Beneficial effect on intestinal growth of a long-acting GLP-2 analog, HM15912, after treatment switching from conventional GLP-2 drug or other long-acting GLP-2 analogs under clinical development in animal model

#P6855

Cho Rong Park, Jaehyuk Choi, Eun Jin Park, Hyunjoo Kwon, Sungmin Bae, Daejin Kim, Young Hoon Kim, **In Young Choi**

[1]Hanmi Pharm. Co., Ltd, Seoul, South Korea

ABSTRACT

The widespread use of teduglutide, which is only approved GLP-2 analog drug for short bowel syndrome (SBS), may be still limited due to insufficient efficacy and leading to a significant burden for patients by daily administration. Hence, several long-acting GLP-2 analog drugs targeting once a week are currently in clinical development. Here, we investigated that HM15912, a novel long-acting GLP-2 analog, enabled to achieve additional small bowel trophic effect after switching from those existing GLP-2 analogs.

To investigate additional intestinotrophic efficacy after switching from teduglutide to HM15912, C57BL/6 mice treated with twice daily administration of teduglutide for 2 weeks were switched to once-weekly administration of HM15912, or continued the typical treatment of teduglutide for the remaining 2 weeks.

HM15912 treatment significantly increased wet weight of SI (72.9% over vehicle) compared to teduglutide treated group (39.4% over vehicle) after 2 weeks. After 2 more weeks treatment, while showing the maintained the increased SI weight in teduglutide treated group (58.3% at week 3 and 41.1% at week 4 over vehicle), SI weight was further increased after switching to HM15912 (61.2% at week 3 and 68.5% at week 4 over vehicle). Next, we synthesized GLP-2 analogs to have same sequences with glepaglutide and apraglutide. SD rats treated with every other day administration of these long acting GLP-2 analogs for 2 weeks were switched to HM15912 or continued the typical treatment of them for the remaining 2 weeks. HM15912 treatment significantly increased wet weight of SI compared to weekly GLP-2 analog drug treated groups after 2 weeks (84.4% versus 41.5% or 26.6% over vehicle). After 2 more weeks treatment, while showing the slight increment in SI weight in the long-acting GLP-2 analogs (50.5% and 37.4% over vehicle), SI weight was further increased after switching to HM15912 (82.1% and 90.7% over vehicle, respectively). In line with these results, D-xylose absorption capacity was also significantly increased after switching from existing GLP-2 analogs to HM15912.

As a result, HM15912 significantly promoted small intestinal growth than existing GLP-2 analogs even with a less frequent dosing interval of once a week in rodents.

BACKGROUND

HM15912, a long-acting GLP-2 analog, provided significant morphological and functional improvement in small intestine compared to daily or weekly GLP-2 medications via substantially extended half-life and systemic exposure.

