Synergistic tumor clearing effect of a novel long-acting IL-2 analog, HM16390, in combination with immune checkpoint inhibitors in cold tumor syngeneic mouse models

736-P Hanmi

Jinyoung Kim[1], Jaehyuk Choi[1], Sang Hyun Park[1], Yu Yon Kim[1], Jooyun Byun[1], Hocheol Shin[1], Sungmin Bae[1], Daejin Kim[1], In Young Choi[1,*]

[1]Hanmi Pharm. Co., Ltd., South Korea

BACKGROUND

Introduction & Objective: HM16390 is a novel long-acting IL-2 analog designed to induce the activation and expansion of cytotoxic effector cells through finely tuned binding affinities to IL-2 receptor subunits.

In this study, we investigated the immune cell profiles in the tumor microenvironment following the HM16390 injection, as well as the potential as a combination partner for immune checkpoint inhibitors (CPIs) in poorly immunogenic tumor models.

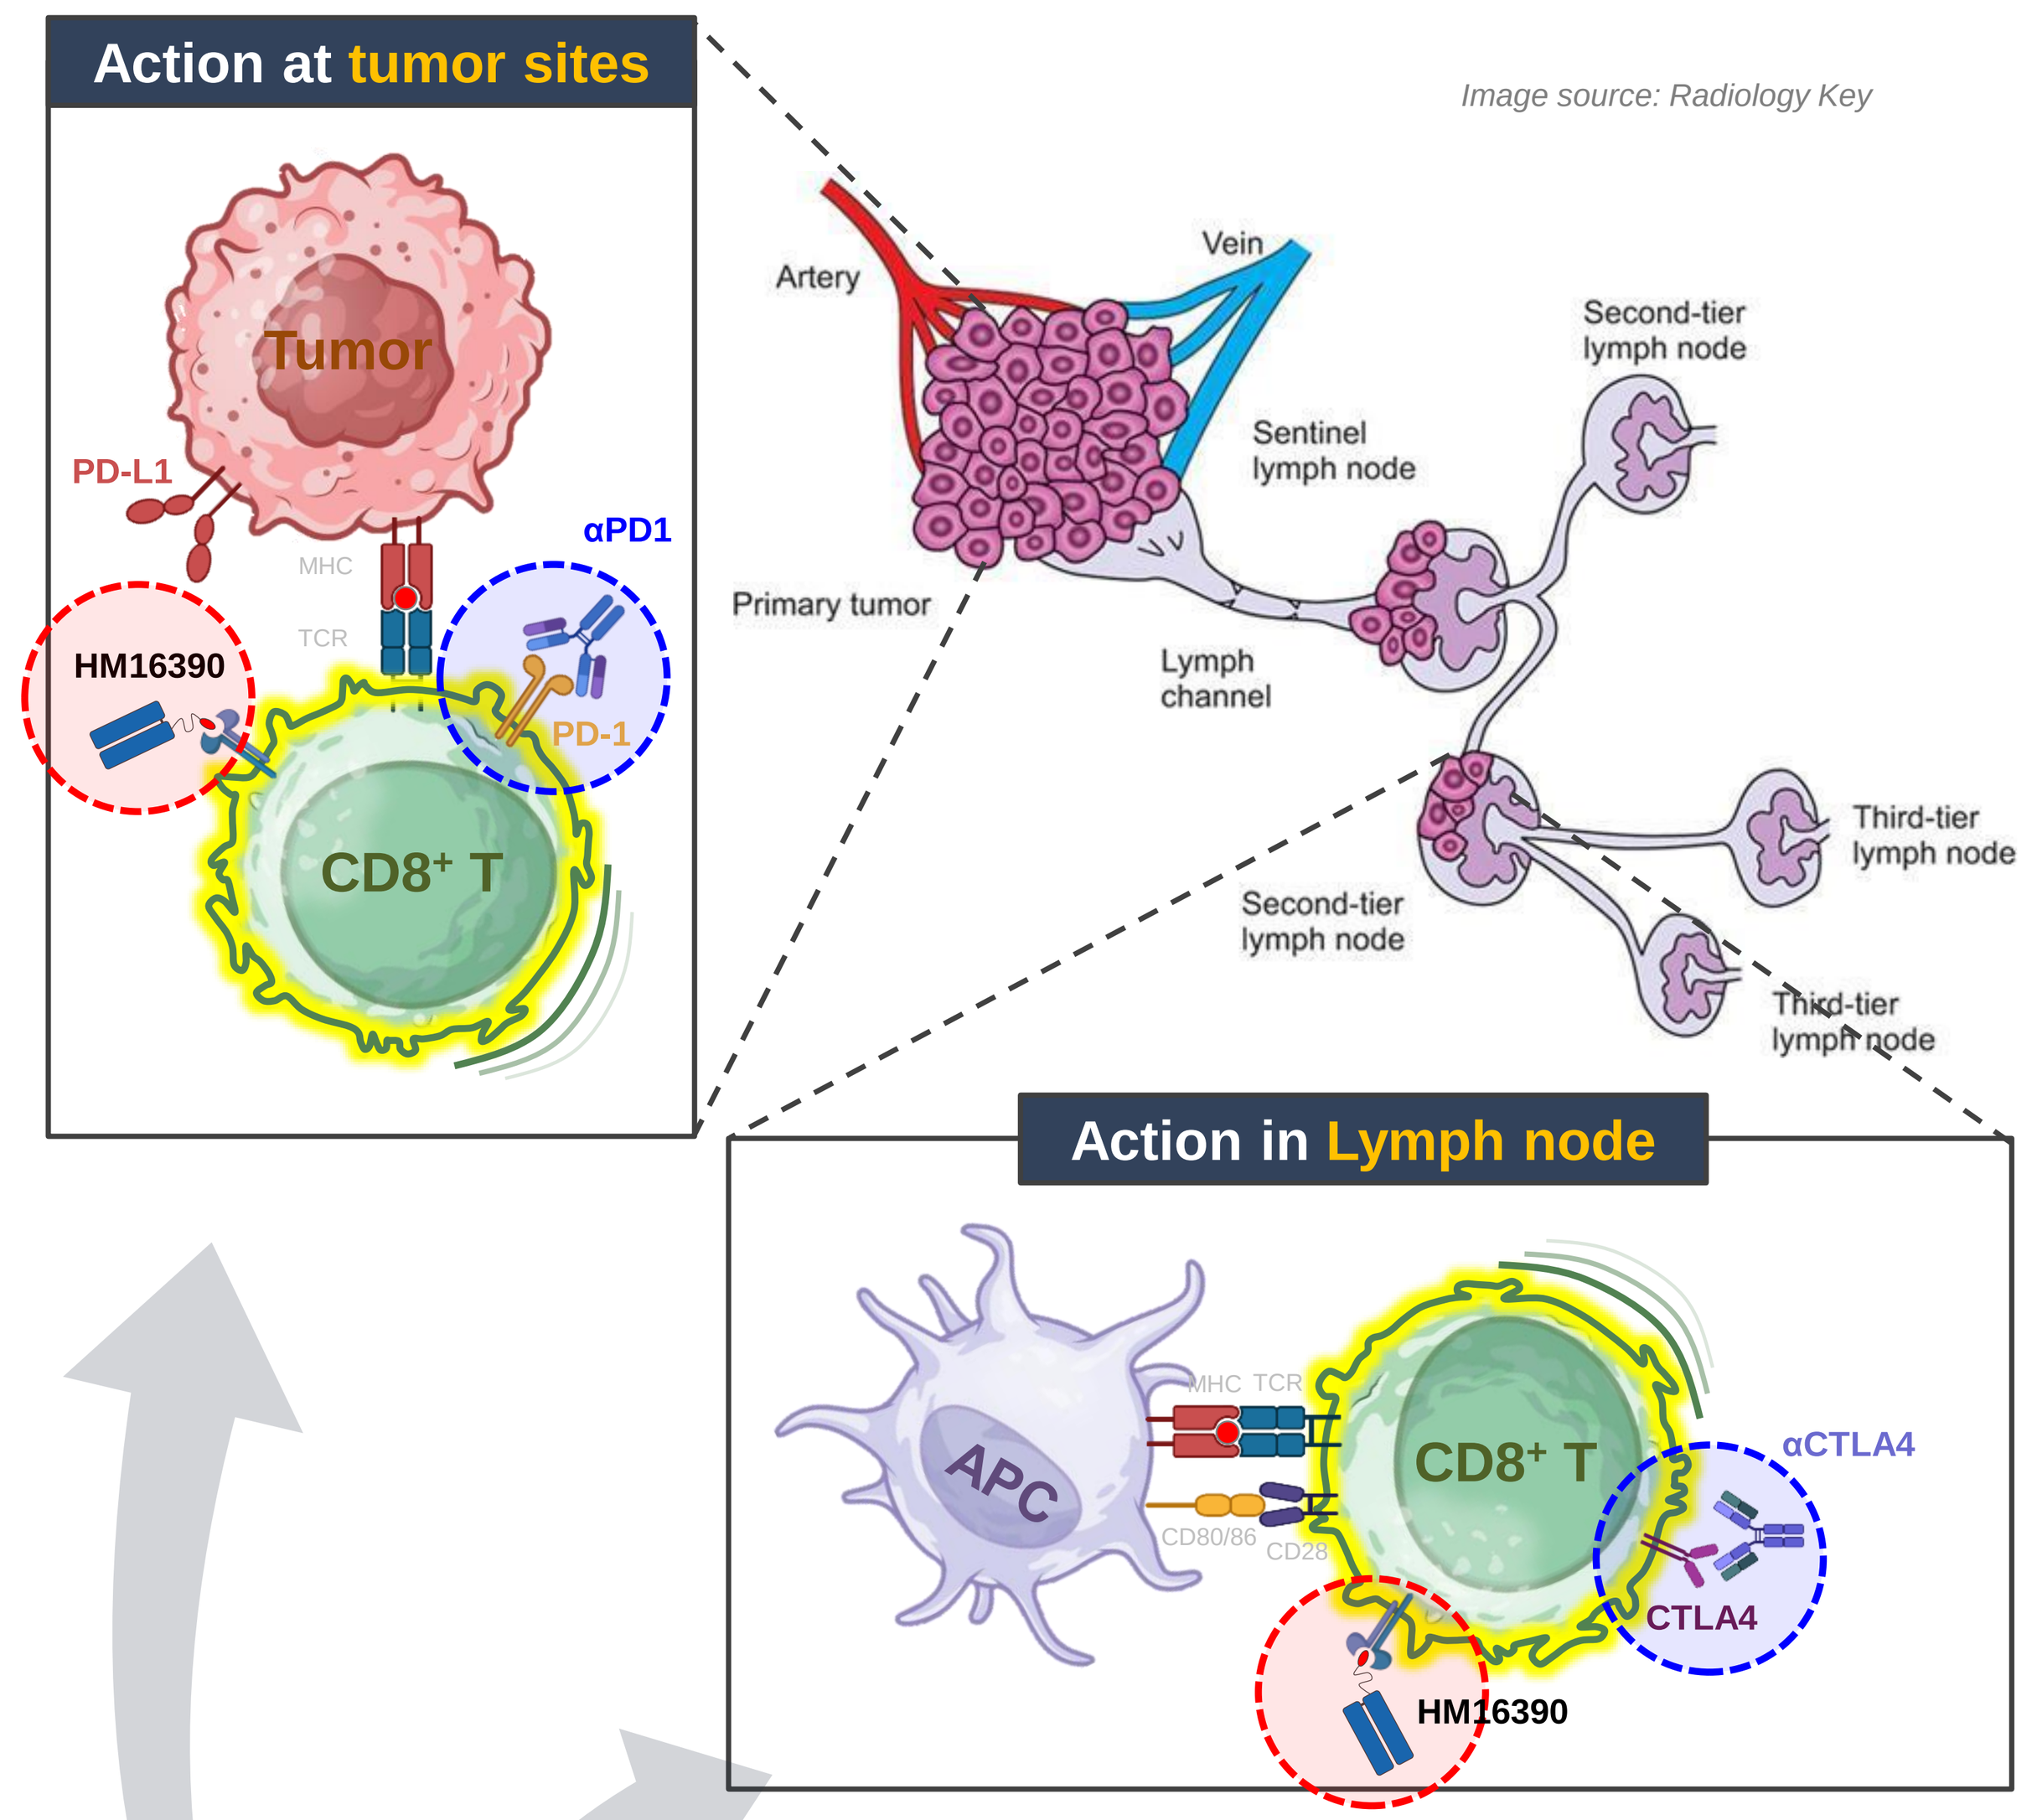
Action at tumor sites
Image source: Radiology Key
Tumor
PD-L1
MHC
TCR
αPD1
PD-1
HM16390
CD8+ T
Artery
Vein
Sentinel lymph node
Second-tier lymph node
Primary tumor
Lymph channel
Third-tier lymph node
Second-tier lymph node
Third-tier lymph node
Action in Lymph node
APC
MHC TCR
CD80/86
CD28
CD8+ T
αCTLA4
CTLA4
HM16390

