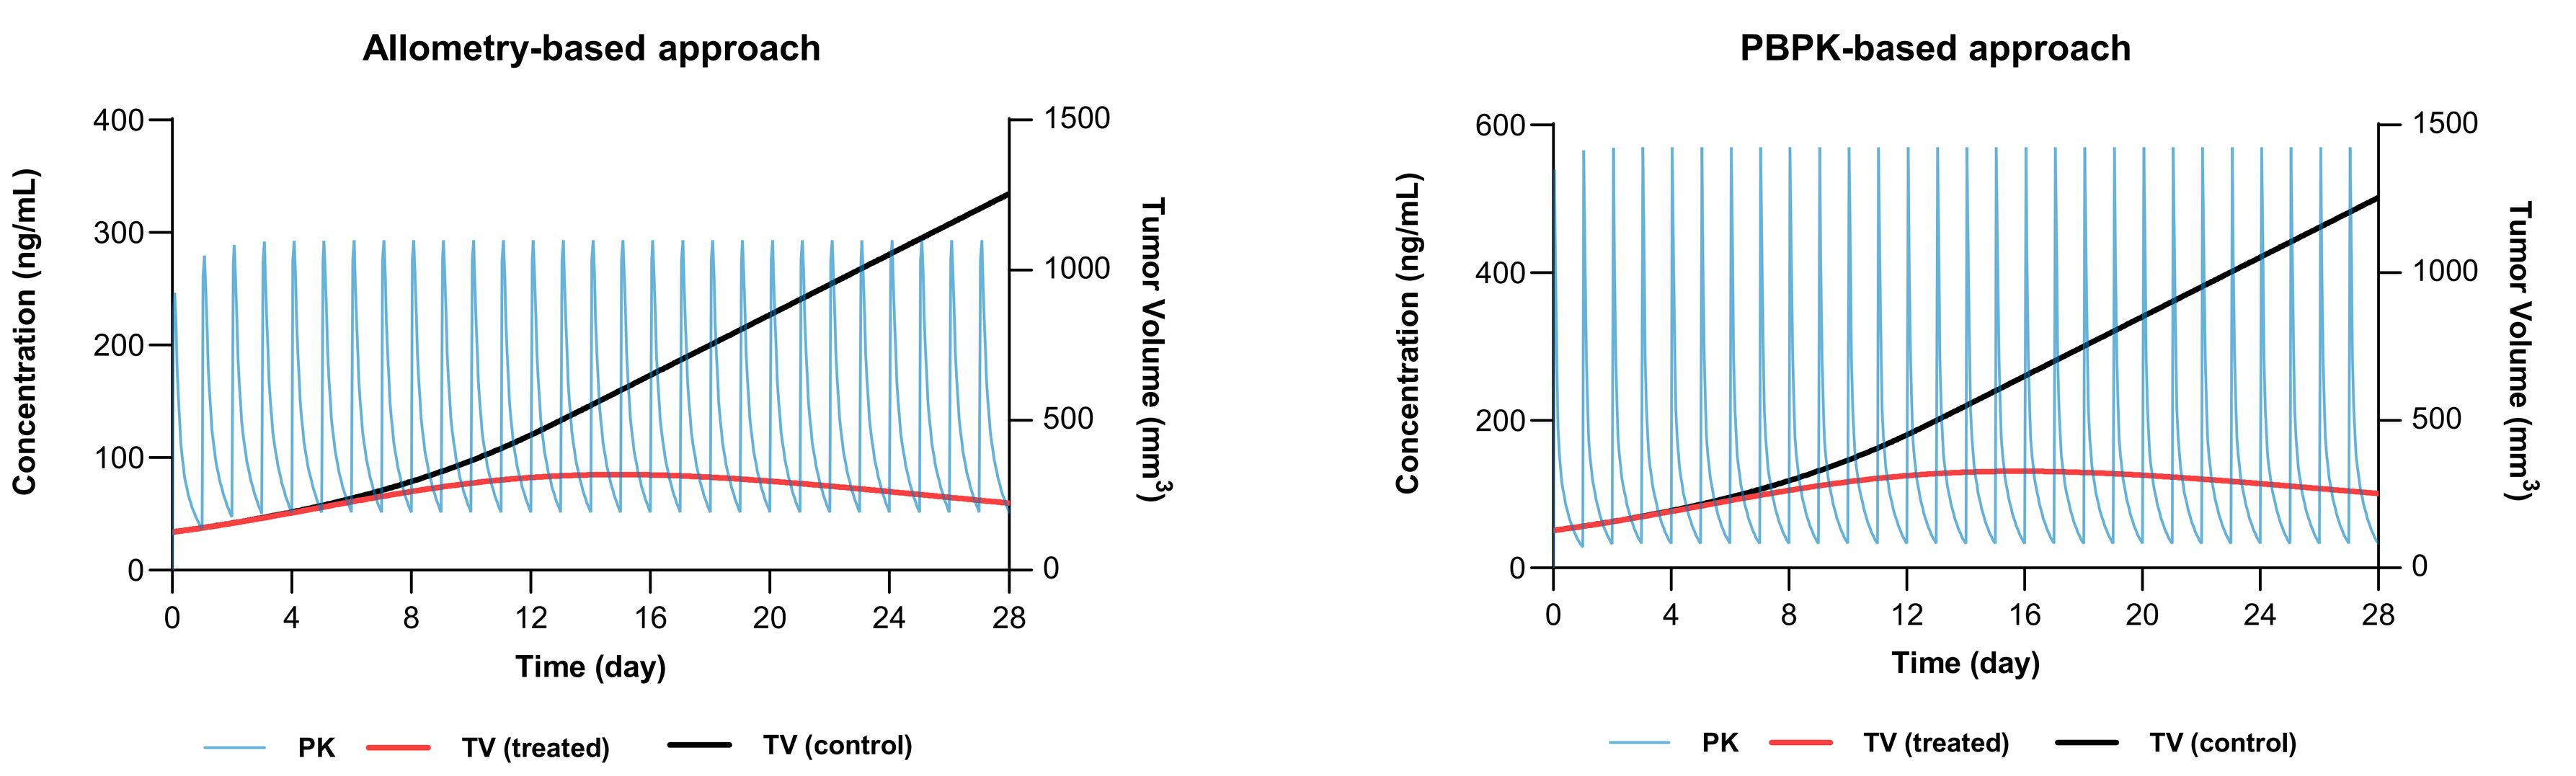
Allometry-based approach
PBPK-based approach
Concentration (ng/mL)
Tumor Volume (mm^3)
Time (day)
PK
TV (treated)
TV (control)

