Effect of a novel long-acting GLP-1/GIP/glucagon triple agonist (HM15211) in a NASH and liver fibrosis animal models

Hanmi

1100-P

Jung Kuk Kim[1], Jong Suk Lee[1], Dae Jin Kim[1], Eun Jin Park[1], Aram Lee[1] , Young Hoon Kim[1], and In Young Choi[1]
[1]Hanmi Pharm. Co., Ltd, Seoul, South Korea

BACKGROUND

Modulation of multiple aspects of NASH and liver fibrosis by HM15211 in comparison to the action of other drug candidates for NASH

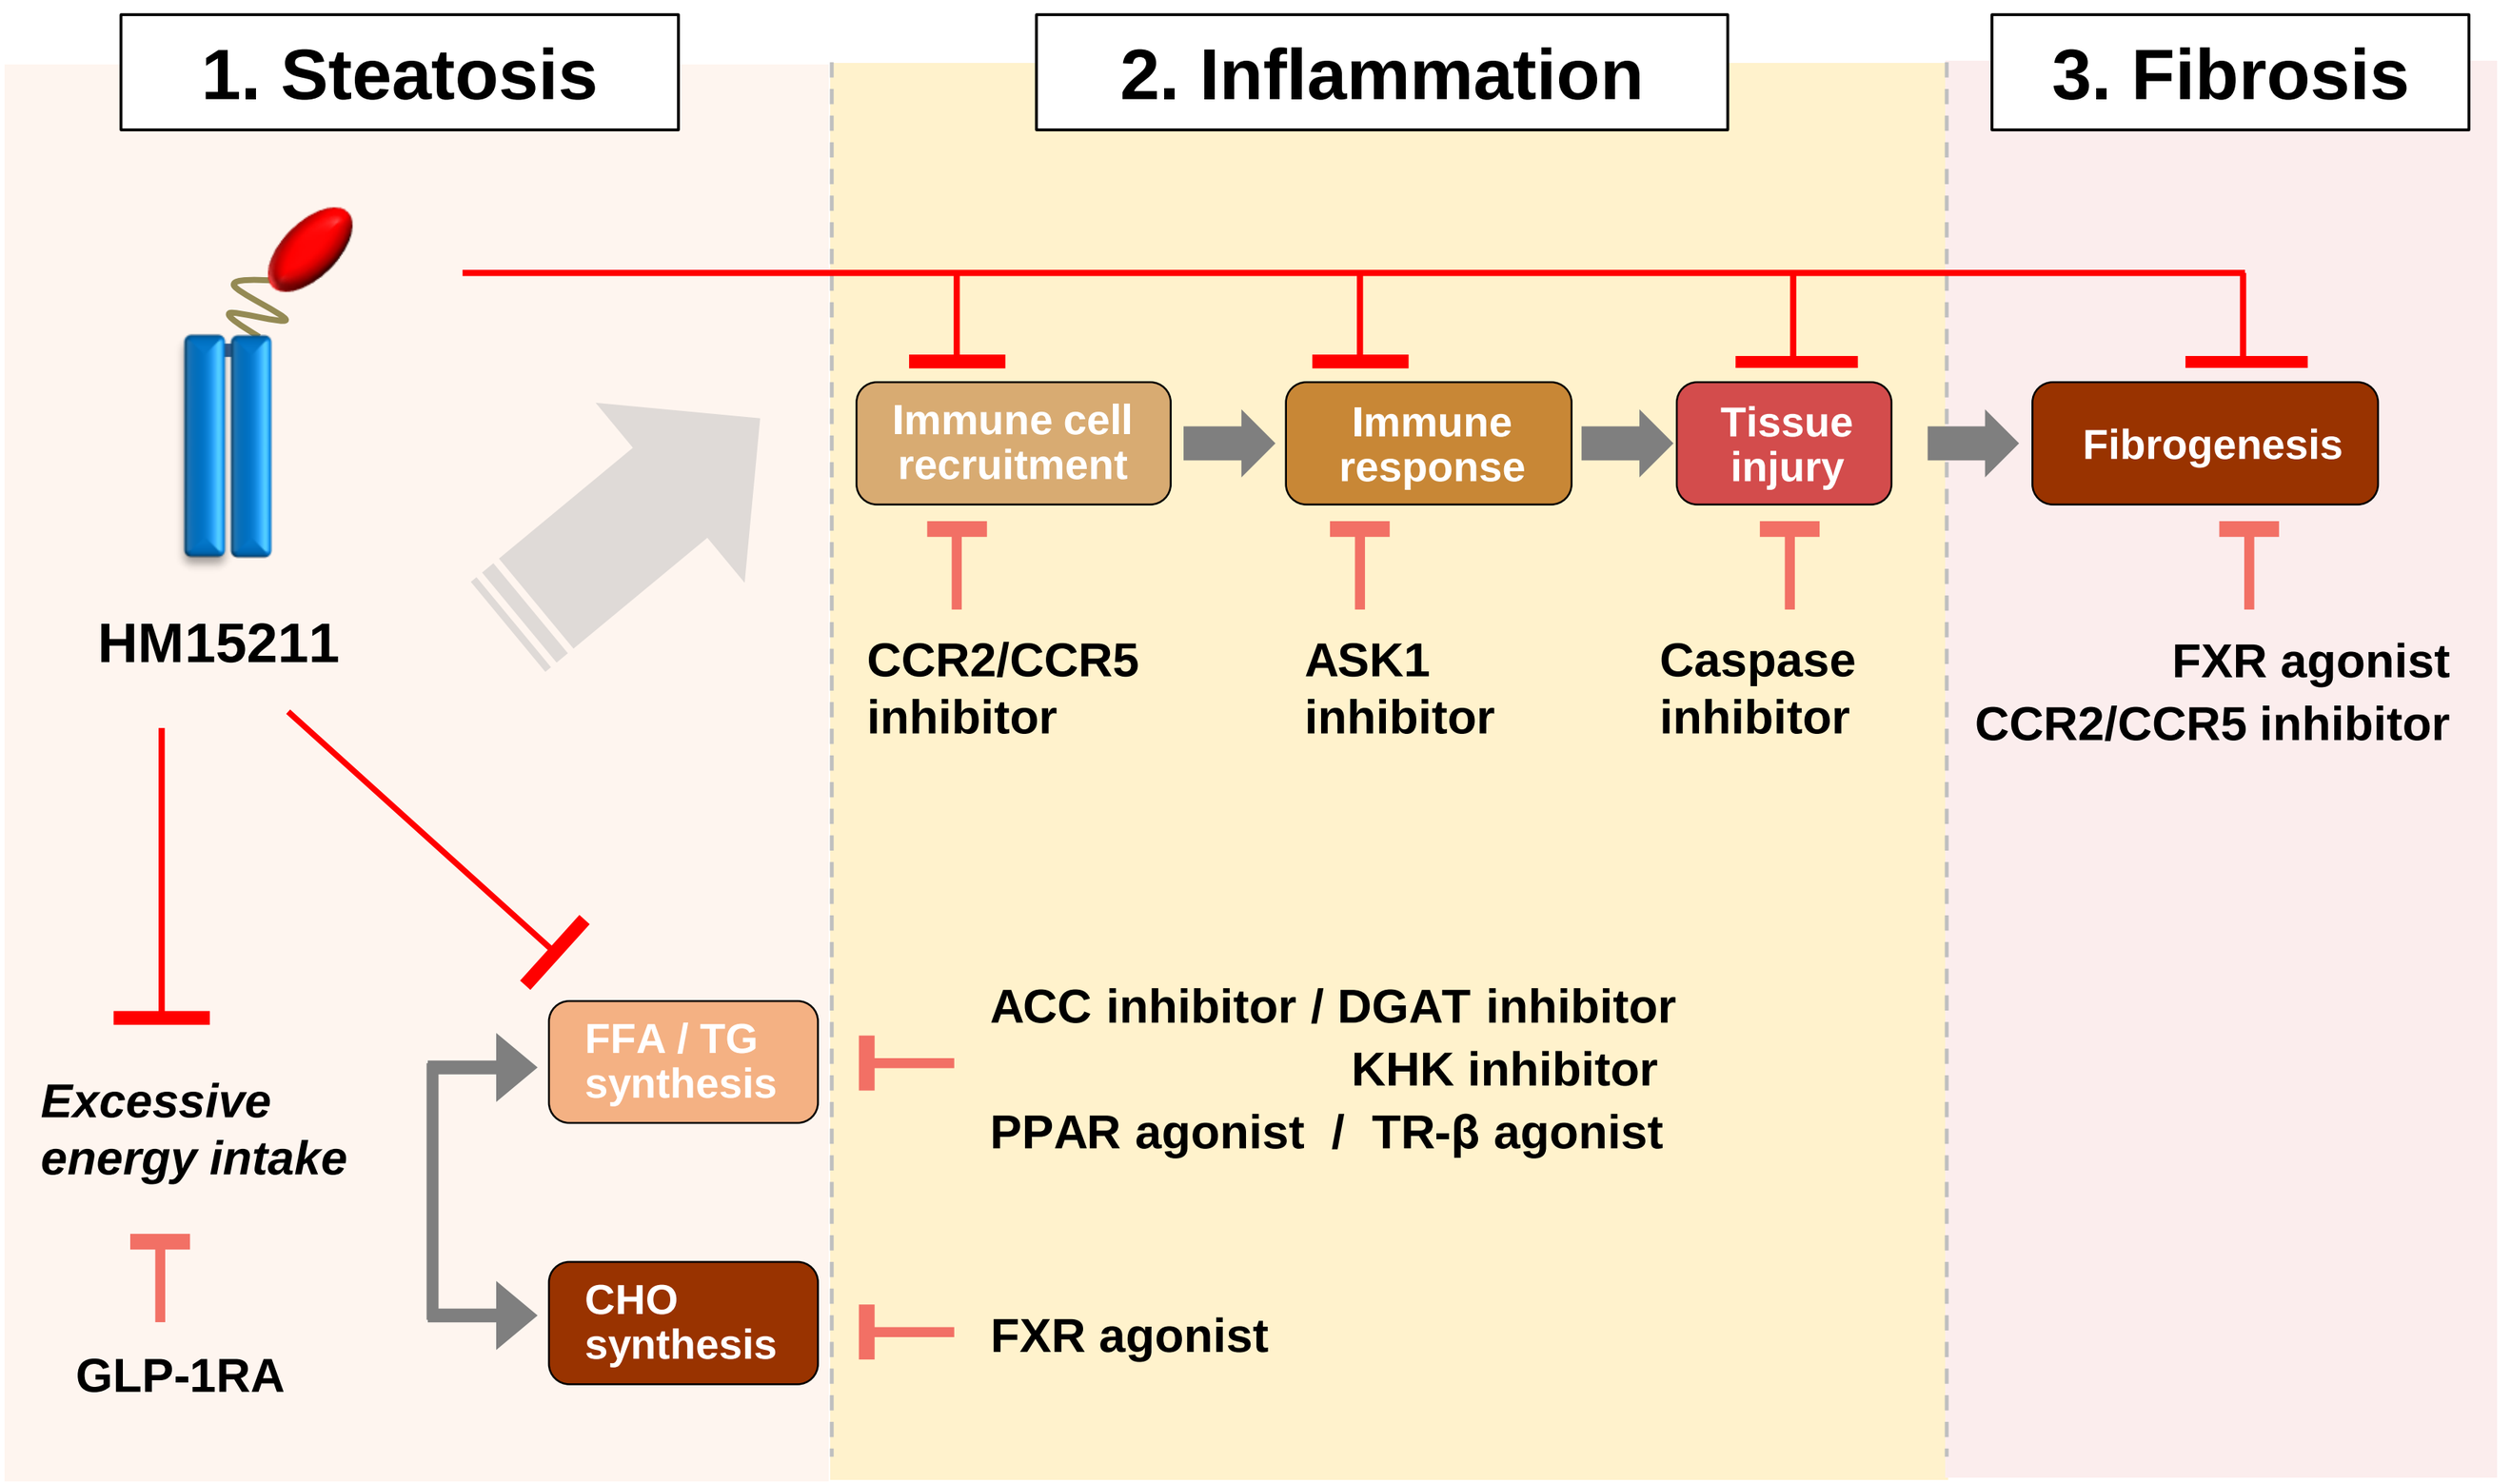
1. Steatosis
2. Inflammation
3. Fibrosis
HM15211
Immune cell recruitment
Immune response
Tissue injury
Fibrogenesis
CCR2/CCR5 inhibitor
ASK1 inhibitor
Caspase inhibitor
FXR agonist
CCR2/CCR5 inhibitor
Excessive energy intake
GLP-1RA
FFA / TG synthesis
CHO synthesis
ACC inhibitor / DGAT inhibitor
KHK inhibitor
PPAR agonist / TR-β agonist
FXR agonist