HM16390 which is engineered to enhance the proliferation and stimulation of cytotoxic effector cells, could serve as a promising combination partner for CPIs by remodeling the tumor microenvironment (TME) into an immune-favorable condition.

HM16390

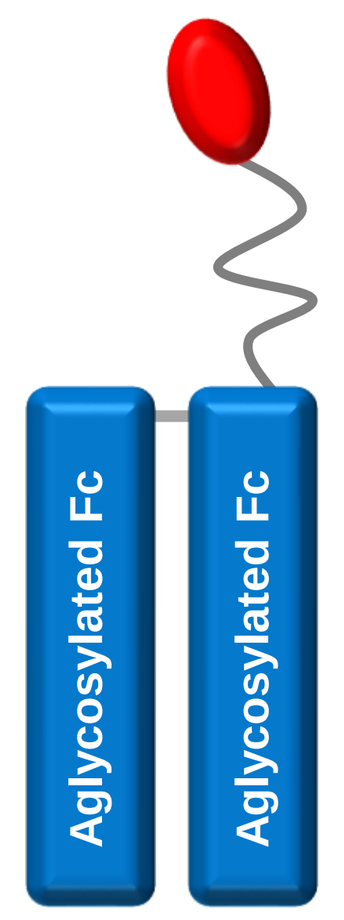
Aglycosylated Fc
Aglycosylated Fc

IL-2 analog rationally designed for intensive anti-tumor activity with immune balance

: Intensified IL-2Rβ binding elicits outstanding lymphocytes expansion

: Optimal IL-2Rα binding minimizes a risk of VLS and buffers an intensified IL-2Rβ binding derived CRS

Extended half-life allows once per immuno-therapies

Convenient s.c treatment option for patient adherence

METHOD & RESULTS

Tumor infiltrating lymphocytes (TILs) composition

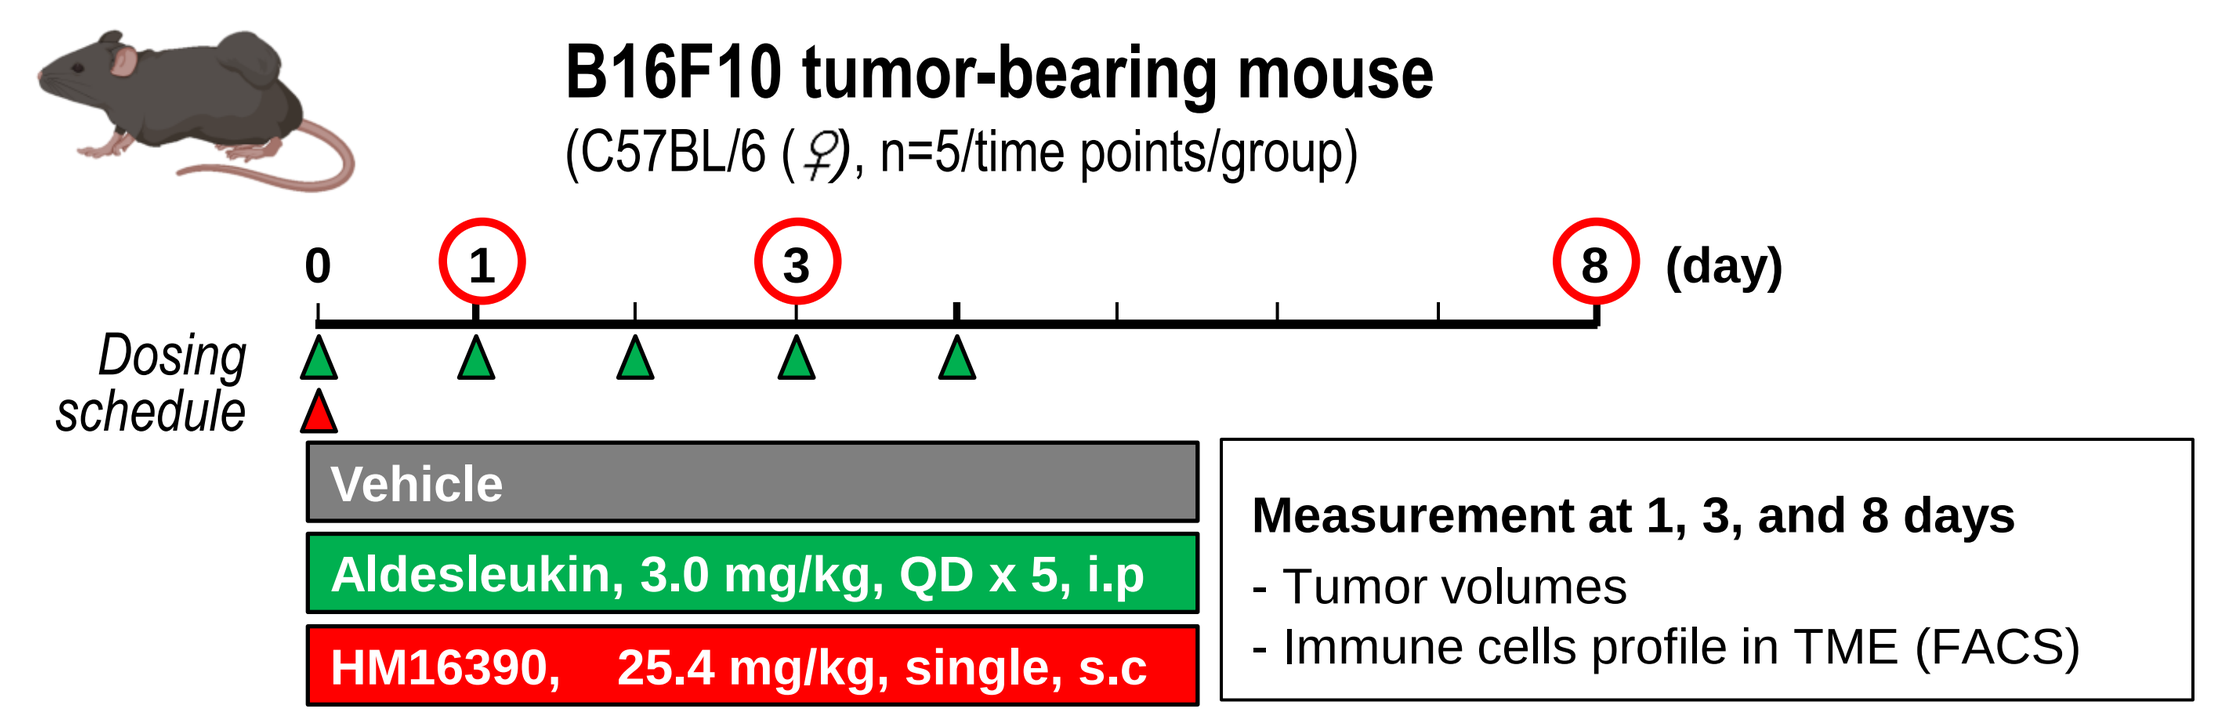
B16F10 tumor-bearing mouse
(C57BL/6 (♀), n=5/time points/group)
0
1
3
8
(day)
Dosing schedule
Vehicle
Aldesleukin, 3.0 mg/kg, QD x 5, i.p
HM16390, 25.4 mg/kg, single, s.c
Measurement at 1, 3, and 8 days
- Tumor volumes
- Immune cells profile in TME (FACS)