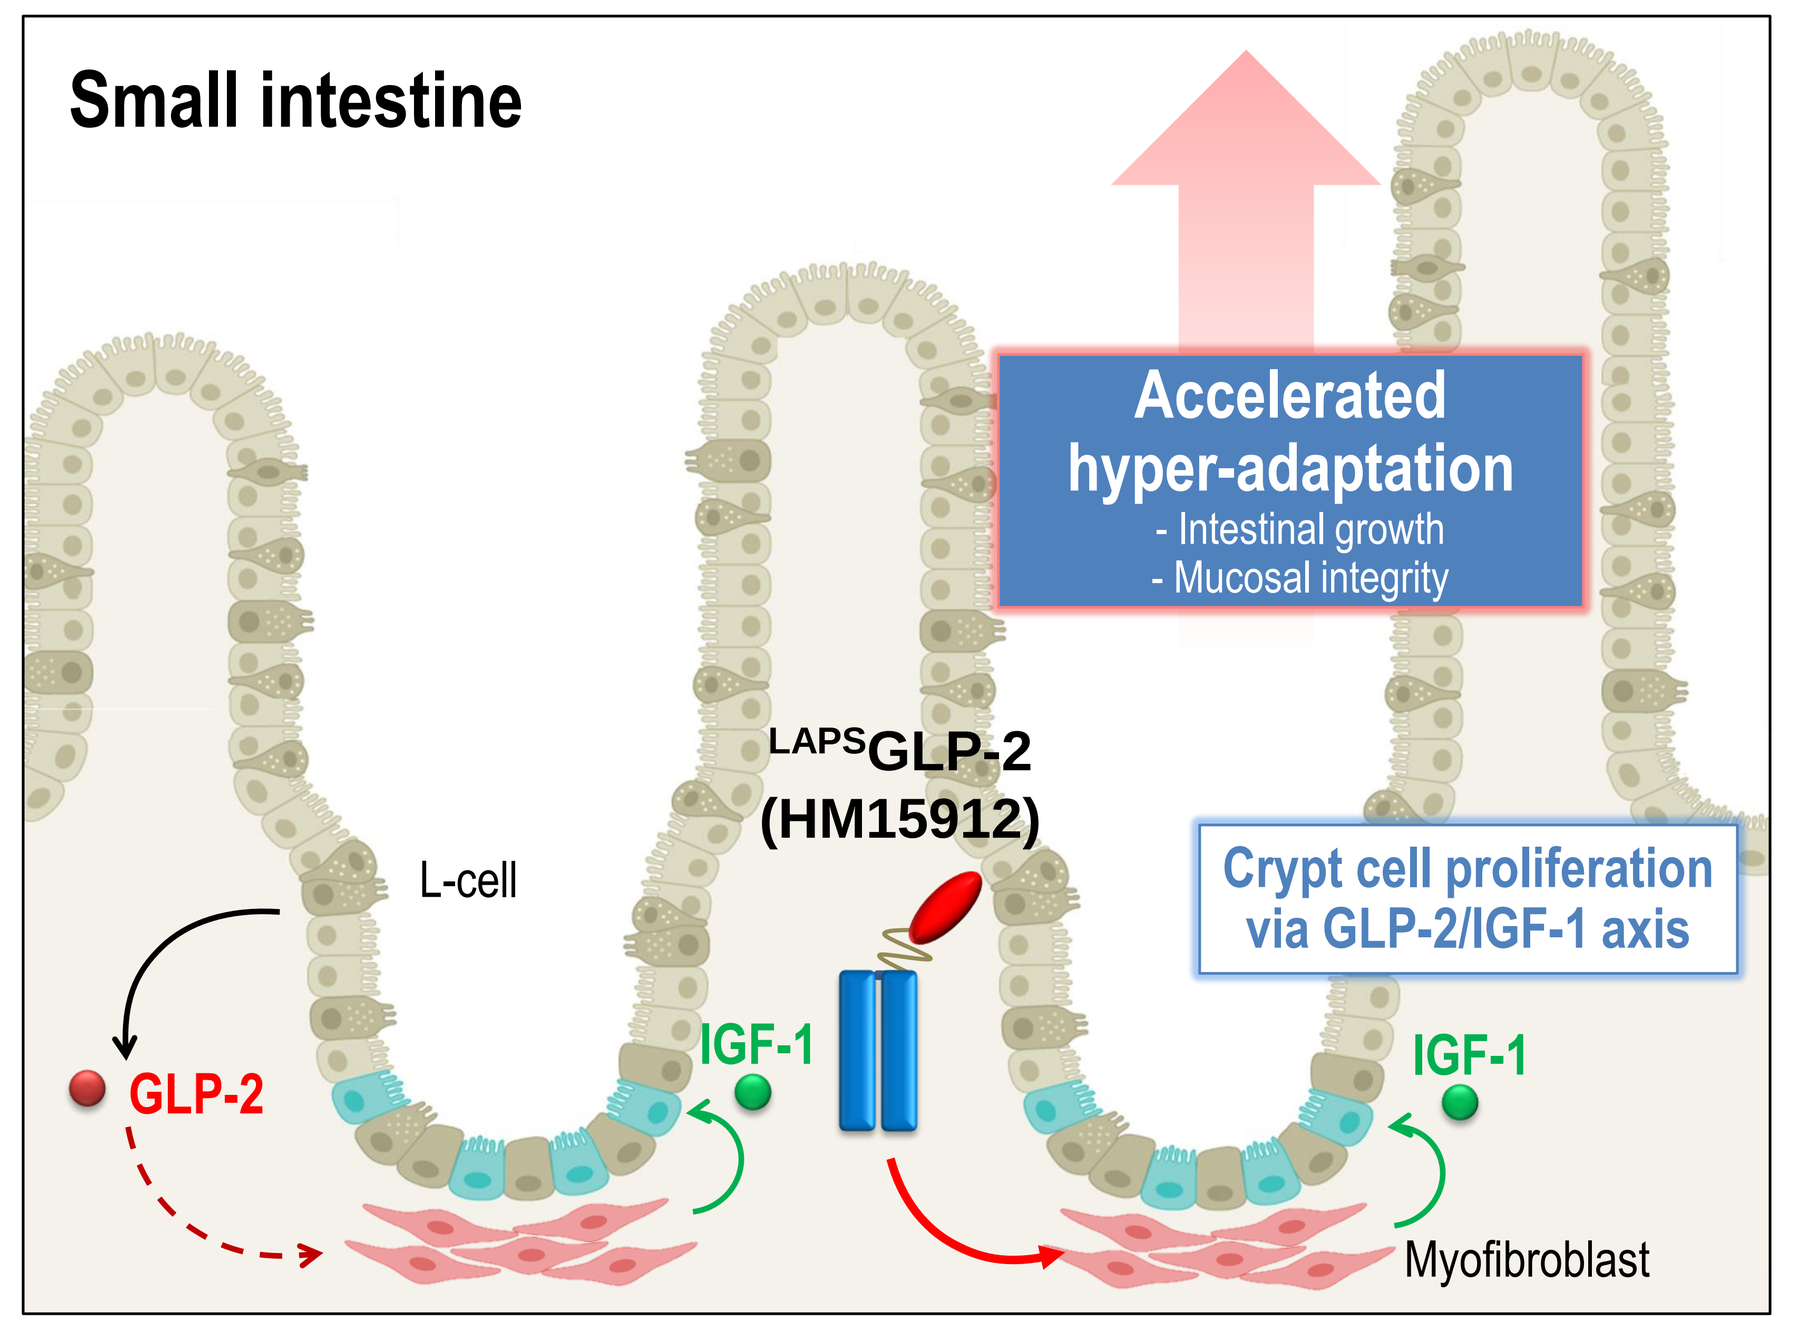
Small intestine
Accelerated hyper-adaptation
- Intestinal growth
- Mucosal integrity
LAPSGLP-2 (HM15912)
L-cell
Crypt cell proliferation via GLP-2/IGF-1 axis
GLP-2
IGF-1
IGF-1
Myofibroblast

HM15912 is conjugated with a human IgG4 Fc *via* flexible PEG linker, and has a same biological mode of action with human GLP-2 via IGF-1

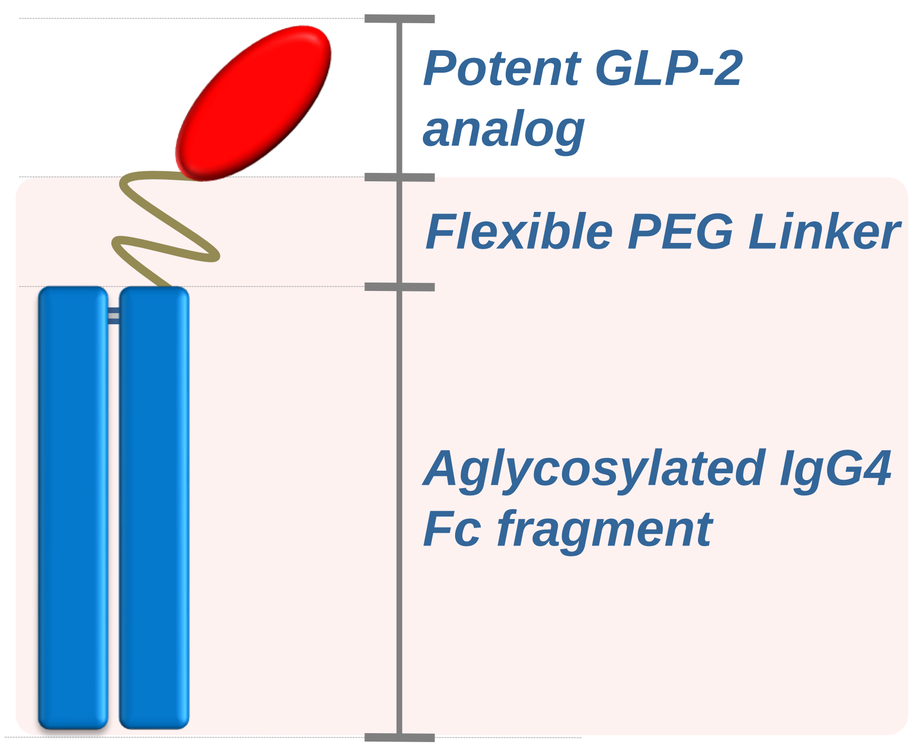
Potent GLP-2 analog
Flexible PEG Linker
Aglycosylated IgG4 Fc fragment

[General profile]

- **Rationally designed GLP-2 analog to have a more potent intestinotrophic action *vs* human GLP-2**
- **Extended half-life allows once-monthly dosing in human**
- **Ready-to-inject with soluble formation**