AIMS

- **Since multiple biological pathways are involved in the disease progression, therapeutic approach simultaneously targeting these pathways might be required to effectively treat NASH and fibrosis**
- **To address this, HM15211, a novel long-acting GLP-1/GIP/Glucagon triple agonist, has been developed**
- **In this study, we evaluated the therapeutic potential of HM15211 in NASH and fibrosis animal models**

METHODS

- Therapeutic potential of HM15211 in NASH and fibrosis was evaluated in MCD-diet induced NASH mice (6 ~ 12 weeks induction). After 4 ~ 5 weeks treatment of HM15211, liver tissue samples were prepared to measure hepatic TG, TBARS, NASH/fibrosis-related marker gene expression (TNF-α, TGF-β, α-SMA, and Collagen-1α1). Blood liver function markers (ALT, bilirubin) were also determined
- To investigate the therapeutic effects of HM15211 in more human relevant disease model, biopsy-proven obese, NASH, and fibrosis monkeys (BMI >40 kg/m^2, NAS + fibrosis score > 7) were utilized. After 12 weeks treatment of HM15211 including 3 weeks titration period, body weight and blood lipid profiles were determined, and body composition was determined by DEXA. Liver biopsy samples were subjected to histologic analysis.
- To determine NAS (NAFLD activity score), the same region of each liver tissue was subjected to H&E staining. For fibrosis analysis, Sirius red staining and hepatic hydroxyproline analysis were performed

RESULTS

Steatosis and inflammation improvement in MCD mice

Figure 1. Effect of HM15211 on steatosis in MCD-diet mice (n=7)

(a) Hepatic TG **(b) Hepatic TBARS§**

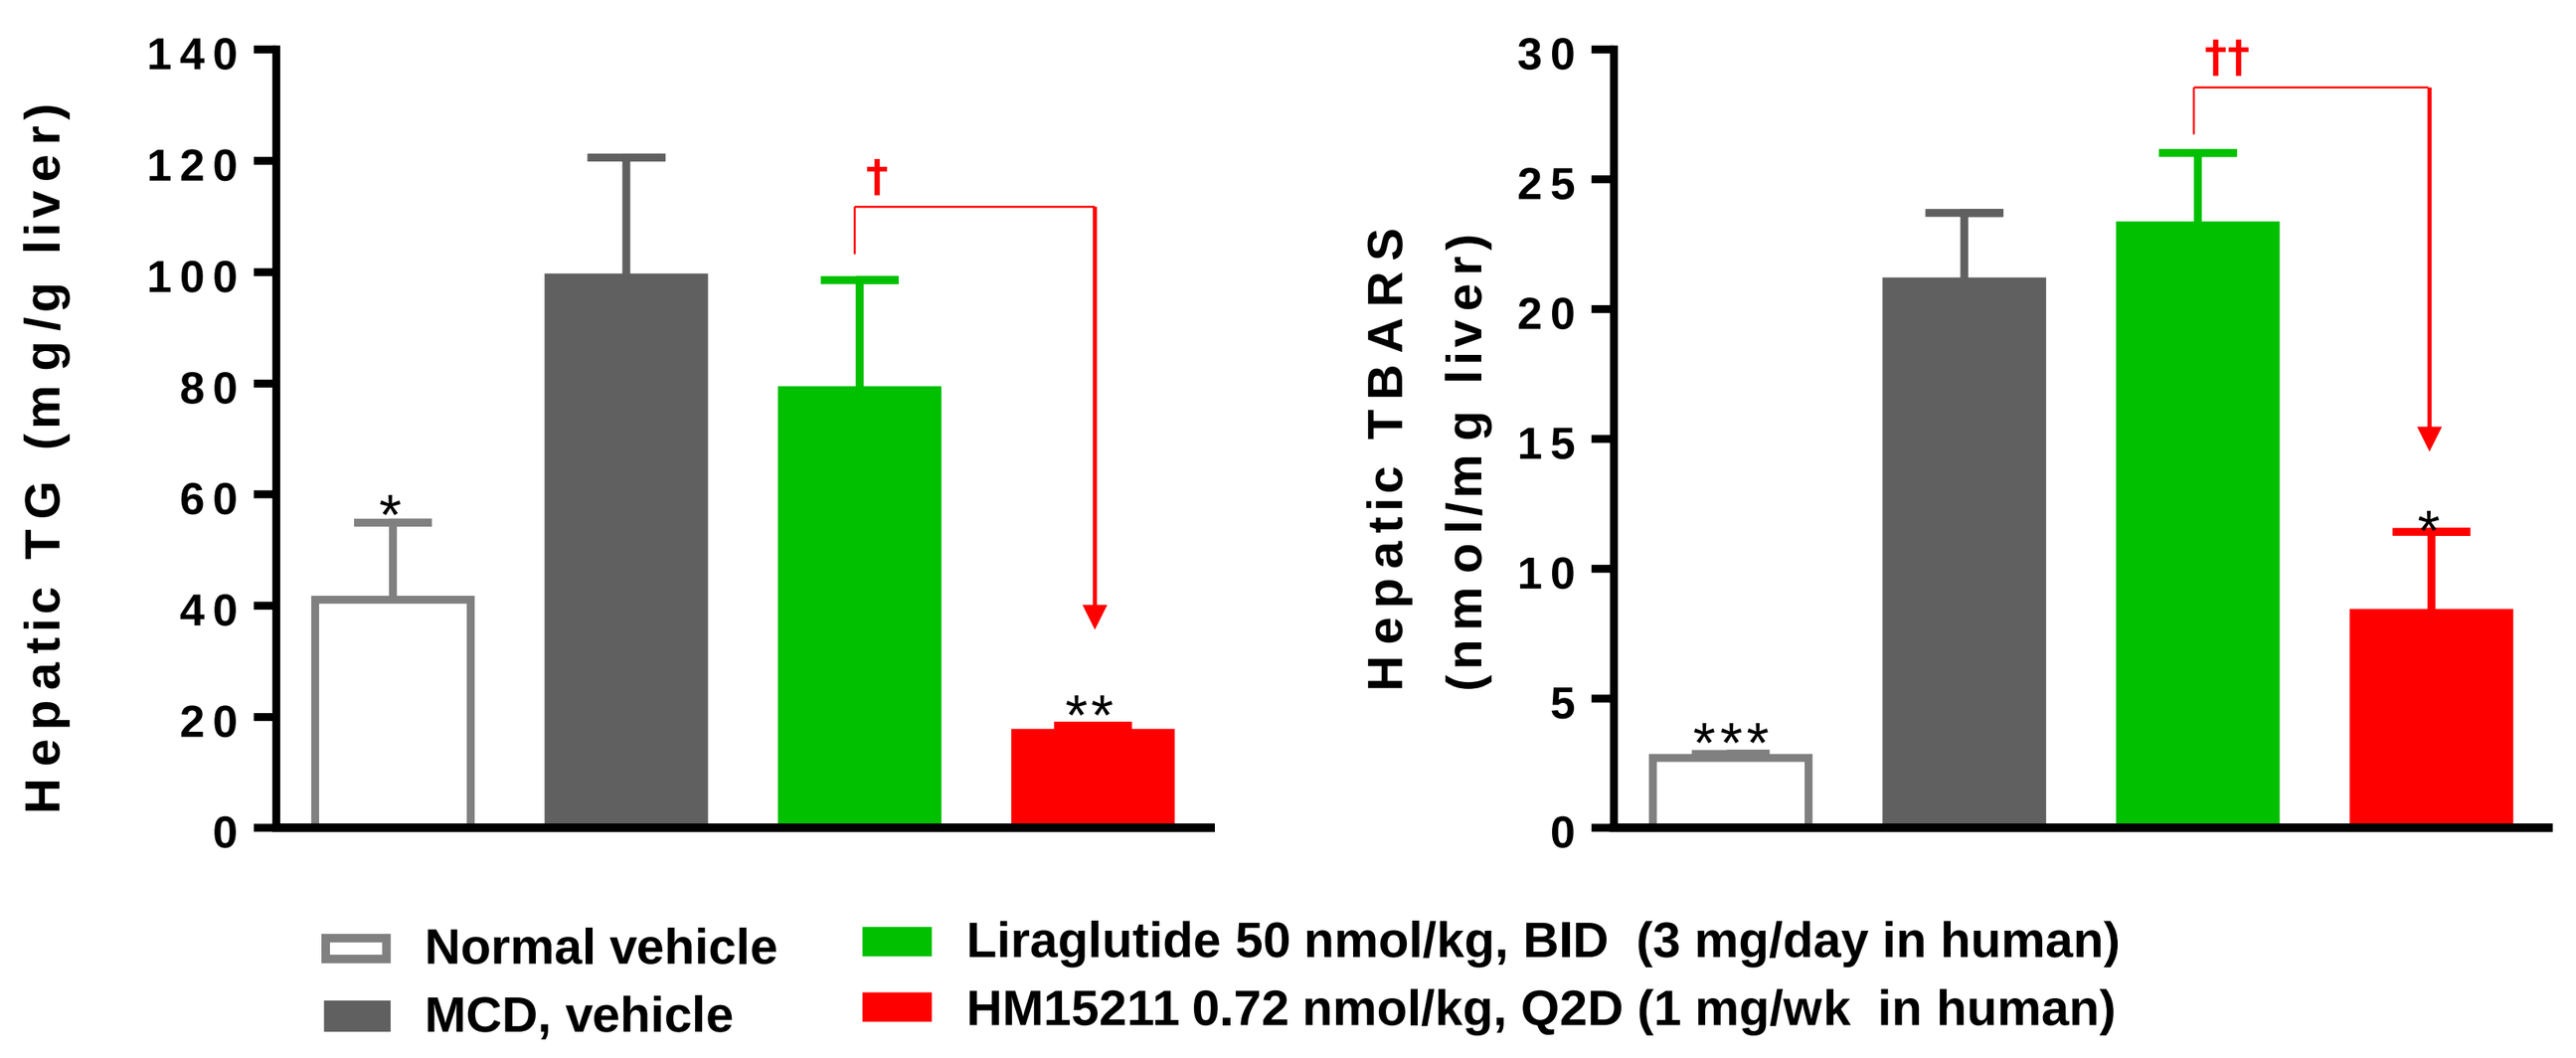
Hepatic TG (mg/g liver)
Hepatic TBARS (nmol/mg liver)
Normal vehicle
MCD, vehicle
Liraglutide 50 nmol/kg, BID (3 mg/day in human)
HM15211 0.72 nmol/kg, Q2D (1 mg/wk in human)