➢ TILs were evaluated in poorly immunogenic tumor model, B16F10 tumor-bearing mouse, after single s.c administration of HM16390 or 5 consecutive day i.p administration of aldesleukin, respectively.

Figure 1. Profiles of Tumor infiltrating lymphocytes in B16F10 syngeneic mouse model

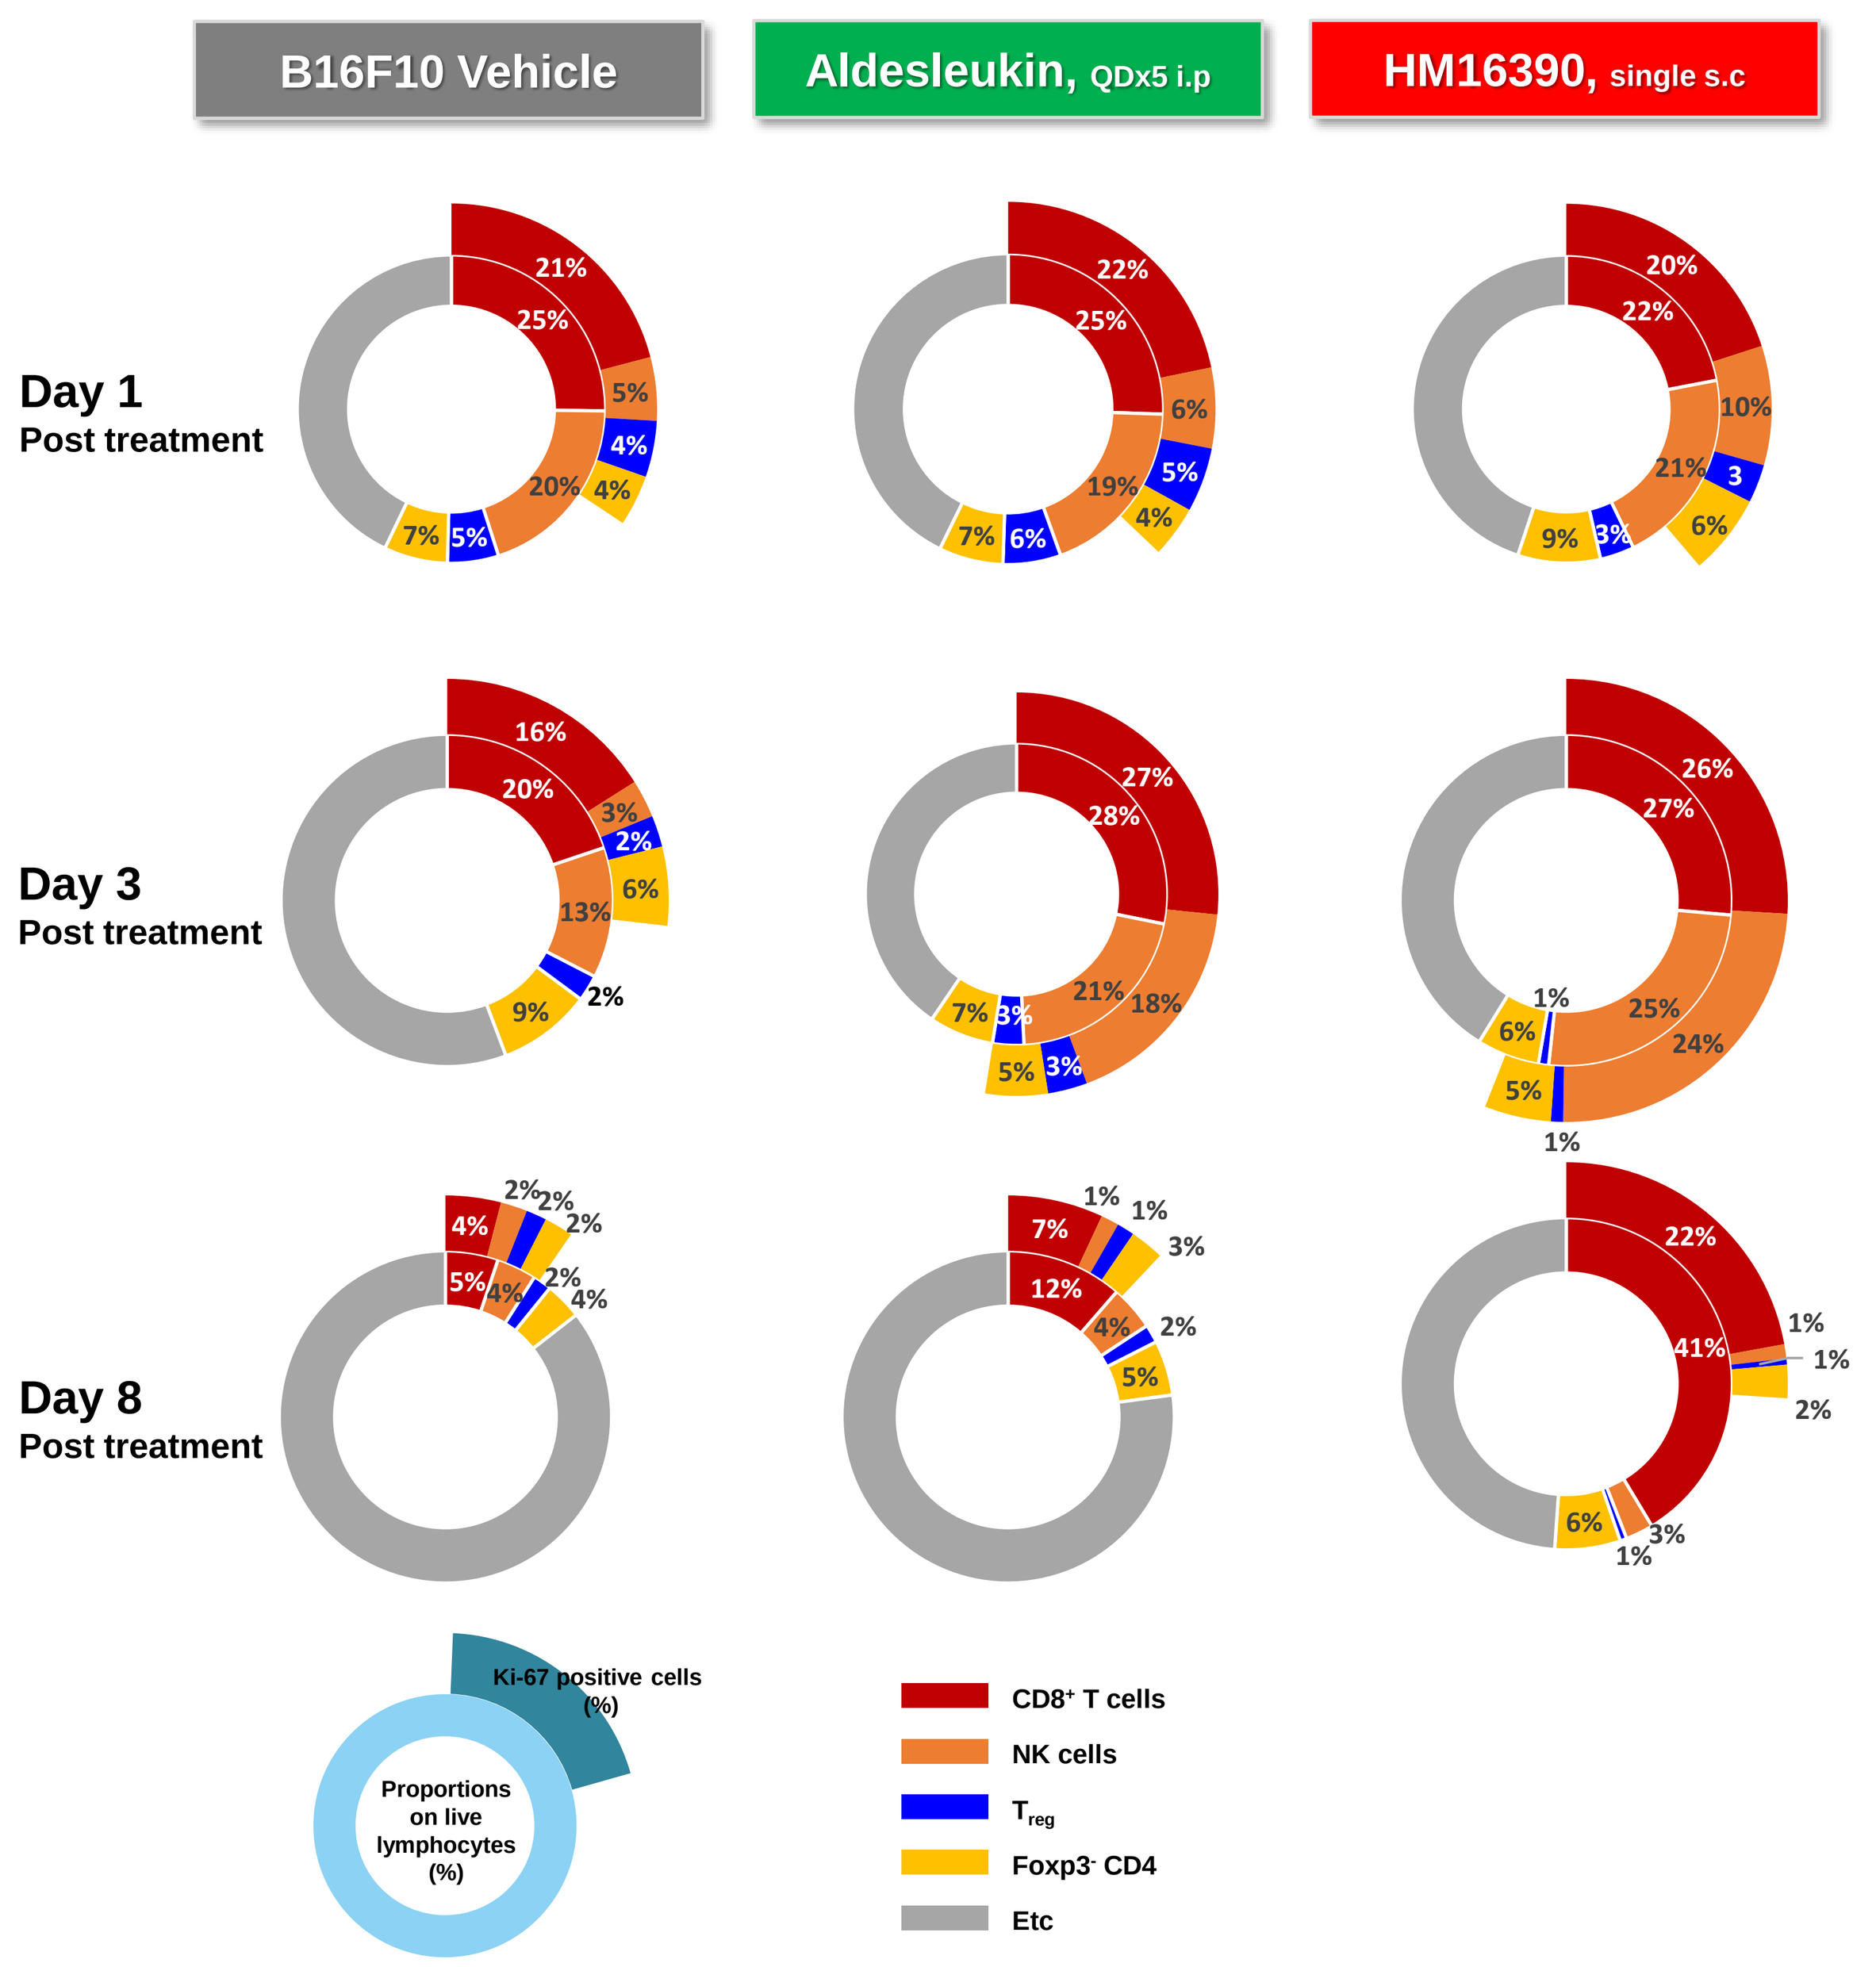
B16F10 Vehicle
Aldesleukin, QDx5 i.p
HM16390, single s.c
Day 1
Post treatment
Day 3
Post treatment
Day 8
Post treatment
Ki-67 positive cells (%)
Proportions on live lymphocytes (%)
CD8+ T cells
NK cells
Treg
Foxp3- CD4
Etc