Figure 7. Simulated correlation between human dose & efficacy

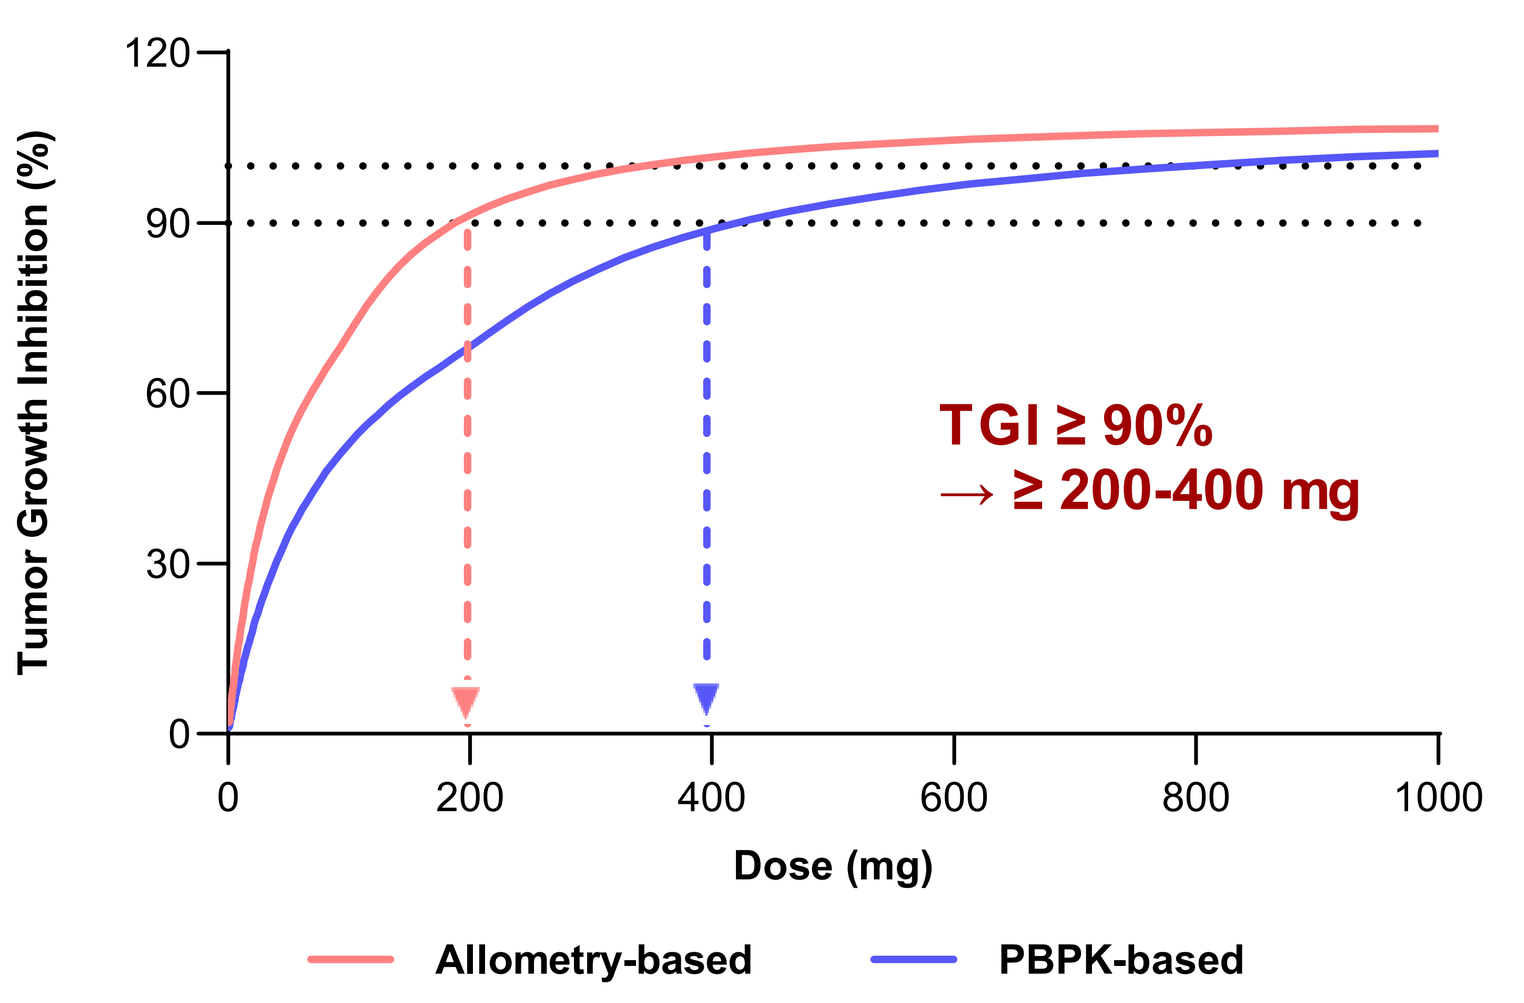
Tumor Growth Inhibition (%)
Dose (mg)
TGI ≥ 90%
→ ≥ 200-400 mg
Allometry-based
PBPK-based