LAPSCOVERY : Long Acting Peptide/Protein DiSCOVERY Technology

METHODS

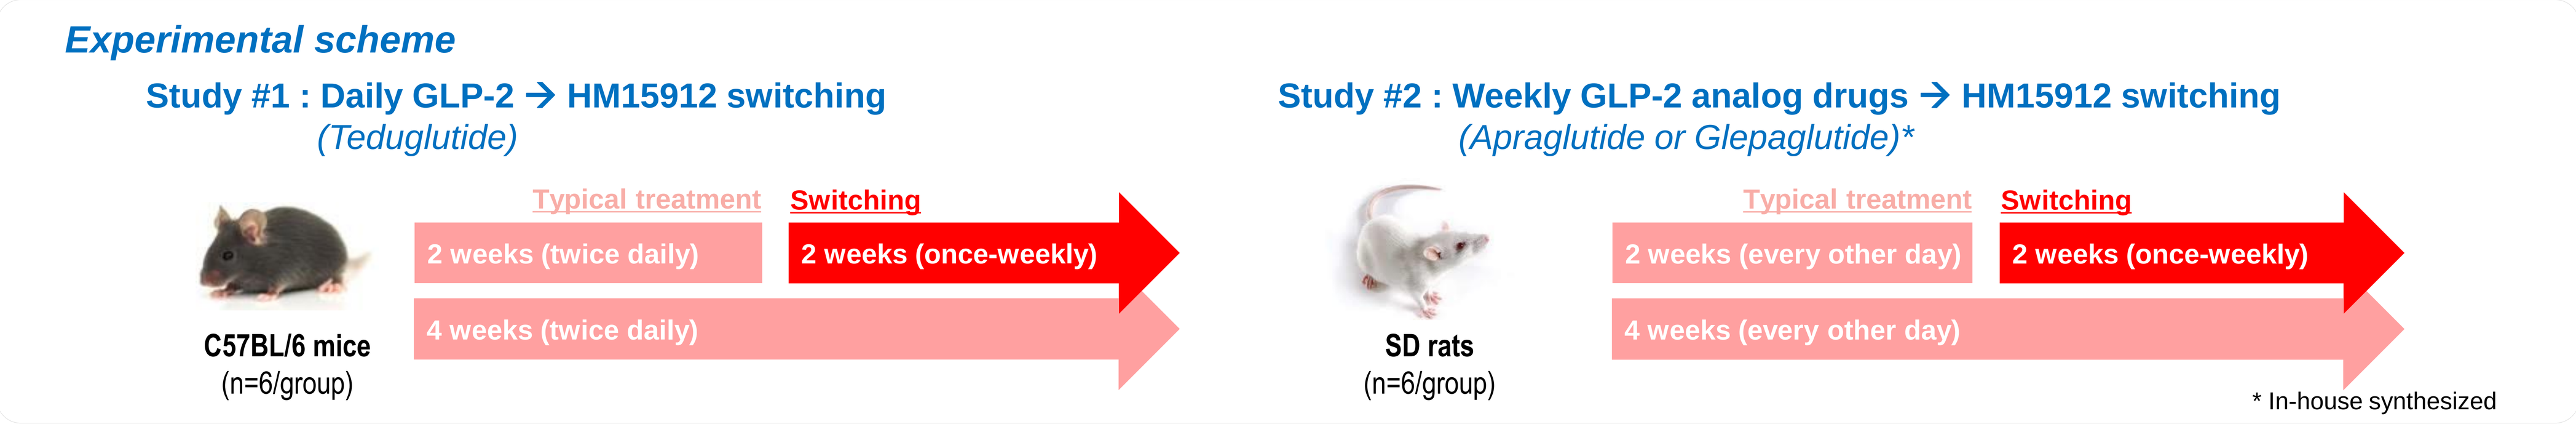
Experimental scheme
Study #1 : Daily GLP-2 → HM15912 switching
(Teduglutide)
Typical treatment
Switching
2 weeks (twice daily)
2 weeks (once-weekly)
4 weeks (twice daily)
C57BL/6 mice
(n=6/group)
Study #2 : Weekly GLP-2 analog drugs → HM15912 switching
(Apraglutide or Glepaglutide)*
Typical treatment
Switching
2 weeks (every other day)
2 weeks (once-weekly)
4 weeks (every other day)
SD rats
(n=6/group)
* In-house synthesized

- In study #1, additional intestinotrophic efficacy after switching from teduglutide to HM15912 was investigated. In study #2, additional intestinotrophic efficacy after switching from weekly GLP-2 analog drugs to HM15912 was investigated. small intestine wet mass was measured at week 2, 3, and 4, and blood D-xylose concentrations were measured to evaluate absorption capacity after oral challenge of D-xylose at the end of study.

RESULTS

Study #1, Beneficial effects after switching from daily GLP-2 analog to HM15912

Fig. 1 Additional intestinotrophic efficacy after switching from teduglutide to HM15912 in C57BL/6 mice