➢ A single s.c treatment with HM16390 at 25.4 mg/kg (6.0 mg/kg as IL-2) resulted in a more favorable anti-tumoral immune cell composition, compared to 5 consecutive day i.p treatments with aldesleukin at 3.0 mg/kg in B16F10 syngeneic mouse model (FACS analysis, n=5/time point).

Synergistic effect with CPIs in poorly immunogenic tumor model

Part A. Synergistic effect by combo with CPIs in melanoma model

Figure 2. Bliss independence analysis when combined with CPIs in B16F10 syngeneic mouse model (2 independent studies)

(a) Combination with anti-PD1 (Study #1, n=8/group)

(b) Combination with anti-CTLA4 (Study #2, n=7/group)

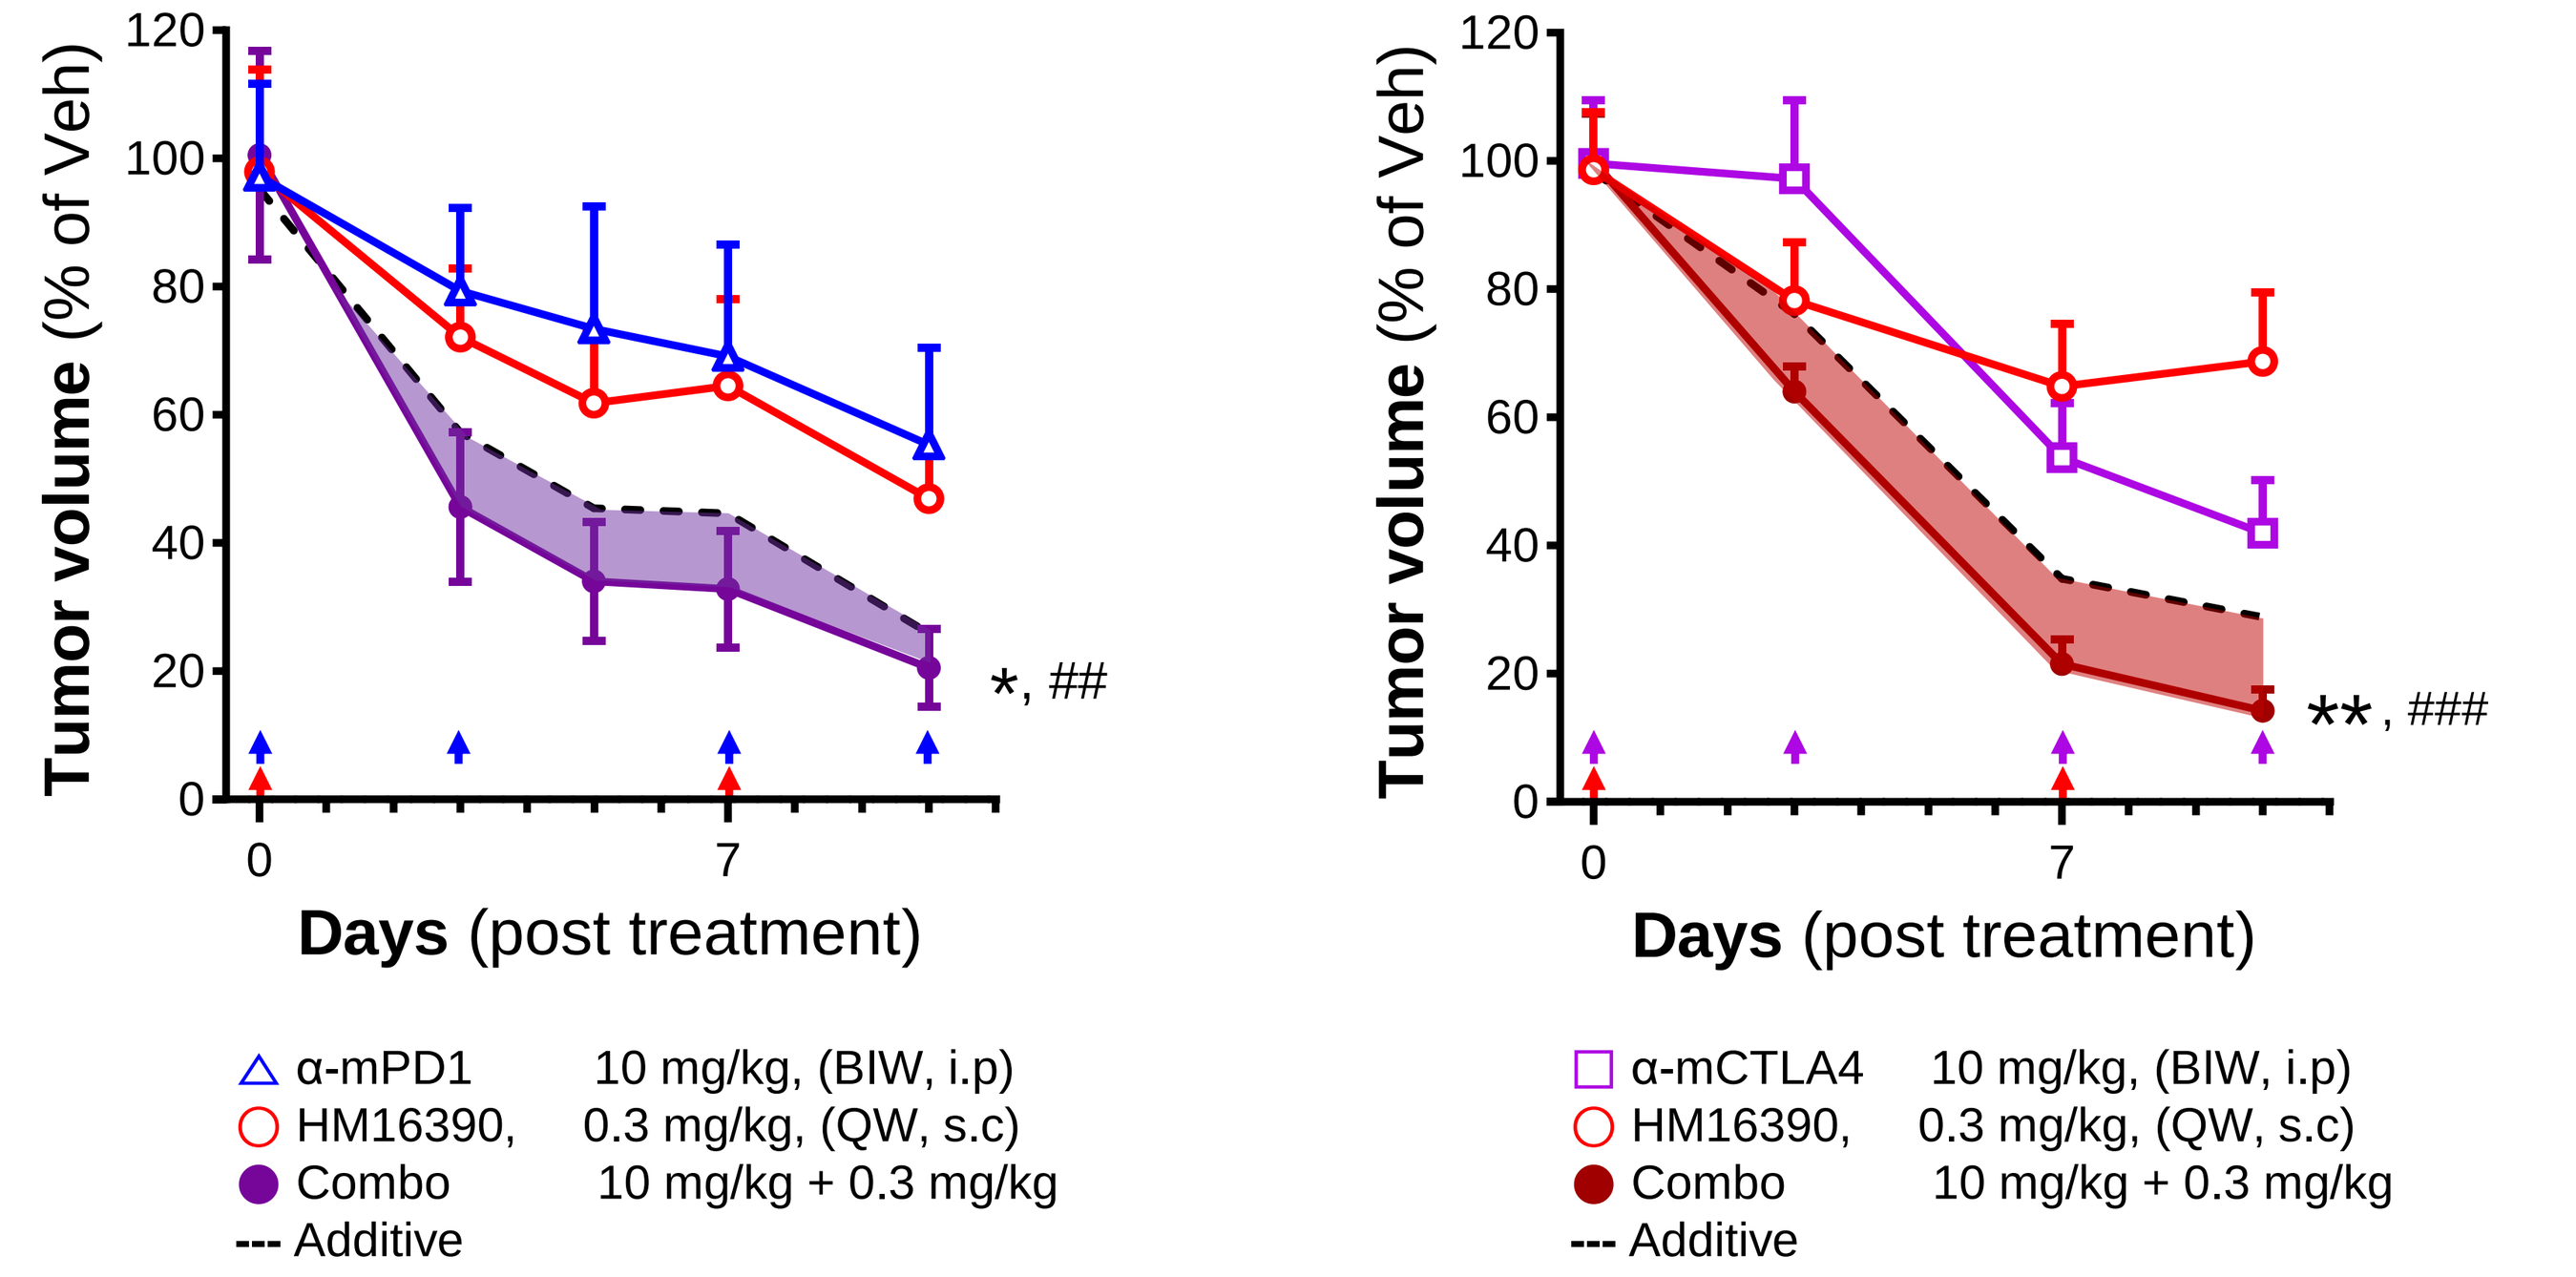
Tumor volume (% of Veh)
Days (post treatment)
*, ##
**, ###
α-mPD1 10 mg/kg, (BIW, i.p)
HM16390, 0.3 mg/kg, (QW, s.c)
Combo 10 mg/kg + 0.3 mg/kg
--- Additive
α-mCTLA4 10 mg/kg, (BIW, i.p)
HM16390, 0.3 mg/kg, (QW, s.c)
Combo 10 mg/kg + 0.3 mg/kg
--- Additive

➢ HM16390 exhibited significant anti-tumor activity compared to α-mPD1 or α-mCTLA4 in the B16F10 mice. Combination with each CPI resulted in a synergistic effect, further enhancing therapeutic efficacy on tumor growth inhibition. Additivity of individual treatments (%) = normalized tumor volume by vehicle (NTV) at day n (%) of one mono-treatment groups volume x NTV at same day of the other mono-treatment group volume/100. α-mPD1, anti-mouse PD1 antibody; α-mCTLA4, anti-mouse CTLA4 antibody. **p<0.01, *p<0.05 *vs.* CPI mono; ###p<0.001, ##p<0.01 *vs.* HM16390 mono by unpaired t-test.