* ~ ***p<0.05 ~ 0.001 *vs.* MCD mice, vehicle by One-way ANOVA, †~†† p<0.05 ~ 0.01 *vs.* liraglutide by One-way ANOVA
§ TBARS: Thiobarbituric acid reactive substances, oxidative stress marker

➢ HM15211 significantly reduced liver TG and TBARS independent of BWL (*data not shown*) in MCD-diet mice, suggesting its direct liver effect on steatosis improvement

Figure 2. Effect of HM15211 on NASH prognosis and inflammation marker expression in MCD-diet mice (n=7)

(a) Blood ALT **(b) Blood total bilirubin**

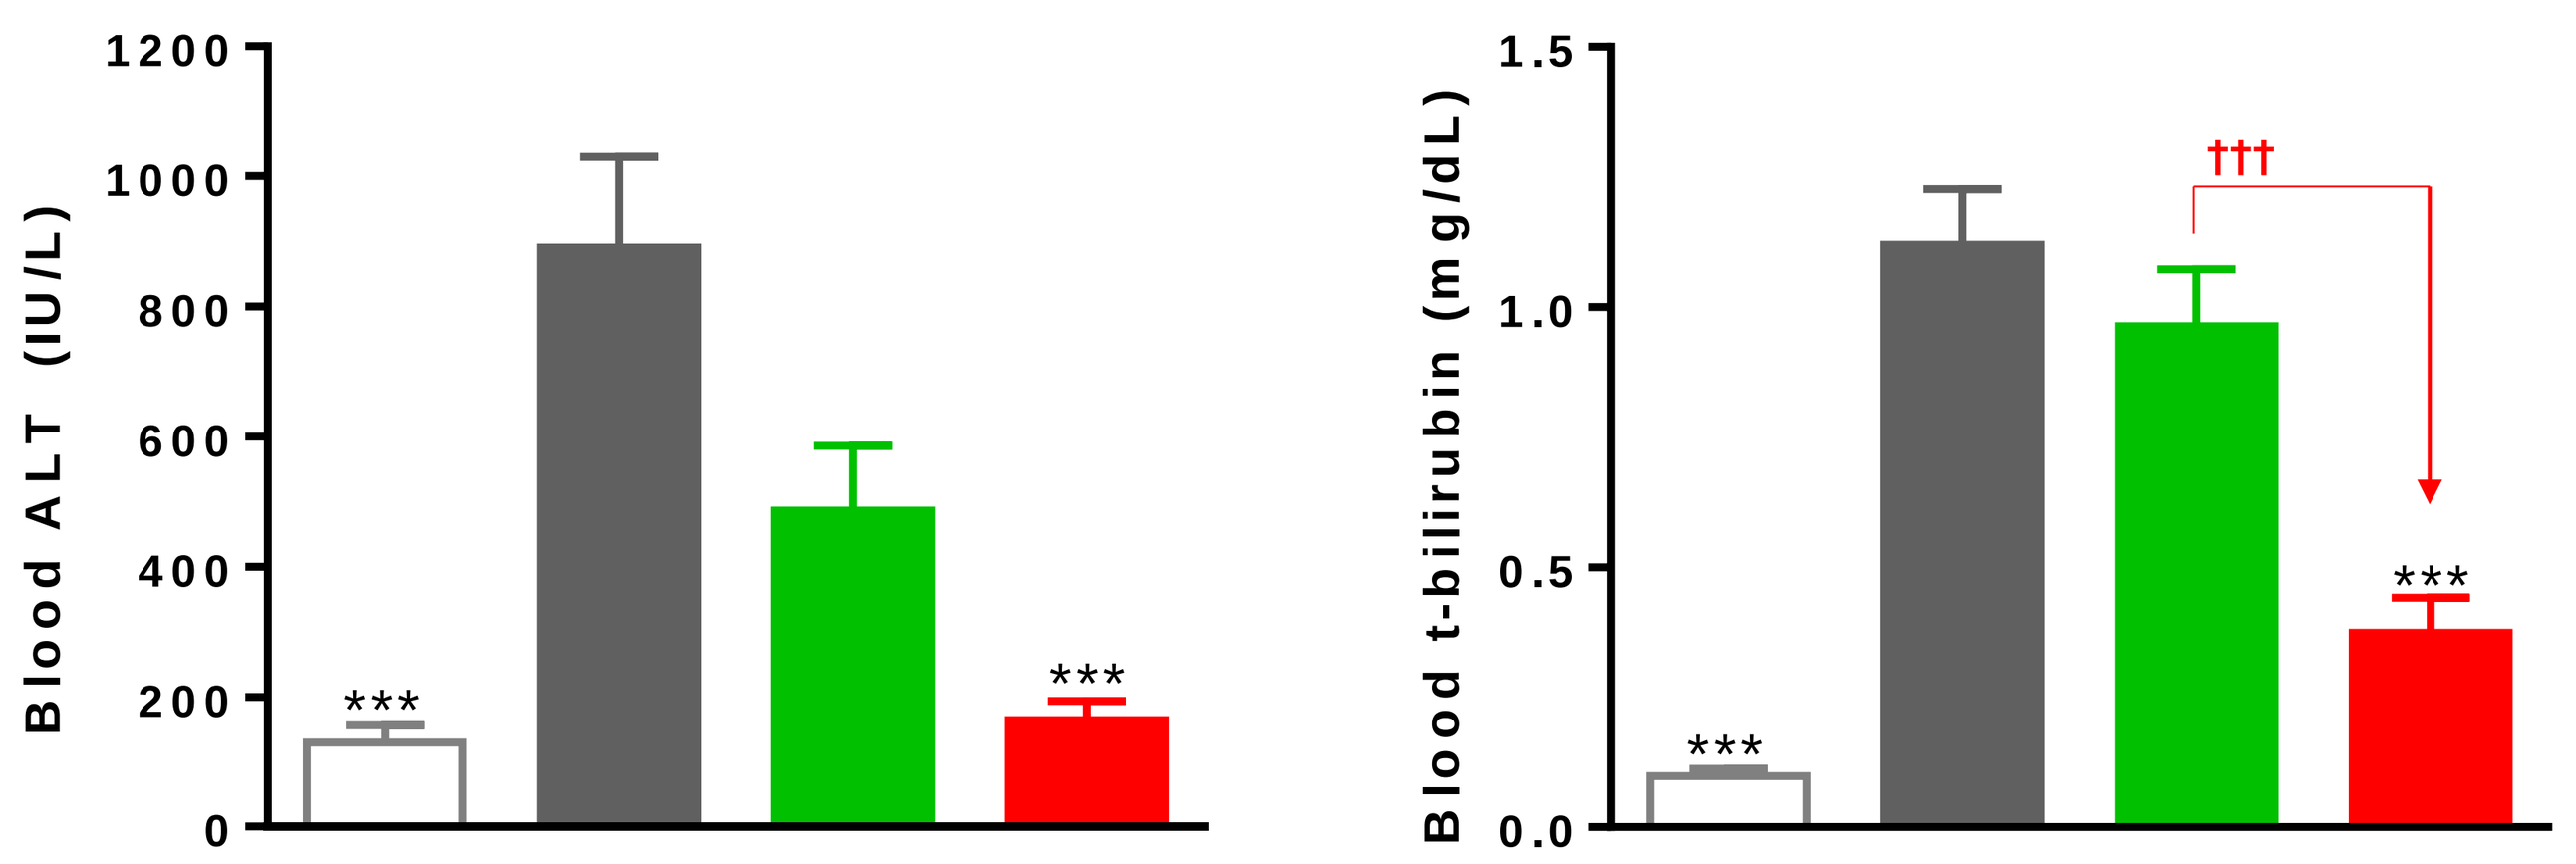
Blood ALT (IU/L)
Blood t-bilirubin (mg/dL)