(a) Small intestine mass

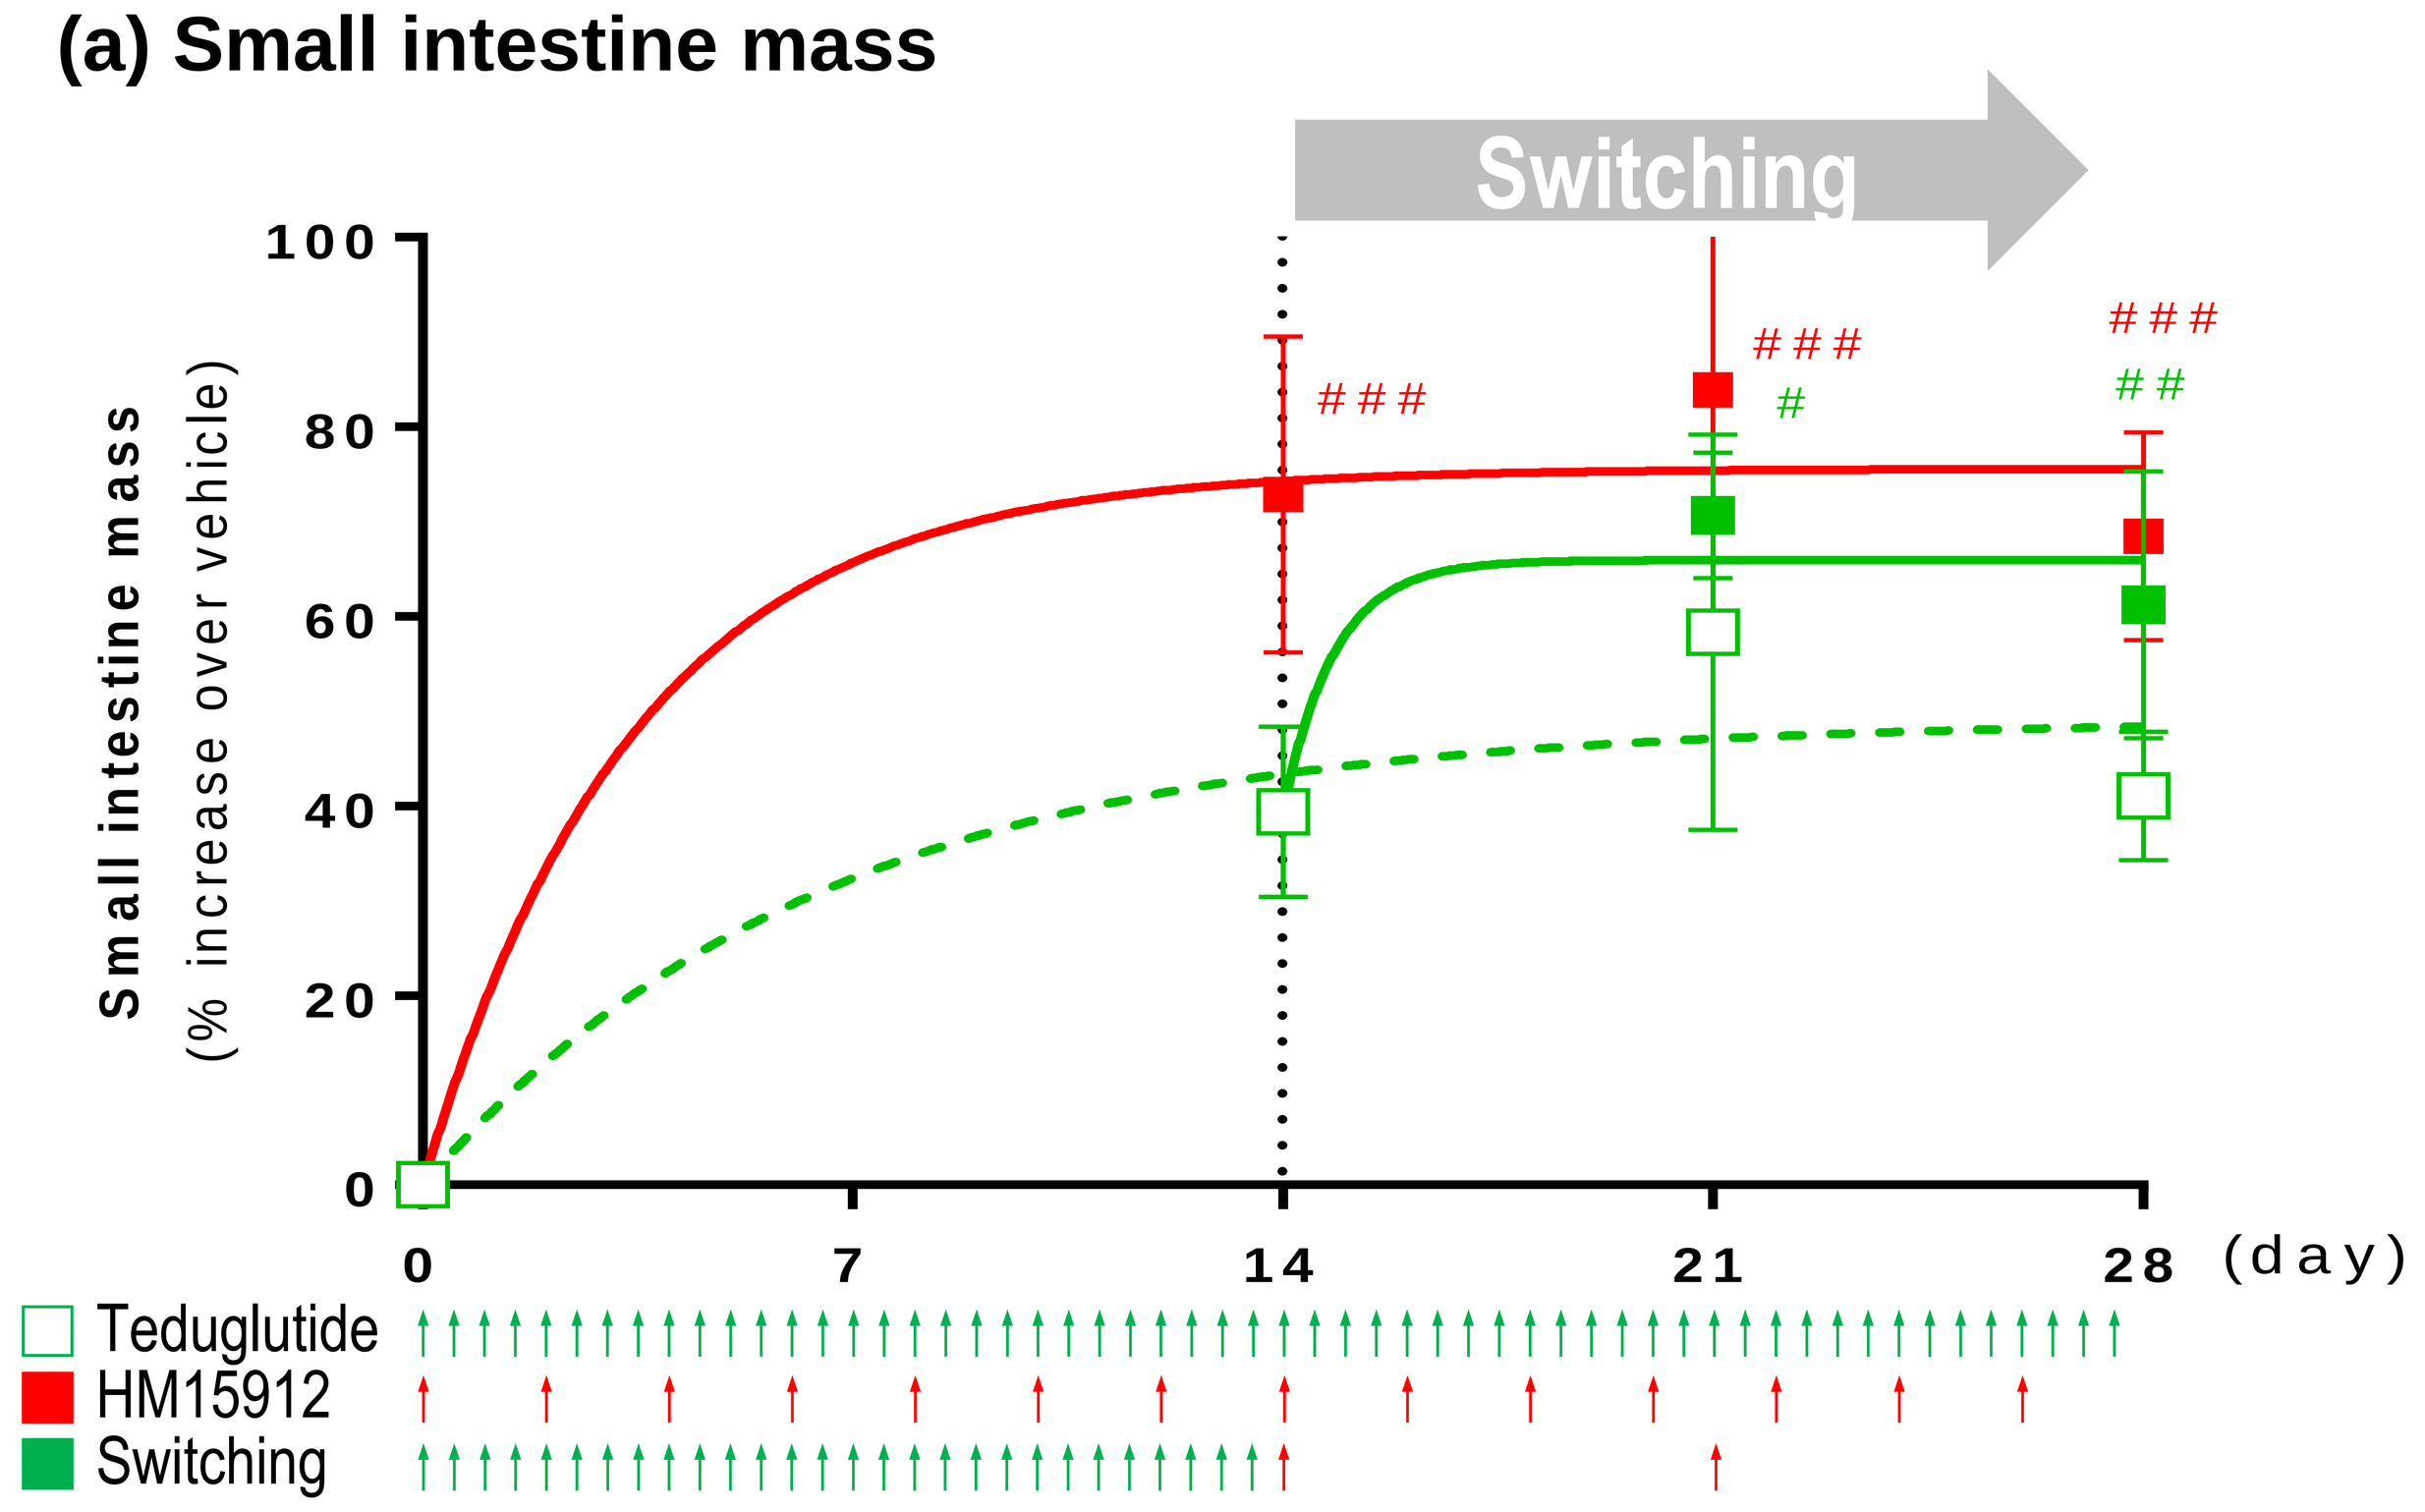
Switching
Small intestine mass
(% increase over vehicle)
100
80
60
40
20
0
0
7
14
21
28
(day)
###
###
#
###
##
Teduglutide
HM15912
Switching