Part B. Synergistic effect by combo with CPIs in TNBC model

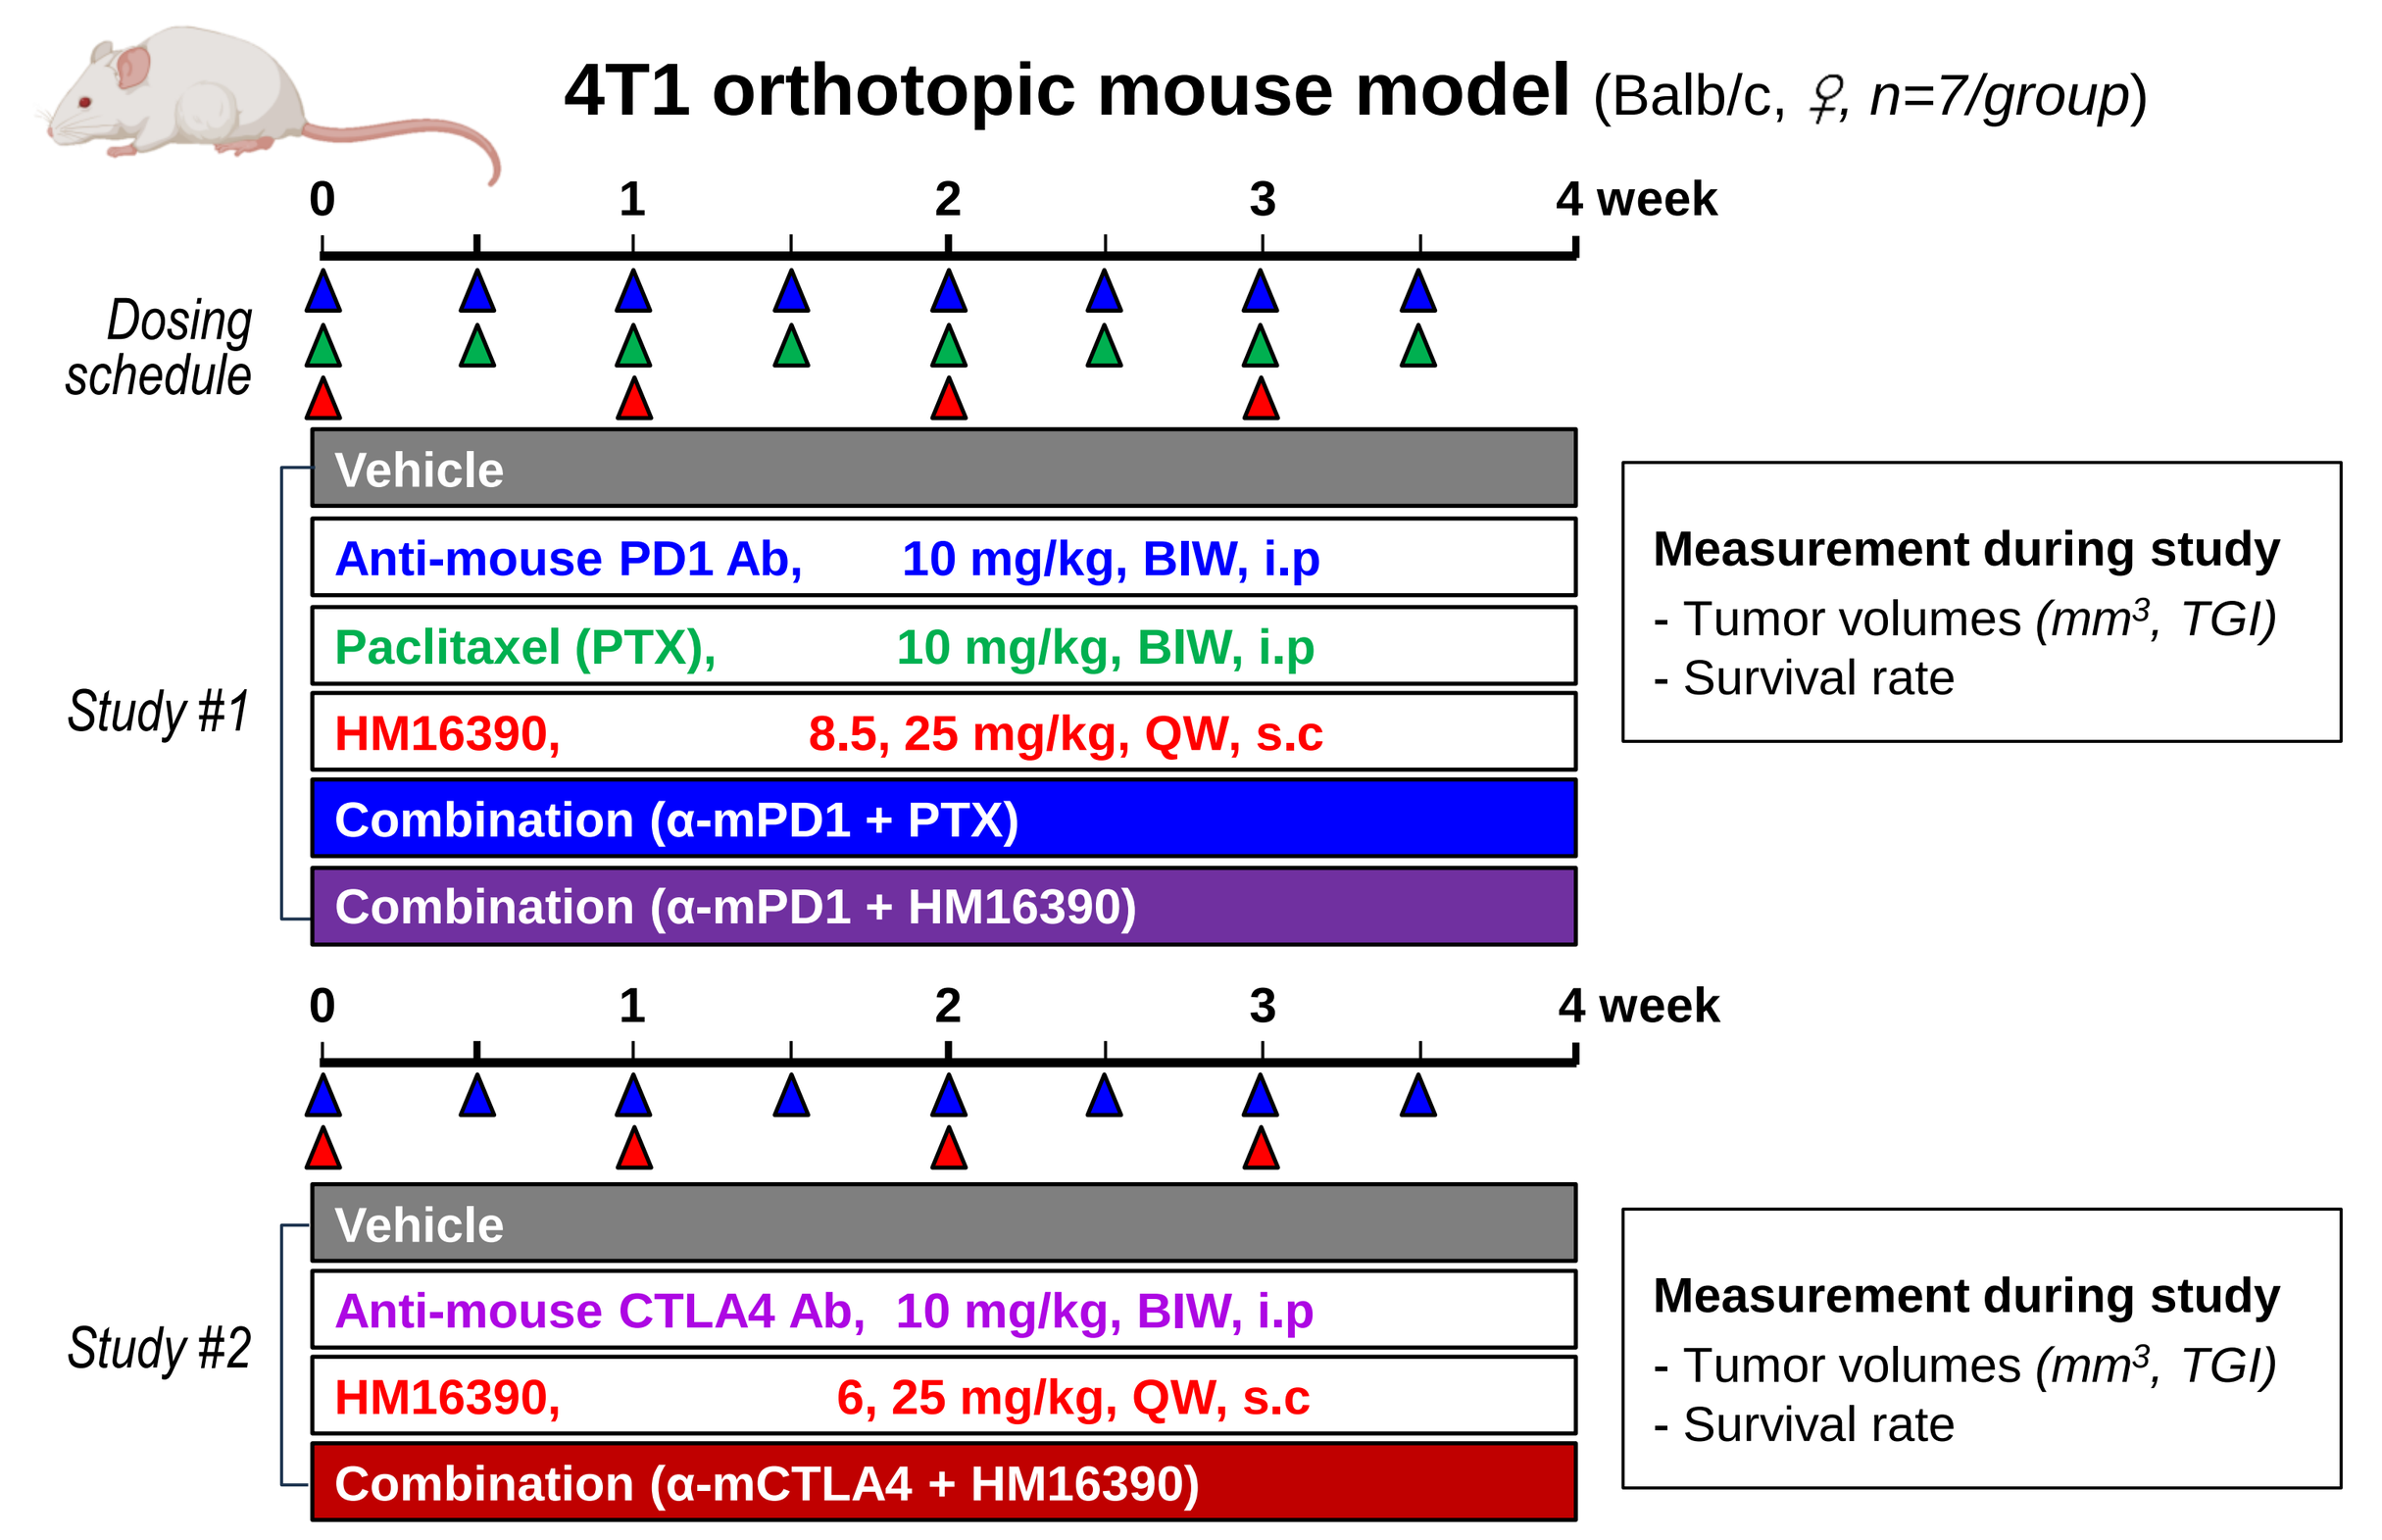
4T1 orthotopic mouse model (Balb/c, ♀, n=7/group)
0
1
2
3
4 week
Dosing schedule
Study #1
Vehicle
Anti-mouse PD1 Ab, 10 mg/kg, BIW, i.p
Paclitaxel (PTX), 10 mg/kg, BIW, i.p
HM16390, 8.5, 25 mg/kg, QW, s.c
Combination (α-mPD1 + PTX)
Combination (α-mPD1 + HM16390)
Measurement during study
- Tumor volumes (mm3, TGI)
- Survival rate
Study #2
Vehicle
Anti-mouse CTLA4 Ab, 10 mg/kg, BIW, i.p
HM16390, 6, 25 mg/kg, QW, s.c
Combination (α-mCTLA4 + HM16390)
Measurement during study
- Tumor volumes (mm3, TGI)
- Survival rate