Target efficacy	Predicted human efficacious dose	
	Allometry-based	PBPK-based
TGI 90% at end of cycle 1 (Day 28)	**200 mg QD**	**400 mg QD**

- The use of the simulated tumor growth inhibition as a target for the prediction of the human efficacious dose was supported from correlation between observed tumor growth inhibition and exposure level in xenograft models.
- Established PK/PD relationship of HM97662 in HT-1376 xenograft models were integrated to human PK model for projection of human PK/PD and therapeutic dose levels.
- HM97662 was predicted of a daily efficacious dose ranges from 200 to 400 mg as 90% tumor growth inhibition.

Conclusions

1. **The two different approaches (allometry-based and PBPK-based) were utilized in human PK prediction. The similar results (≤ 1.9-fold in all predicted parameters) were predicted in these two approaches. Therefore, the confidence in human PK prediction accuracy could be increased.**
2. **A PK/PD model of HM97662 based on preclinical data was developed from HT-1376 xenograft mouse model. Based on the translational PK/PD model incorporate with predicted human PK models of HM97662, the optimal human efficacious dose (≥ 90% tumor growth inhibition) was proposed to be clinically 200-400 mg once daily.**
3. **In general, prediction within 2-fold was considered as good prediction accuracy. The developed models adequately described the observed PK profiles in clinical with the simulated AUC_T, C_{max} and $t_{1/2}$ values within 1.8-fold of the observed values (Cohort 1: 50 mg).**
4. **Clinical data from the on-going study (HM-EZHI-101, NCT05598151) will be used to guide study design at future clinical efficacy levels and further model validation and refinement. Also, PK/PD relationships of HM97662 can be valuable to understand clinical PK/PD relations to therapeutic efficacy.**

References

1) BJ Ring *et al.*, PhRMA CPCDC initiative on predictive models of human pharmacokinetics, part 3: comparative assessment of prediction methods of human clearance. *J Pharm Sci.* **2011**;100(10):4090-4110.
2) RD Jones *et al.*, PhRMA CPCDC initiative on predictive models of human pharmacokinetics, part 2: Comparative assessment of prediction methods of human volume of distribution. *J Pharm Sci.* **2011**;100(10):4074-4089.
3) T Wajima *et al.*, Prediction of human pharmacokinetic profile in animal scale up based on normalizing time course profiles. *J Pharm Sci.* **2004**;93(7):1890-1900.
4) P Poulin *et al.*, *In vitro-in vivo* extrapolation of clearance: modeling hepatic metabolic clearance of highly bound drugs and comparative assessment with existing calculation methods. *J Pharm Sci.* **2012**;101(2):838-851.
5) J Yang *et al.*, Prediction of intestinal first-pass drug metabolism. *Curr Drug Metab.* **2007**;8(7):676-684.
6) HM Jones., Lost in translation? Predict first-in-human PK with Simcyp Discovery. *AAPS PharmSci 360.* **2022**;18th Oct.
7) J Yang *et al.*, Comparison of two pharmacodynamic transduction models for the analysis of tumor therapeutic responses in model systems. *AAPS J.* **2010**;12(1):1-10.
8) A Yin *et al.*, A Review of Mathematical Models for Tumor Dynamics and Treatment Resistance Evolution of Solid Tumors. *CPT Pharmacometrics Syst Pharmacol.* **2019**;8(10):720-737.
9) S Daouti *et al.*, Preclinical in vivo evaluation of efficacy, pharmacokinetics, and pharmacodynamics of a novel MEK1/2 kinase inhibitor RO5068760 in multiple tumor models. *Mol Cancer Ther.* **2010**;9(1):134-144.

Hanmi Pharm. Co., Ltd. (http://www.hanmipharm.com)