(b) D-xylose absorption at week 4

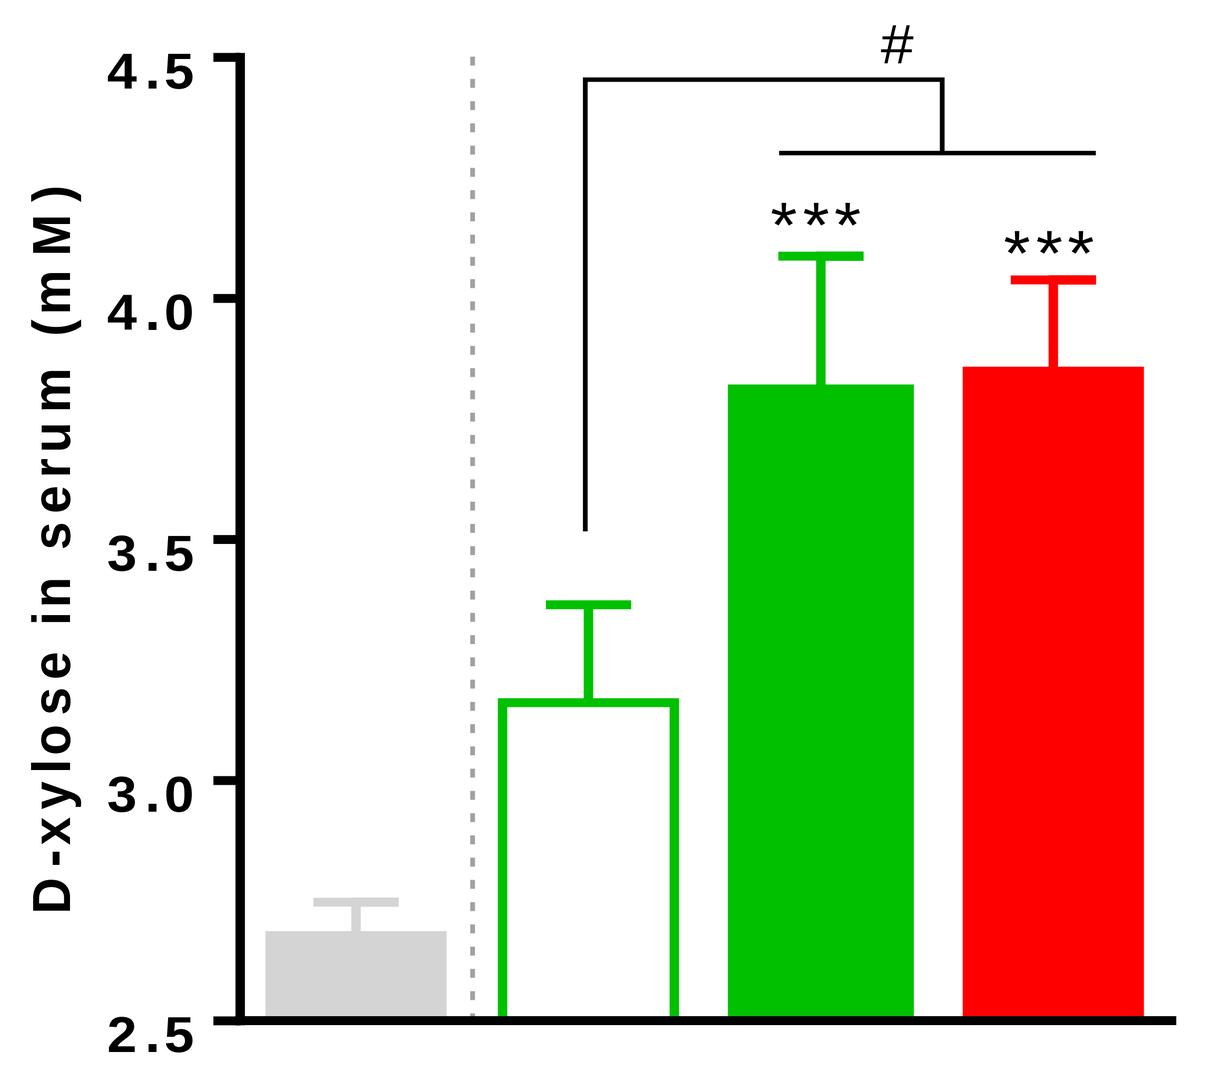
D-xylose in serum (mM)
4.5
4.0
3.5
3.0
2.5
#