Figure 3. Synergistic effect with CPIs in 4T1 mouse model (2 independent studies)

(a) Synergistic anti-tumor effect of HM16390 after combination with anti-PD1

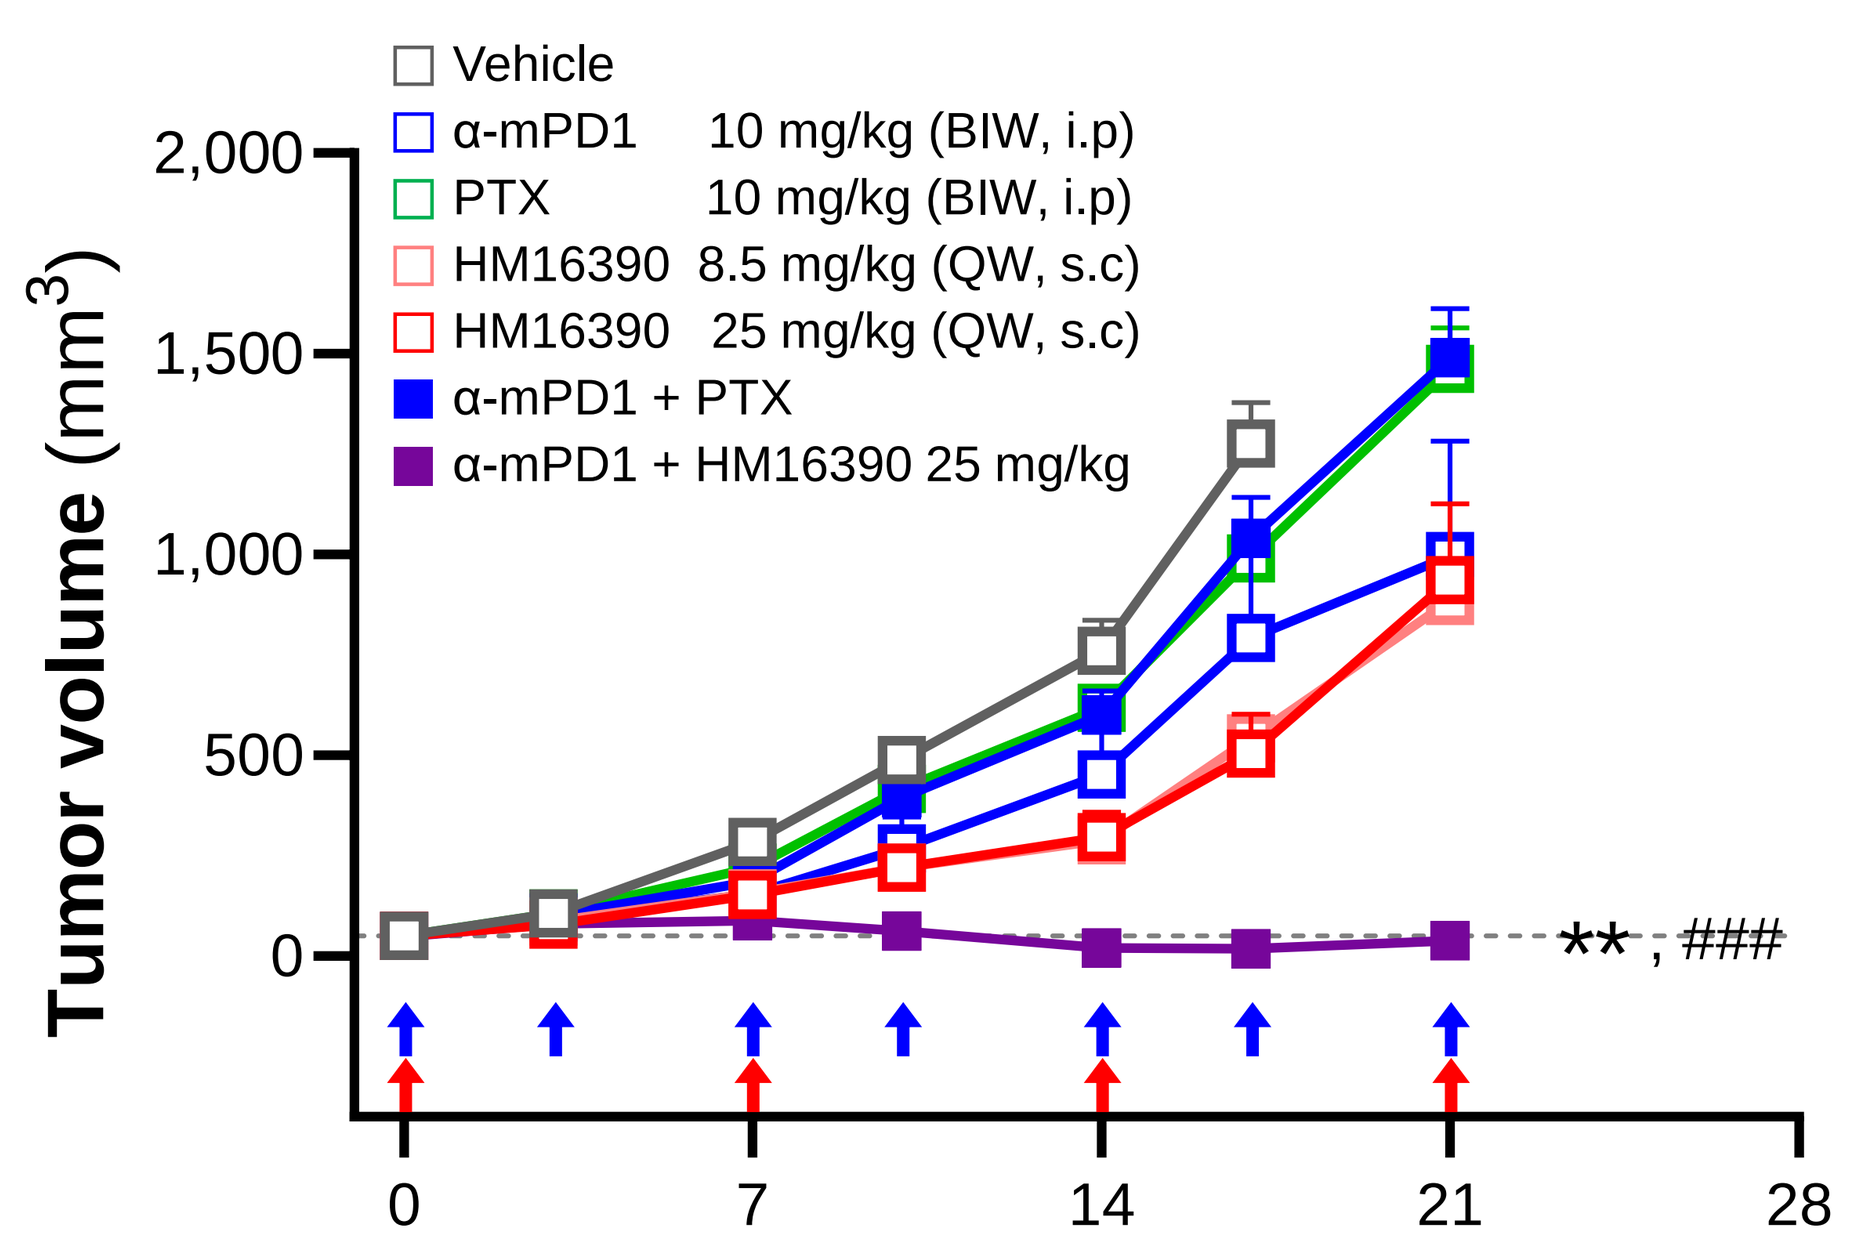
Vehicle
α-mPD1 10 mg/kg (BIW, i.p)
PTX 10 mg/kg (BIW, i.p)
HM16390 8.5 mg/kg (QW, s.c)
HM16390 25 mg/kg (QW, s.c)
α-mPD1 + PTX
α-mPD1 + HM16390 25 mg/kg
Tumor volume (mm3)
**, ###
Time (days)

Therapy	Mono					Combination	
	Vehicle	αPD1 (BIW)	PTX (BIW)	HM16390 (QW)		αPD1 + PTX	HM16390 + αPD1
Dose (mg/kg)	-	10	10	8.5	25	10 + 10	10 + 25
TGI @D17	-	39.4%	23.3%	59.7%	62.9%	19.1%	102.5%
Survival	0%	14.3%	0%	0%	14.3%	0%	85.7%
CR	No	No	No	No	14.3%	No	57.1%