(c) Inflammation and HSC activation marker gene expression

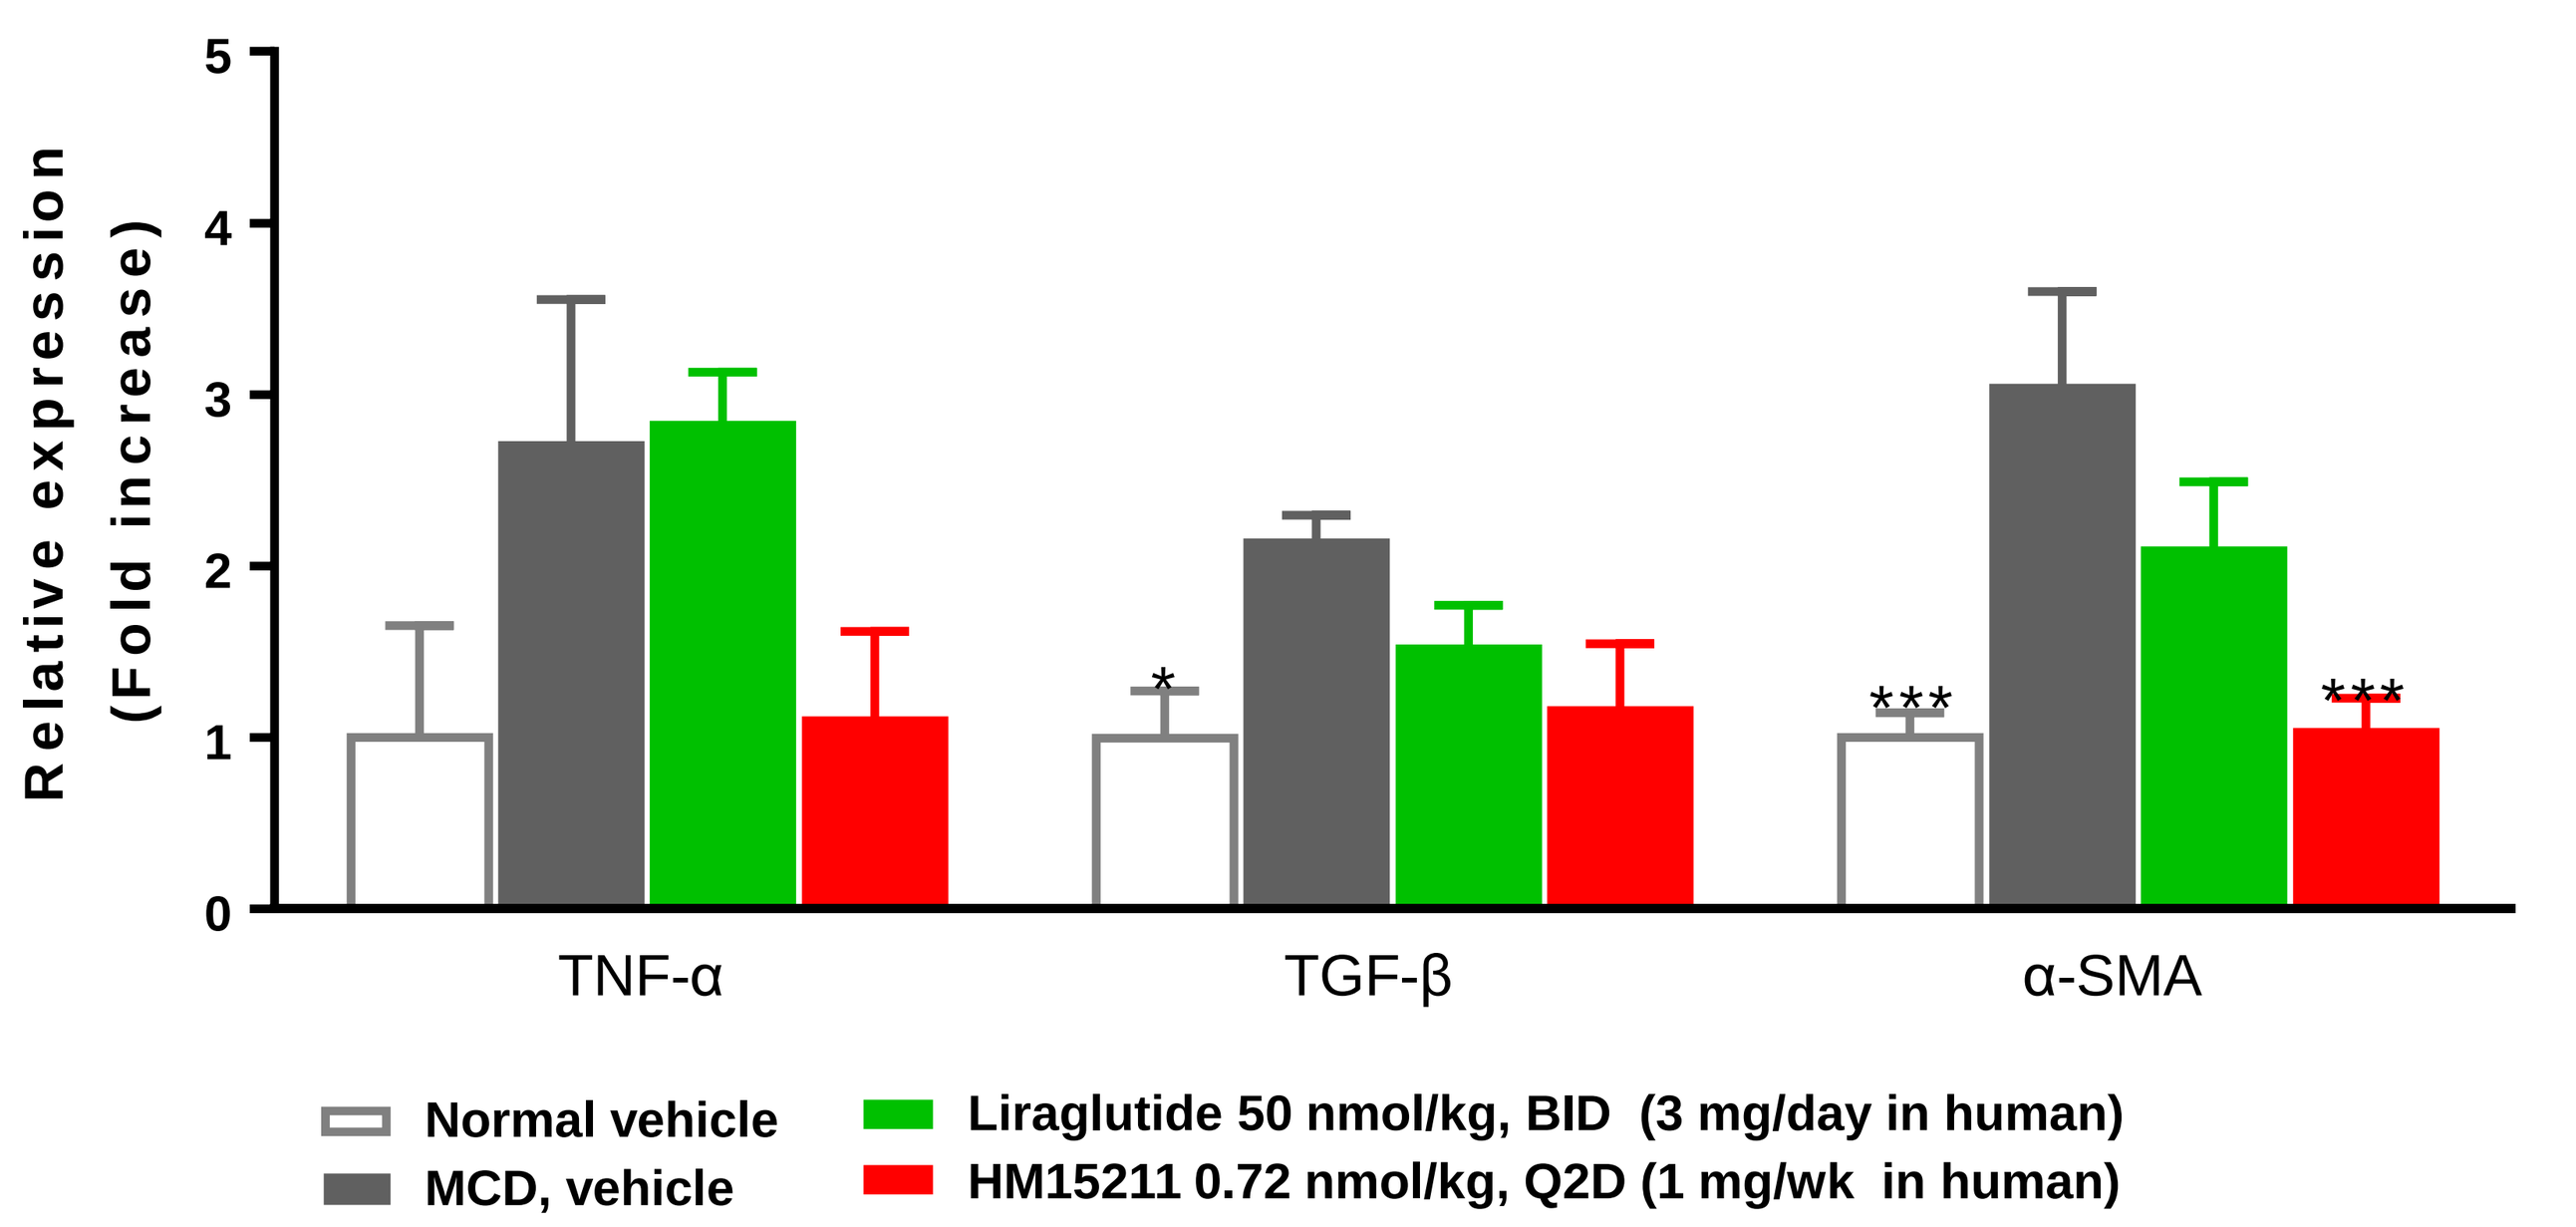
Relative expression (Fold increase)
TNF-α
TGF-β
α-SMA
Normal vehicle
MCD, vehicle
Liraglutide 50 nmol/kg, BID (3 mg/day in human)
HM15211 0.72 nmol/kg, Q2D (1 mg/wk in human)

* ~ ***p<0.05 ~ 0.001 *vs.* MCD mice, vehicle by One-way ANOVA, ††† p<0.001 *vs.* Liraglutide by One-way ANOVA

➢ HM15211 not only reduced blood ALT and bilirubin, but also reduced hepatic inflammation and HSC activation related marker expression, suggesting the anti-inflammatory effects of HM15211

Figure 3. Effect of HM15211 on NASH in MCD-diet mice (n=7)