Vehicle		
Teduglutide		15.0 nmol/kg/BID **[E_{max}, EPAR]**
Teduglutide → HM1591 2		15.0 nmol/kg/BID → 58.1 nmol/kg/QW **[1.0 mg/kg/month HED]**
HM15912		16.6 nmol/kg/Q2D **[0.25 mg/kg/week HED]**

*p<0.05 *vs.* vehicle by One-way ANOVA
#p<0.05 *vs.* Teduglutide by One-way ANOVA

➢HM15912 treatment significantly increased small intestine mass compared to teduglutide treated group after 2 weeks. After 2 more weeks treatment, while showing the maintained small intestine increment in teduglutide treated group, small intestine mass was further increased after switching to HM15912. In consensus with small intestine mass increment, absorption capacity was also significantly increased after switching to once weekly administration of HM15912.

Study #2, Beneficial effects after switching from weekly GLP-2 analog drugs to HM15912

Fig. 2 Additional intestinotrophic efficacy after switching from apraglutide and glepaglutide to HM15912 in SD rats

(a) Small intestine mass

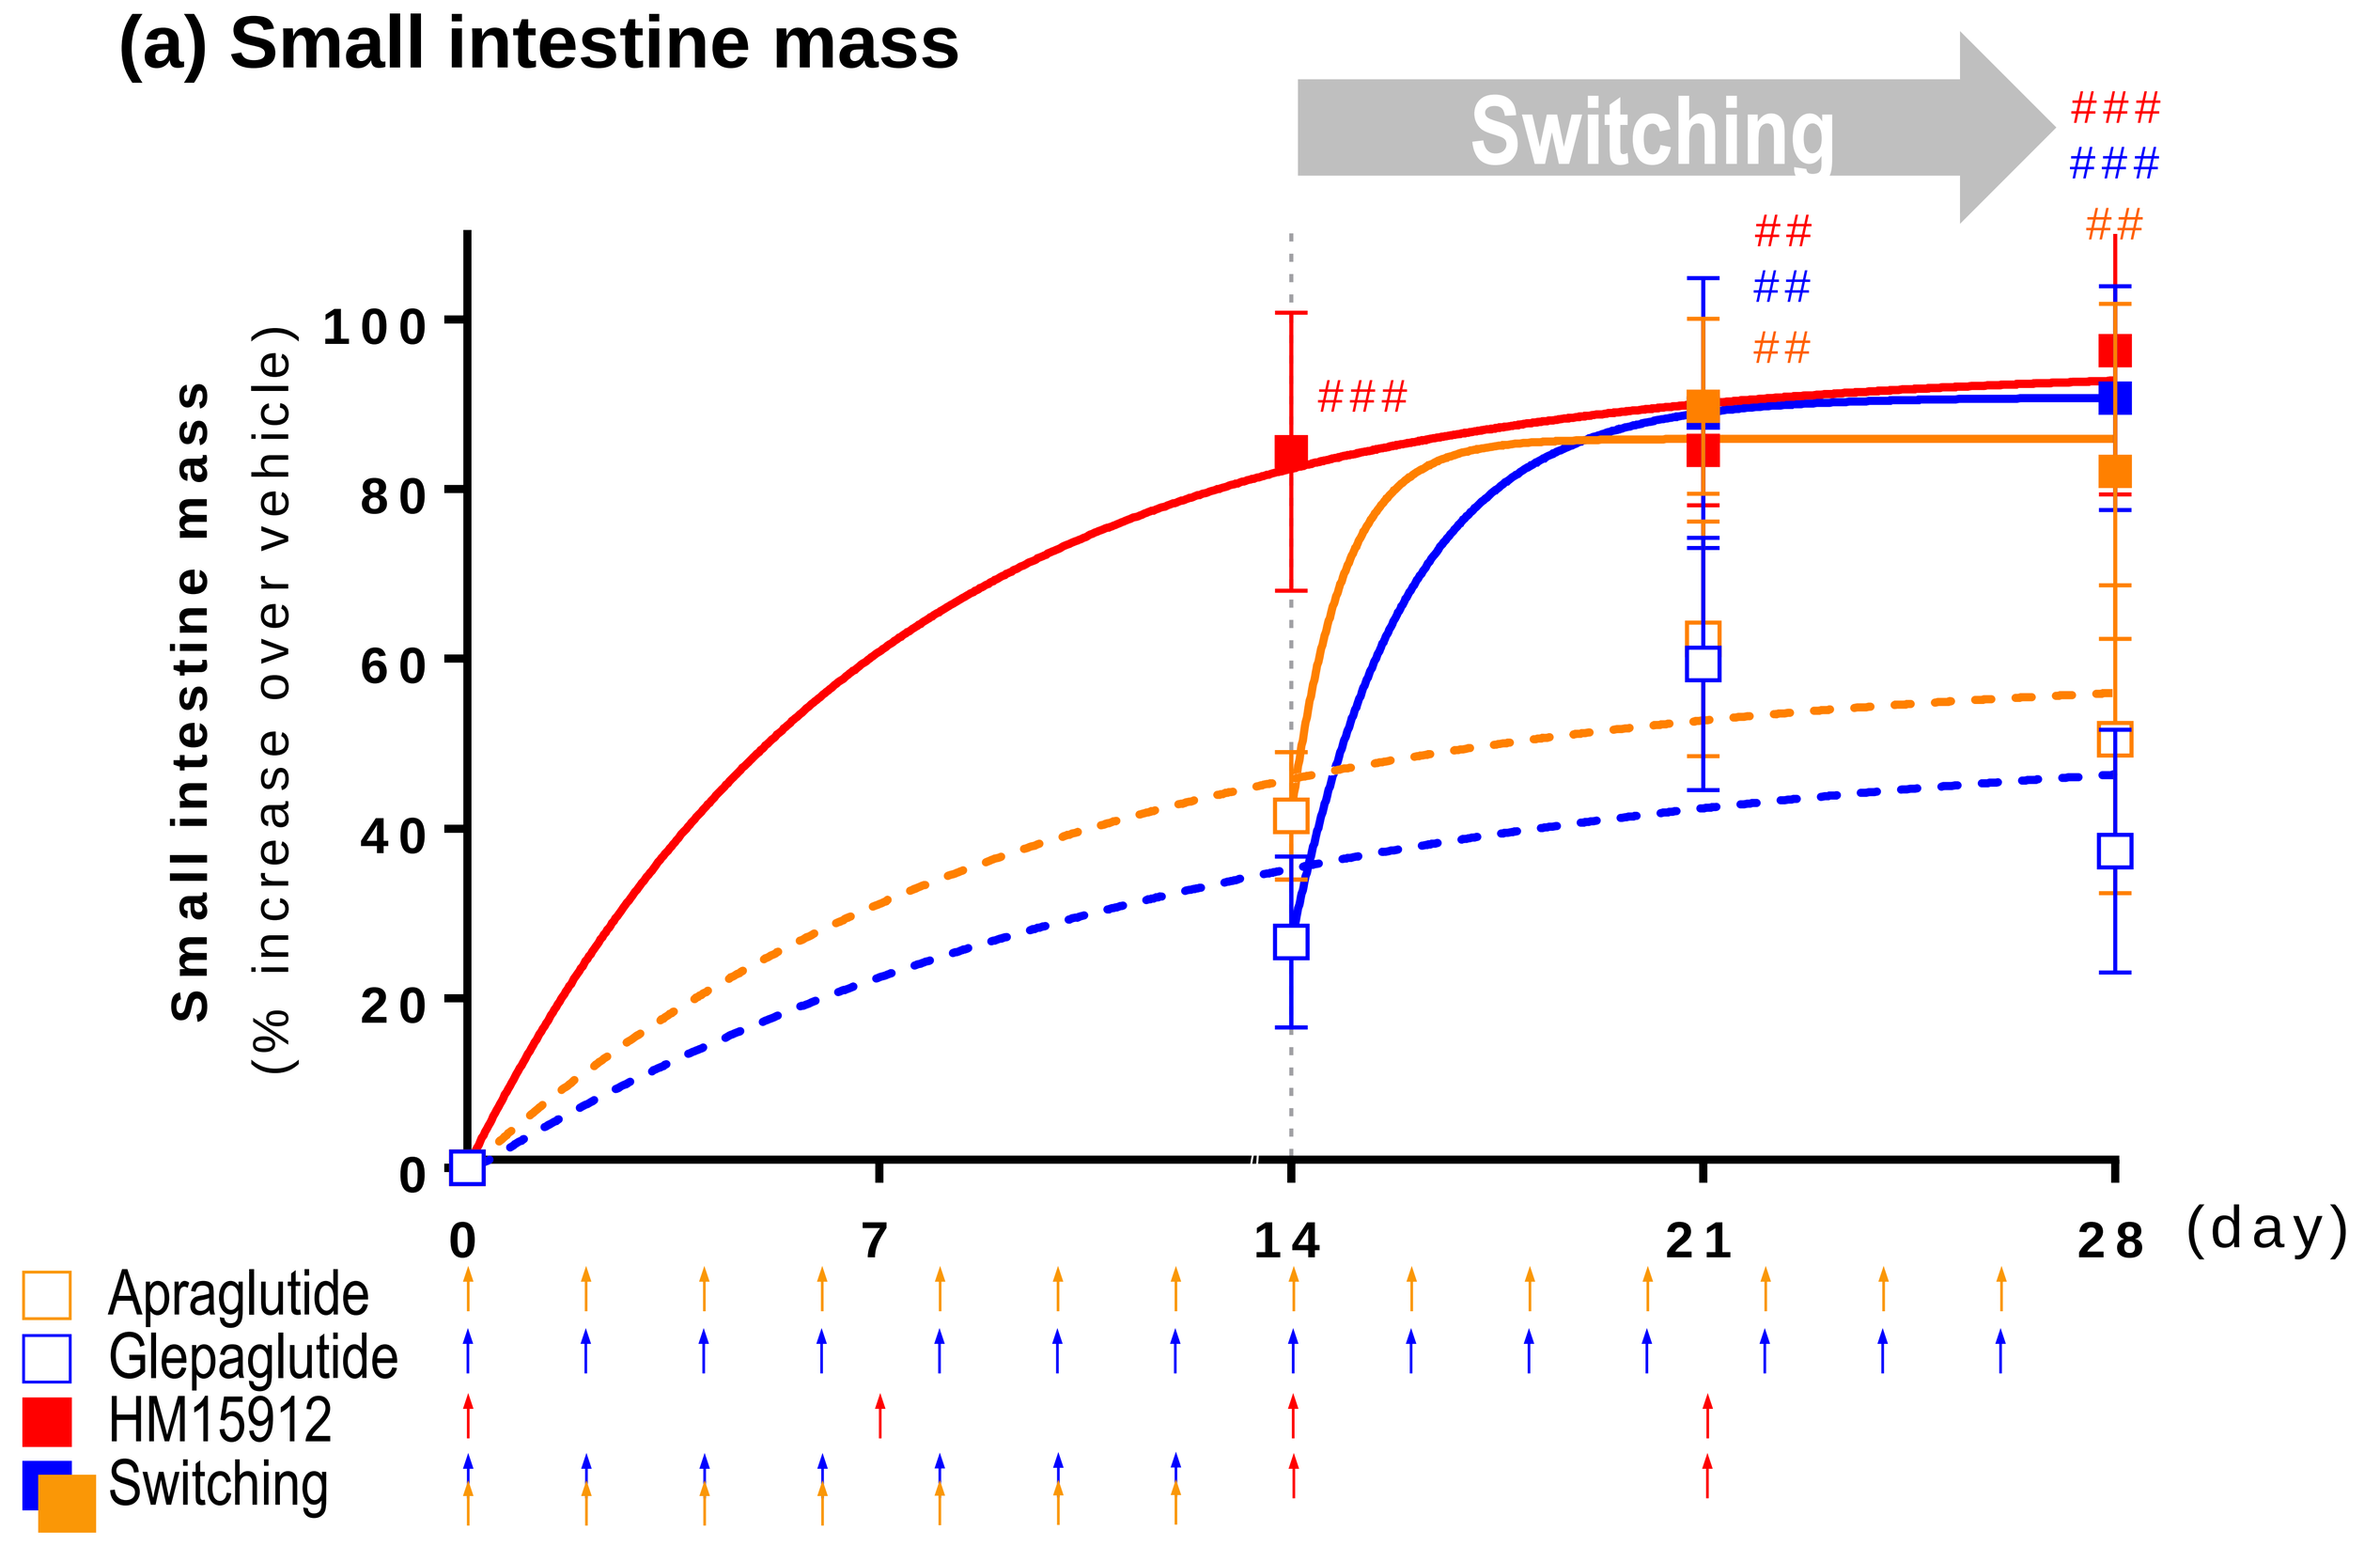
Switching
Small intestine mass
(% increase over vehicle)
100
80
60
40
20
0
0
7
14
21
28
(day)
###
##
##
##
###
###
##
Apraglutide
Glepaglutide
HM15912
Switching