(b) Synergistic anti-tumor effect of HM16390 after combination with anti-CTLA4

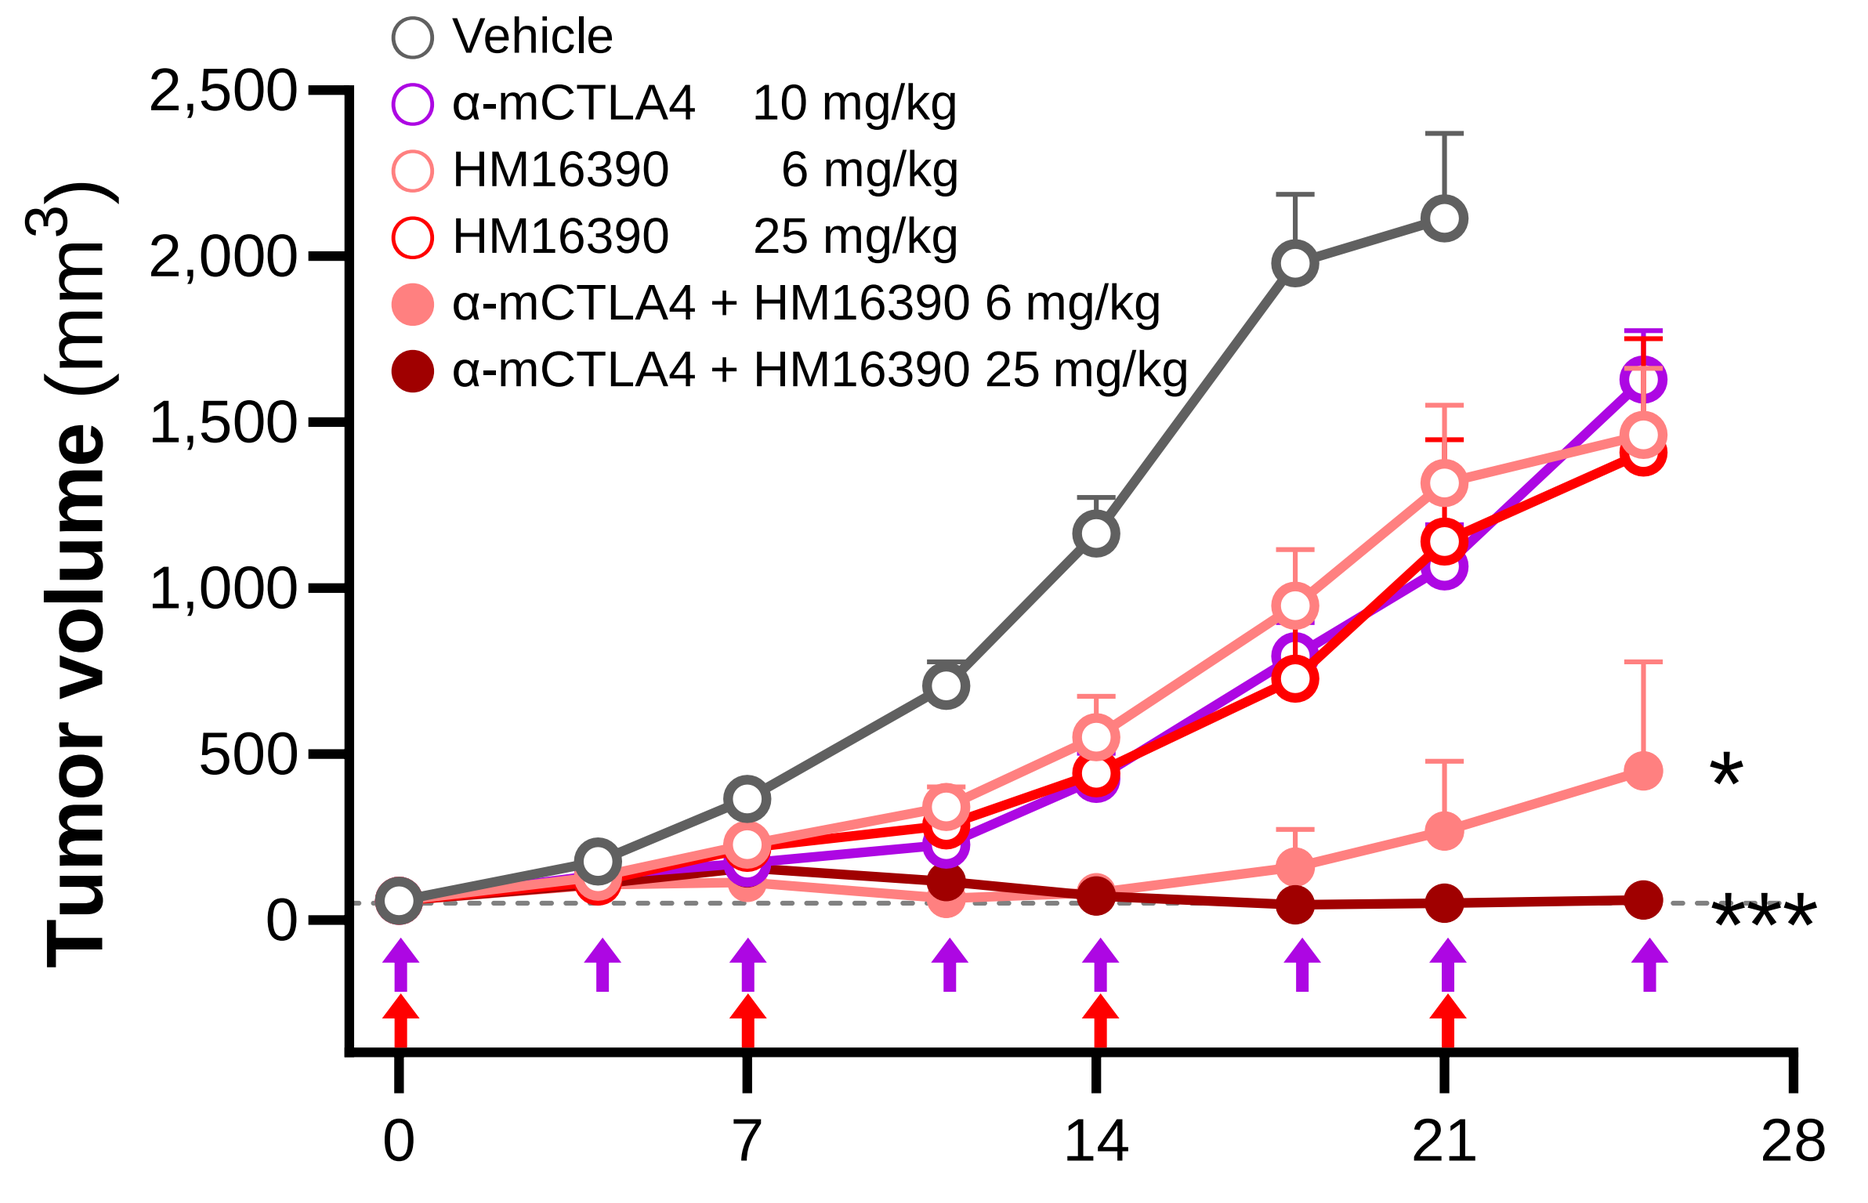
Vehicle
α-mCTLA4 10 mg/kg
HM16390 6 mg/kg
HM16390 25 mg/kg
α-mCTLA4 + HM16390 6 mg/kg
α-mCTLA4 + HM16390 25 mg/kg
Tumor volume (mm3)
*

Time (days)

Therapy	Mono				Combination	
	Vehicle	αCTLA4 (BIW)	HM16390 (QW)		αCTLA4 + HM16390	
Dose (mg/kg)	-	10	6	25	6 + 25	10 + 25
TGI @D21		51.0%	29.7%	47.4%	89.8%	100.3%
Survival	0%	14.3%	14.3%	57.1%	85.7%	100%
CR	No	No	No	14.3%	42.9%	42.9%

➢ HM16390 demonstrated synergistic anti-tumor activity in terms of survival rate, TGI, and CR when combined with α-mPD1 (a) or α-mCTLA4 antibodies (b) in the 4T1 triple negative breast cancer mouse model. Mice with tumor volumes of more than 2,000 mm^3 were sacrificed or considered as sacrificed. ###p<0.001 *vs.* PTX + α-mPD-1, ***p<0.001, **p<0.01, *p<0.05 *vs.* CPI mono by one-way ANOVA test. α-mPD1, anti-mouse PD1 antibody; PTX, paclitaxel; α-mCTLA4, anti-mouse CTLA4 antibody.

CONCLUSIONS

- **HM16390, a novel long-acting IL-2, is designed to enhance proliferation and activation of cytotoxic effector cell via intensified IL-2Rβ binding, while controlling excessive immune responses through optimal IL-2Rα binding.**
- **Anti-CTLA4 plays a critical role in the early activation phase of T cells, particularly in the lymph nodes, while anti-PD1 acts later in the immune response, after T cells have been activated and have reached the tumor site. The results suggest that HM16390 may have a synergistic effect when combined with these CPIs, which target different stages of the anti-tumor immune response through significant modulation of the TME, making it a potentially ideal combination partner.**

This research was supported by Korea Drug Development Fund funded by Ministry of Science and ICT, Ministry of Trade, Industry, and Energy, and Ministry of Health and Welfare (RS-2022-00165557, Republic of Korea)

*Corresponding author: In Young Choi (iychoi@hanmi.co.kr)

Hanmi Pharm. Co., Ltd.