(a) NAS **(b) H&E staining**

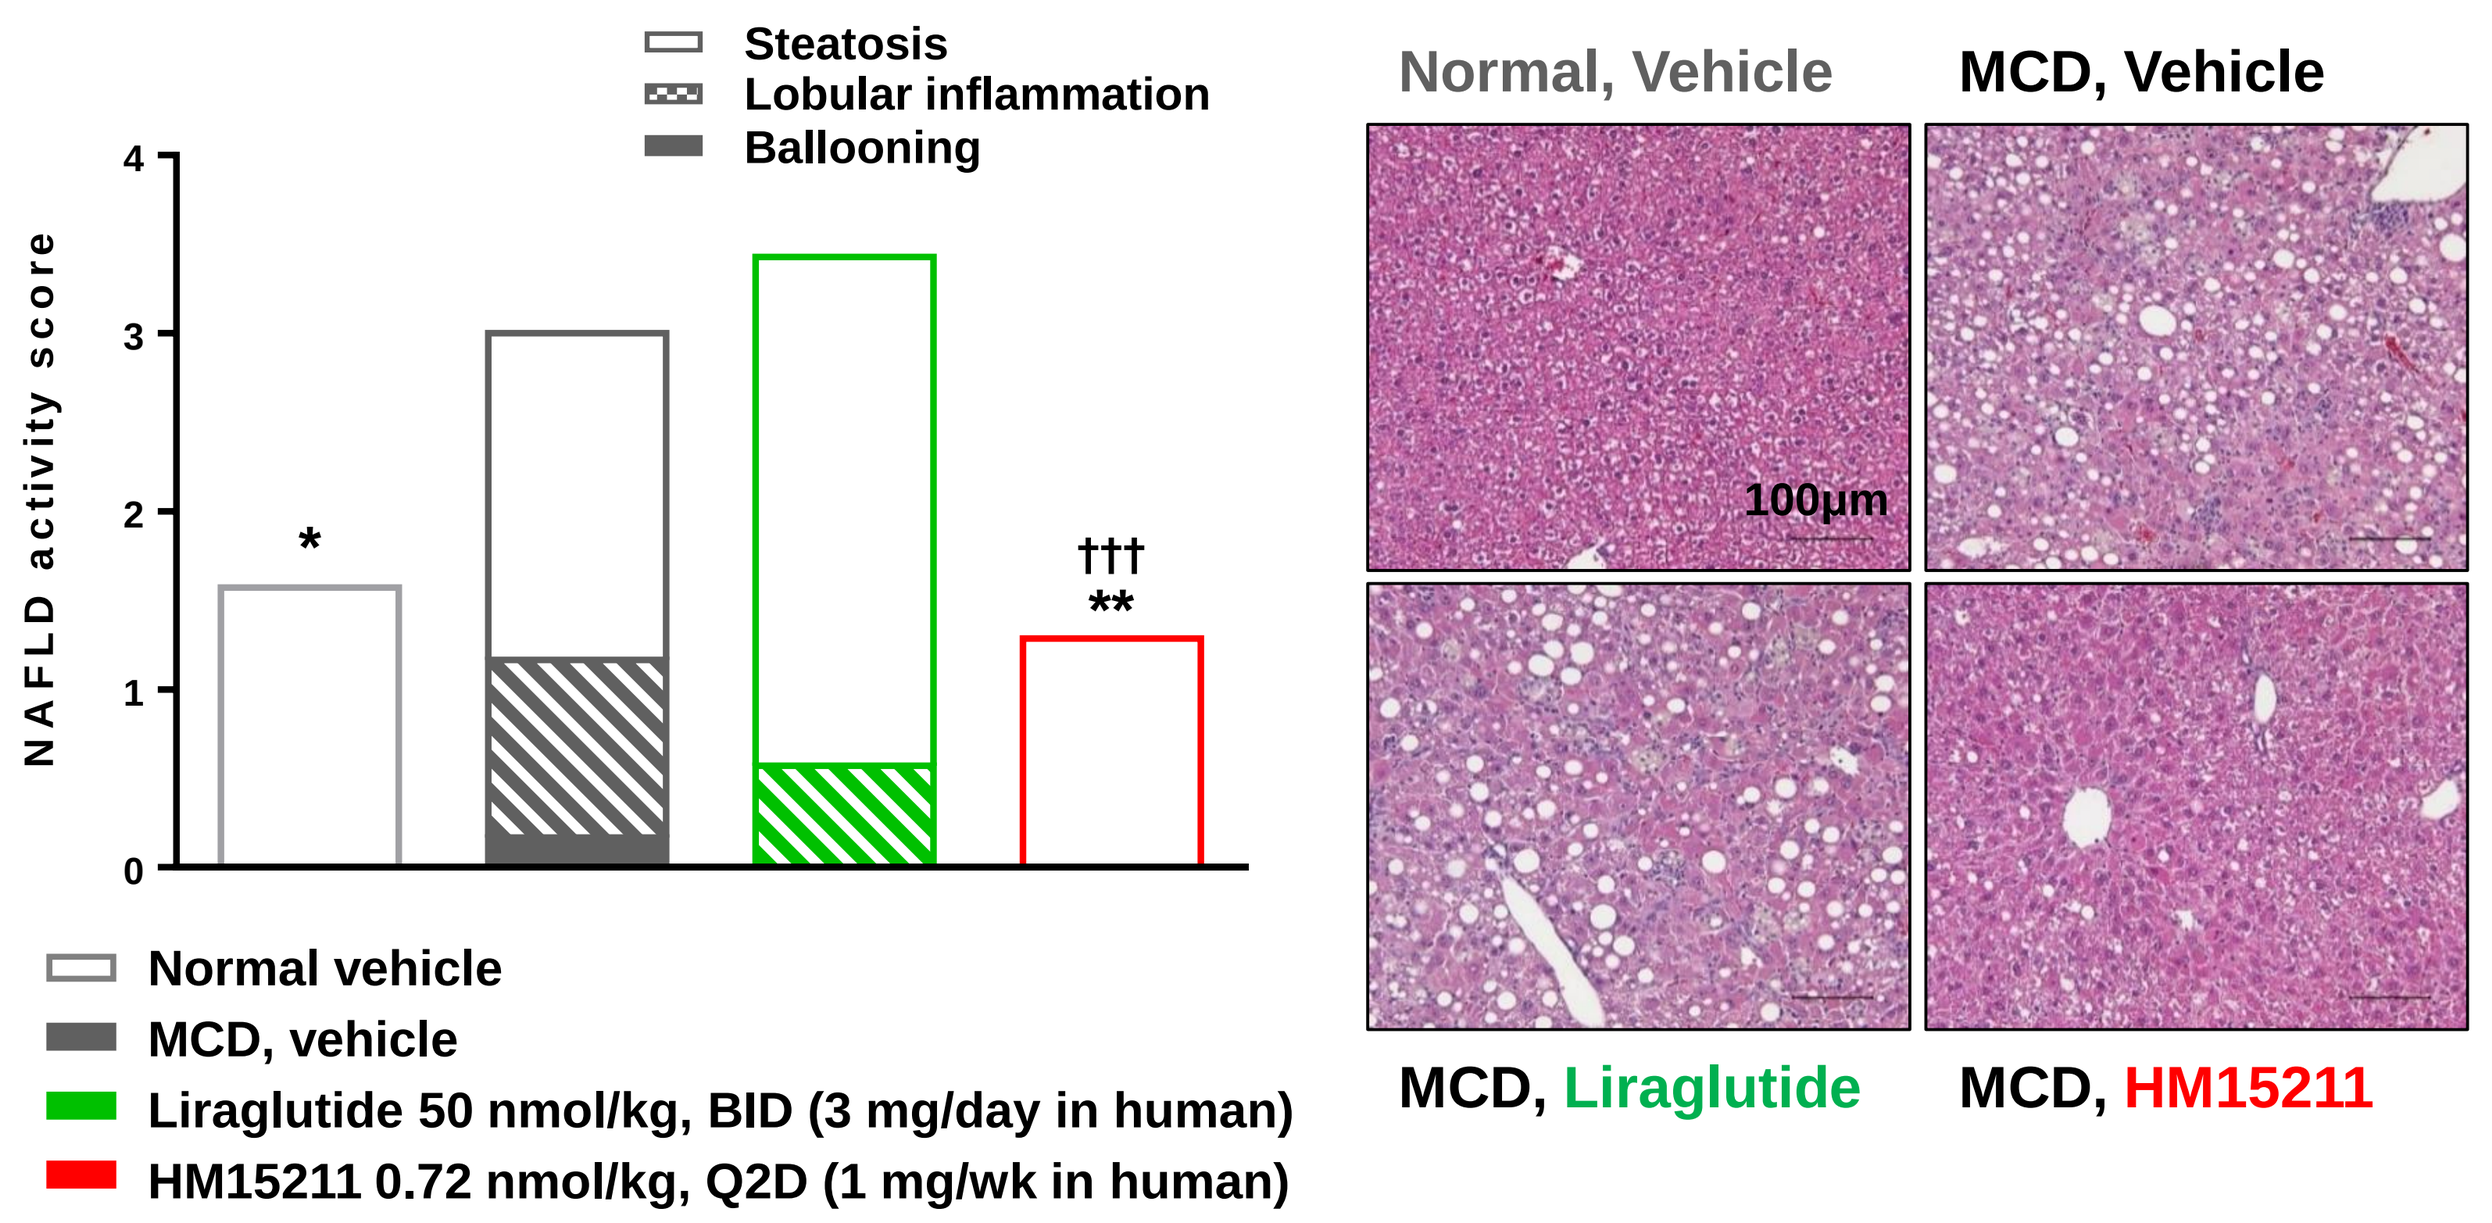
Steatosis
Lobular inflammation
Ballooning
NAFLD activity score
Normal vehicle
MCD, vehicle
Liraglutide 50 nmol/kg, BID (3 mg/day in human)
HM15211 0.72 nmol/kg, Q2D (1 mg/wk in human)
Normal, Vehicle
MCD, Vehicle
100μm
MCD, Liraglutide
MCD, HM15211

* ~ **p<0.05 ~ 0.01 *vs.* MCD mice, vehicle by One-way ANOVA; †††p<0.001 *vs.* Liragluitide by One-way ANOVA

➢ Consistently, HM15211 completely reversed NAS to normal level

Fibrosis improvement in MCD mice

Figure 4. Effect of HM15211 on hepatic fibrosis in MCD-diet mice (n=7)