(b) D-xylose absorption at week 4

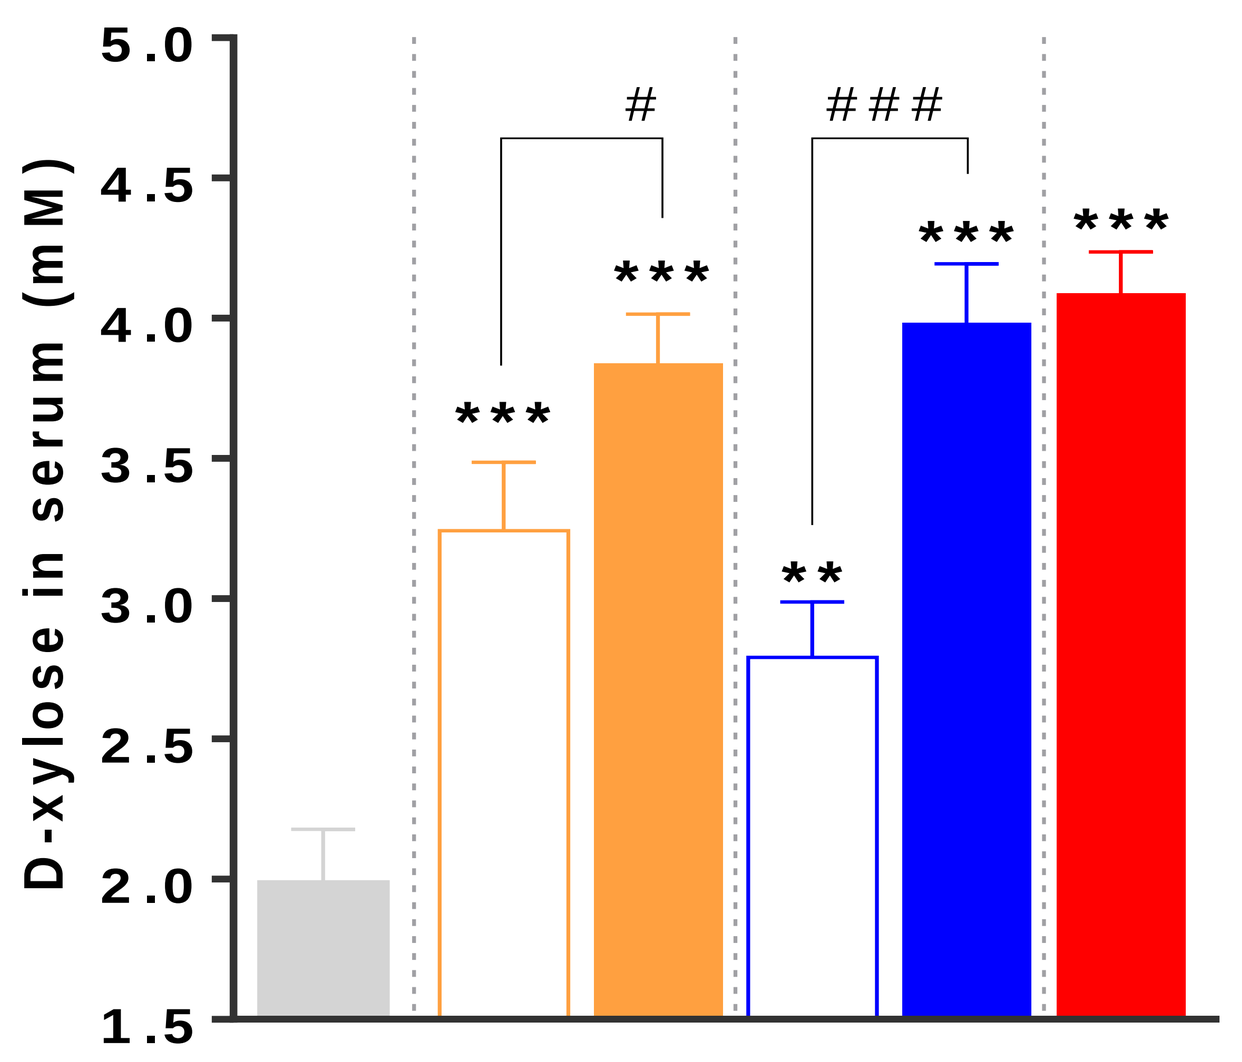
D-xylose in serum (mM)
5.0
4.5
4.0
3.5
3.0
2.5
2.0
1.5
#
###

**

Vehicle	
Apraglutide (Synthesized)	63.7 nmol/kg/Q2D **[10 mg/week HED]**
Glepaglutide (Synthesized)	55.6 nmol/kg/Q2D **[10 mg/week HED]**
Apraglutide (Synthesized) → HM15912	63.7 nmol/kg/Q2D → 57.9 nmol/kg/QW
Glepaglutide (Synthesized) → HM15912	55.6 nmol/kg/Q2D → 57.9 nmol/kg/QW
HM15912	57.9 nmol/kg/QW **[2 mg/kg/month HED]**

*p<0.05 *vs.* vehicle by One-way ANOVA
#p<0.05 *vs.* weekly GLP-2RA (synthesized) by One-way ANOVA

➢HM15912 treatment significantly increased small intestine mass compared to weekly GLP-2 analog drug treated groups after 2 weeks. After 2 more weeks treatment, while showing the slight increment in small intestine mass in weekly GLP-2 analogs, small intestine mass was further increased after switching to HM15912. In consensus with small intestine mass increment, absorption capacity was also significantly increased after switching to once weekly administration of HM15912.

CONCLUSIONS

- **After switching from existing GLP-2 analogs (daily or weekly GLP-2 analogs), HM15912 further increased mucosal growth as well as nutrition absorption capacity in small intestine even though less frequent administration.**
- **HM15912 will provide benefits for less PN dependency and more convenient treatment option to SBS patients, who are still suffered from parenteral support with typical GLP-2 medication, such as teduglutide or weekly GLP-2 analog drugs in clinical development.**

ENDO 2023 Conference June 15 - 18, 2023

ENDOCRINE SOCIETY

Hanmi Pharm. Co., Ltd.