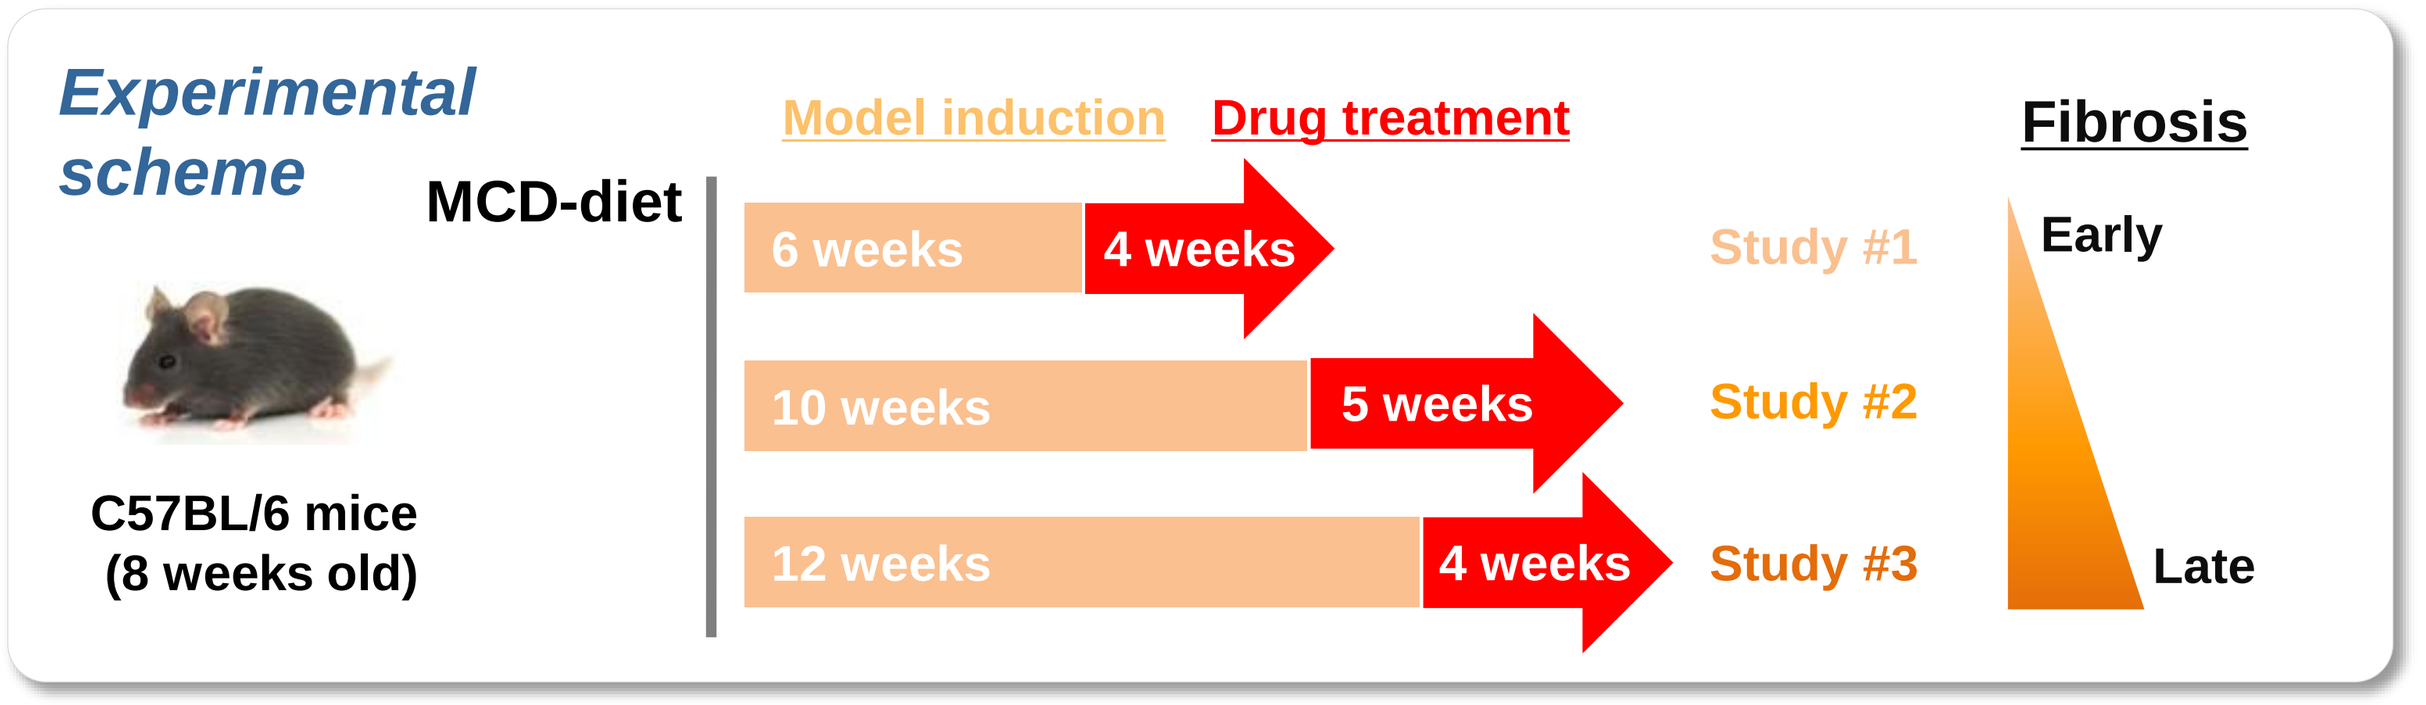
Experimental scheme
MCD-diet
C57BL/6 mice
(8 weeks old)
Model induction
Drug treatment
6 weeks
4 weeks
Study #1
10 weeks
5 weeks
Study #2
12 weeks
4 weeks
Study #3
Fibrosis
Early
Late

(a) Hepatic collagen-1α1 expression

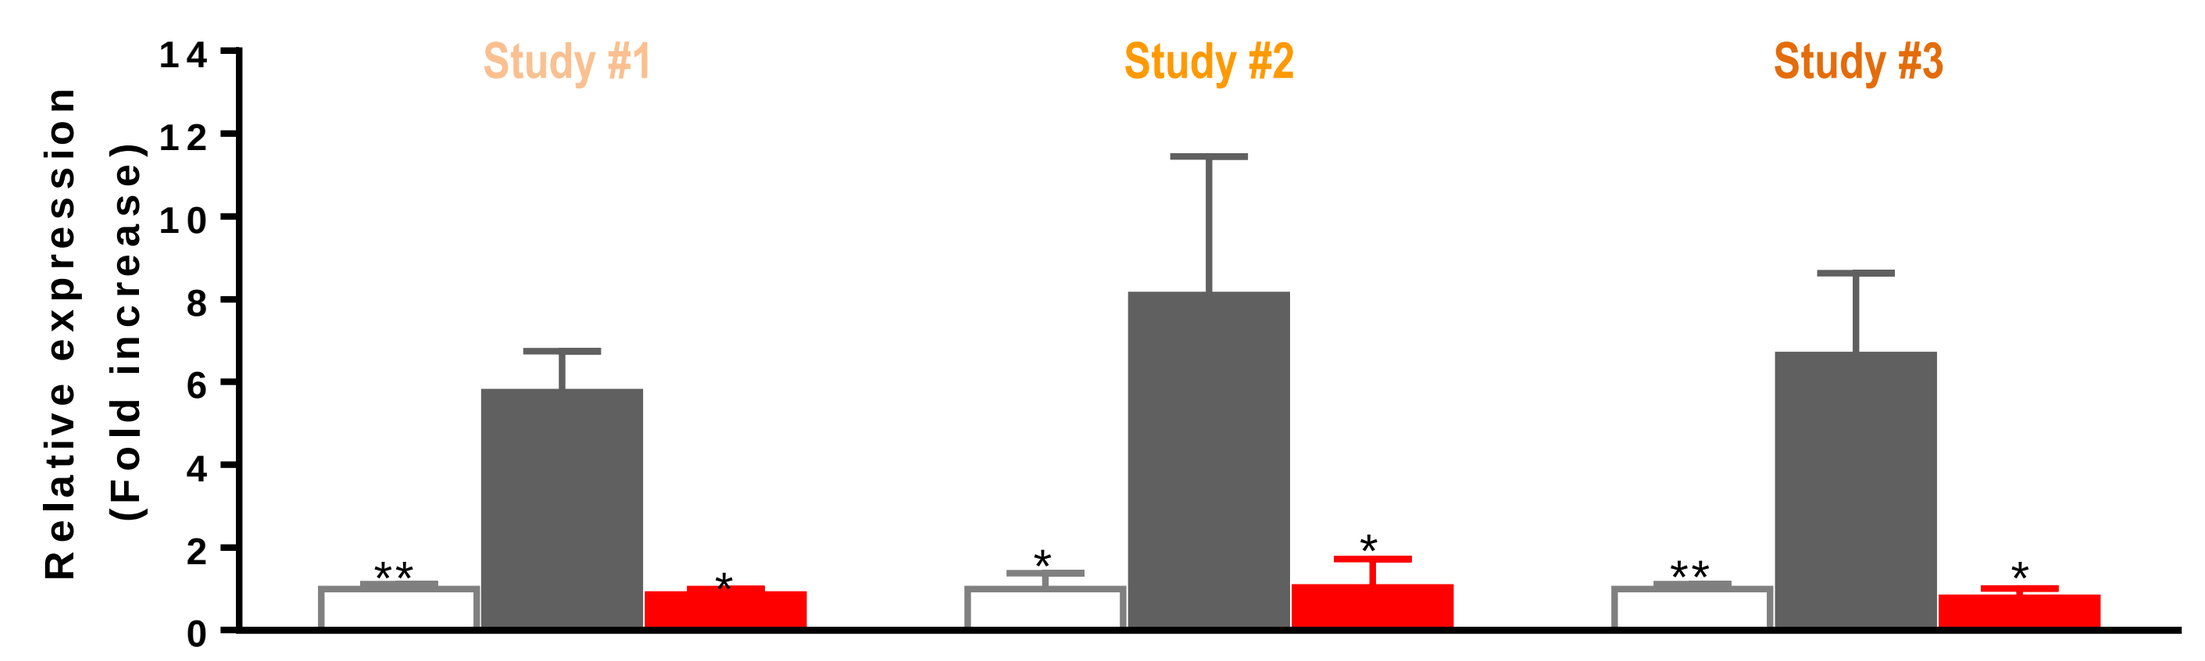
Relative expression (Fold increase)
Study #1
Study #2
Study #3

(b) Hepatic hydroxyproline and fibrosis score

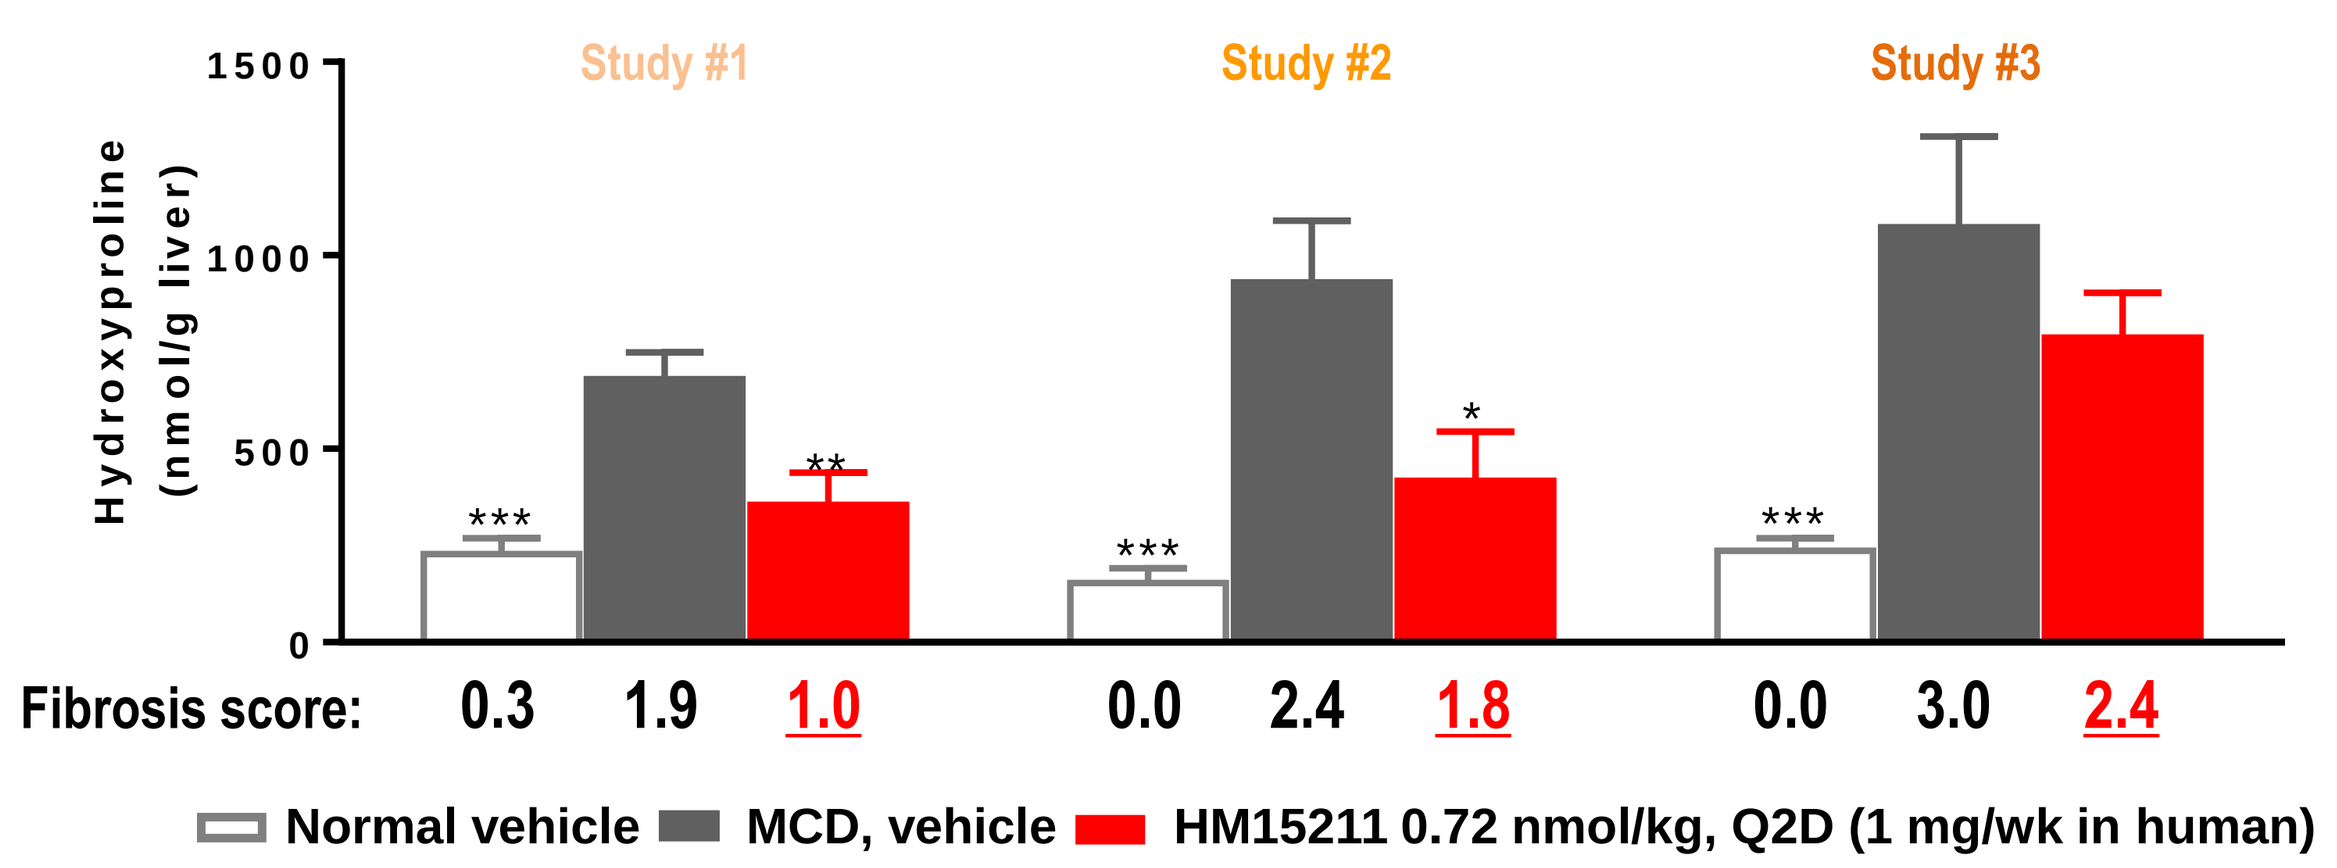
Hydroxyproline (nmol/g liver)
Study #1
Study #2
Study #3
Fibrosis score: 0.3 1.9 1.0 0.0 2.4 1.8 0.0 3.0 2.4
Normal vehicle
MCD, vehicle
HM15211 0.72 nmol/kg, Q2D (1 mg/wk in human)

* ~ ***p<0.0 5 ~ 0.001 *vs.* MCD mice, vehicle by One-way ANOVA

➢ HM15211 reduced hepatic expression of collagen-1α1, hydroxyproline contents, and fibrosis score in MCD-diet mice regardless of fibrosis stage

Therapeutic efficacy in obese/NASH monkeys

Figure 5. Effect of HM15211 on body composition and blood lipid profiles in obese/NASH monkeys

(a) DEXA image

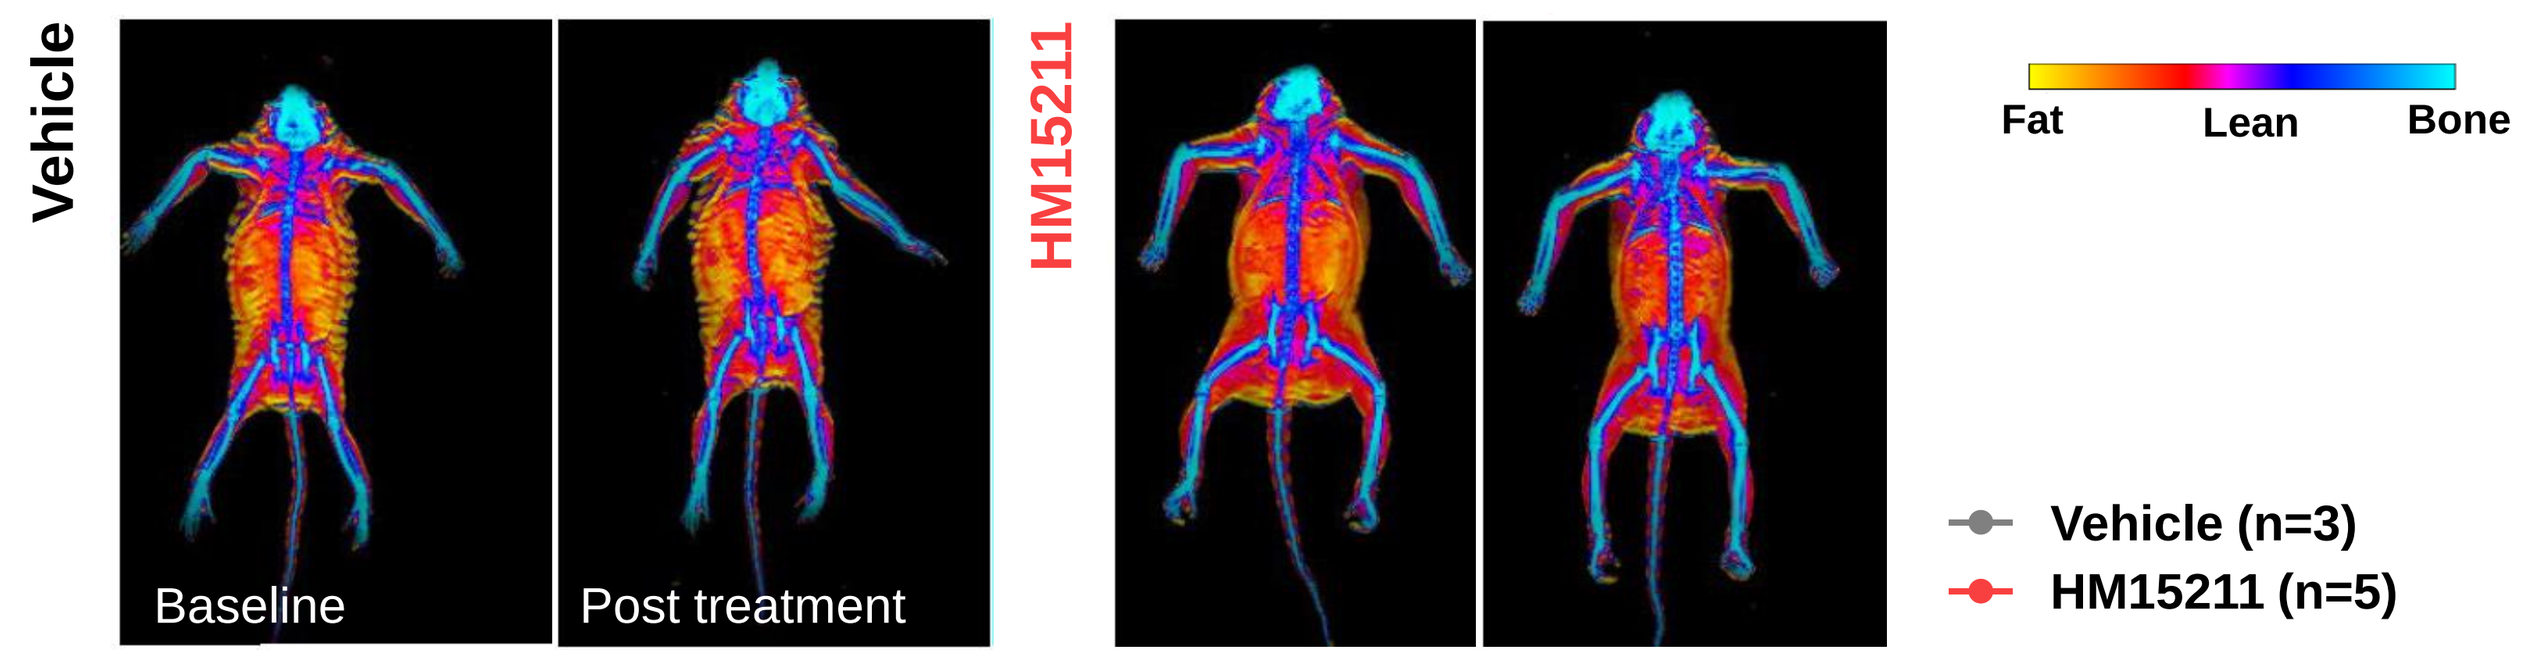
Vehicle
HM15211
Baseline
Post treatment
Fat
Lean
Bone
Vehicle (n=3)
HM15211 (n=5)

(b) Changes in blood lipid profiles

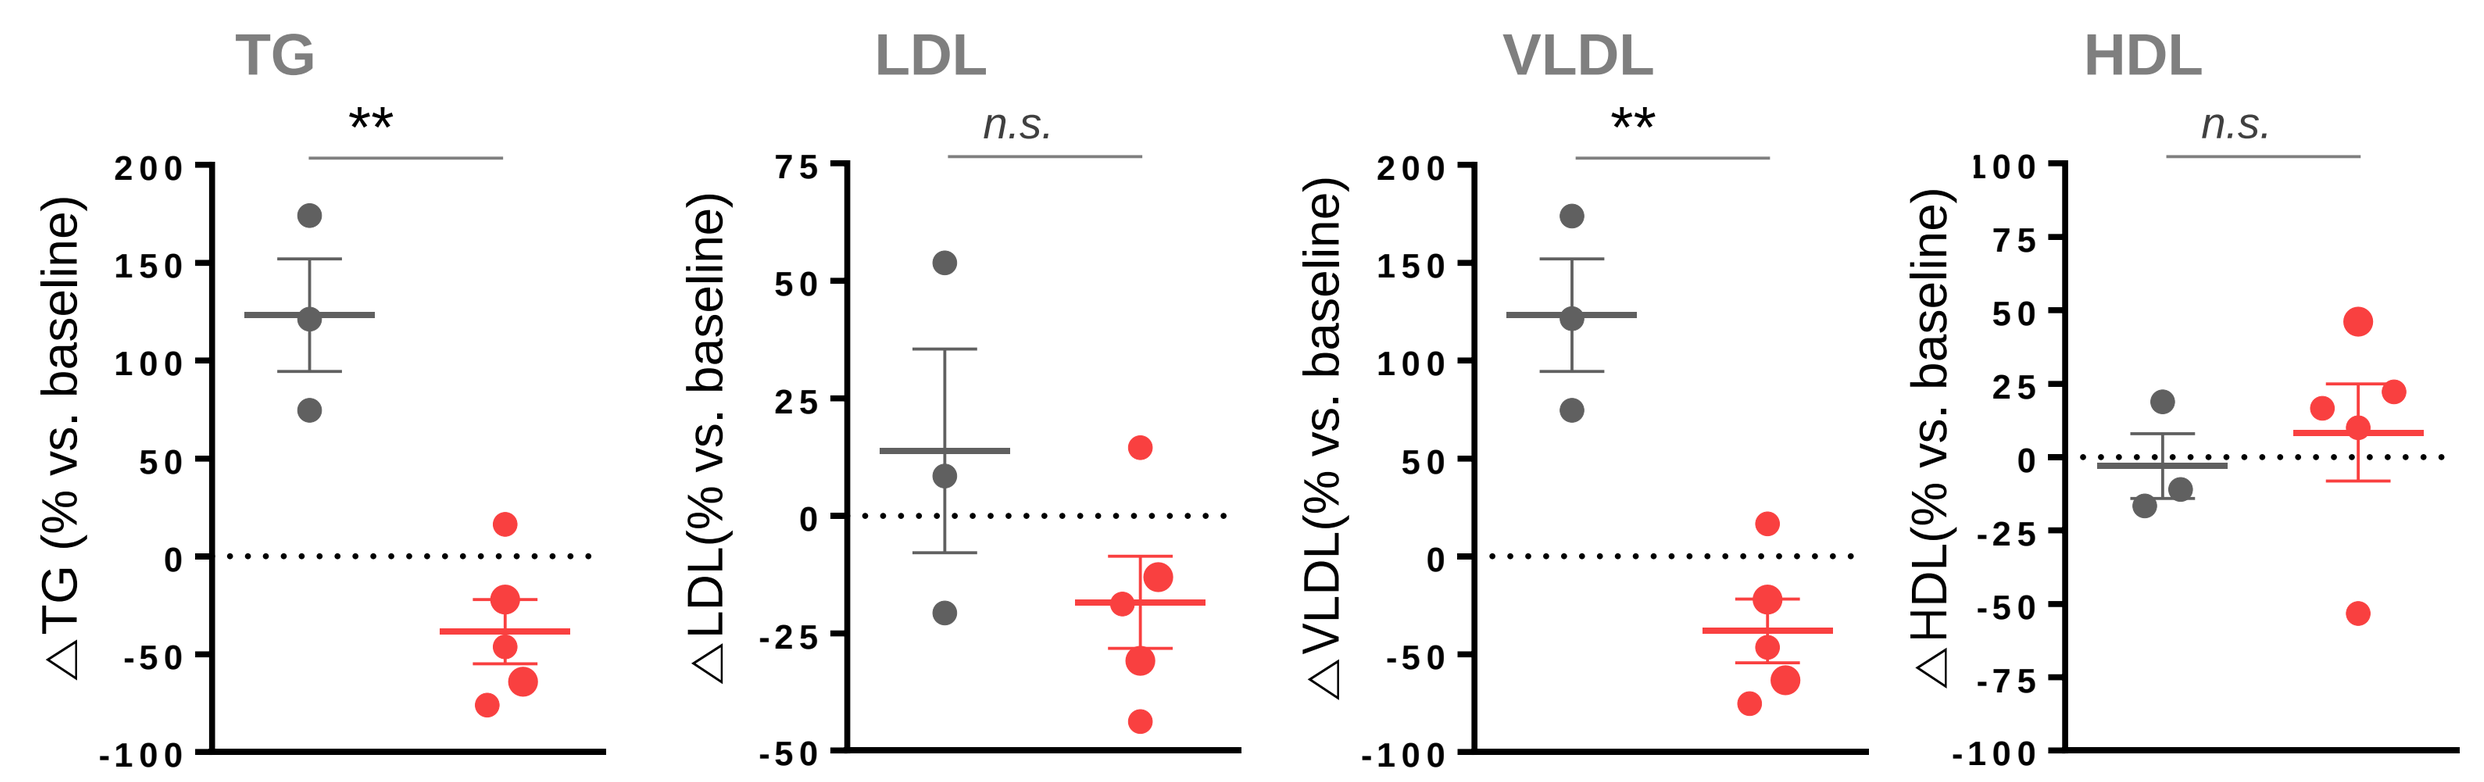
TG
LDL
VLDL
HDL
n.s.
△TG (% vs. baseline)
△LDL(% vs. baseline)
△VLDL(% vs. baseline)
△HDL(% vs. baseline)

➢ In obese/NASH NHP, HM15211 reduced fat mass, and improved blood lipid profiles

Figure 6. Effect of HM15211 in obese/NASH monkeys

(a) NAS **(b) H&E staining**

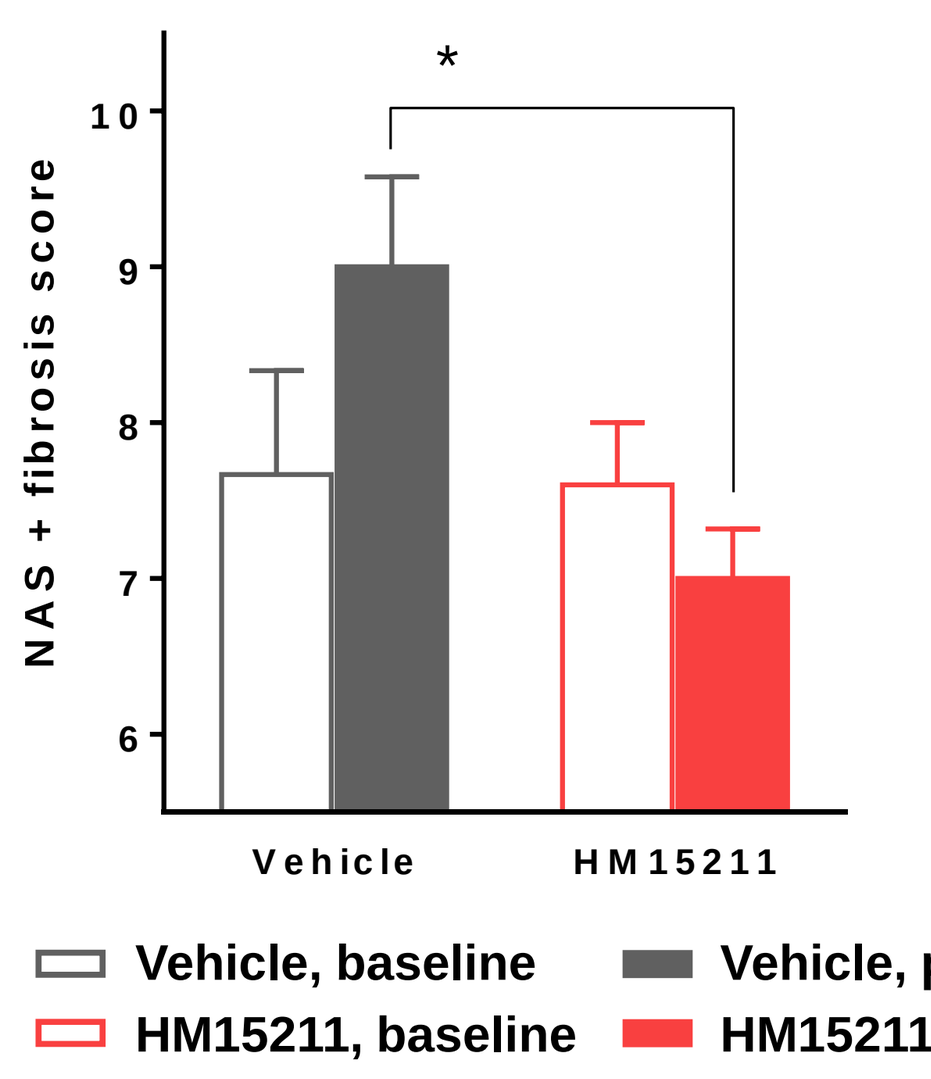
NAS + fibrosis score
Vehicle
HM15211
Vehicle, baseline
Vehicle, post-treatment (n=3)
HM15211, baseline
HM15211, post-treatment (n=5)

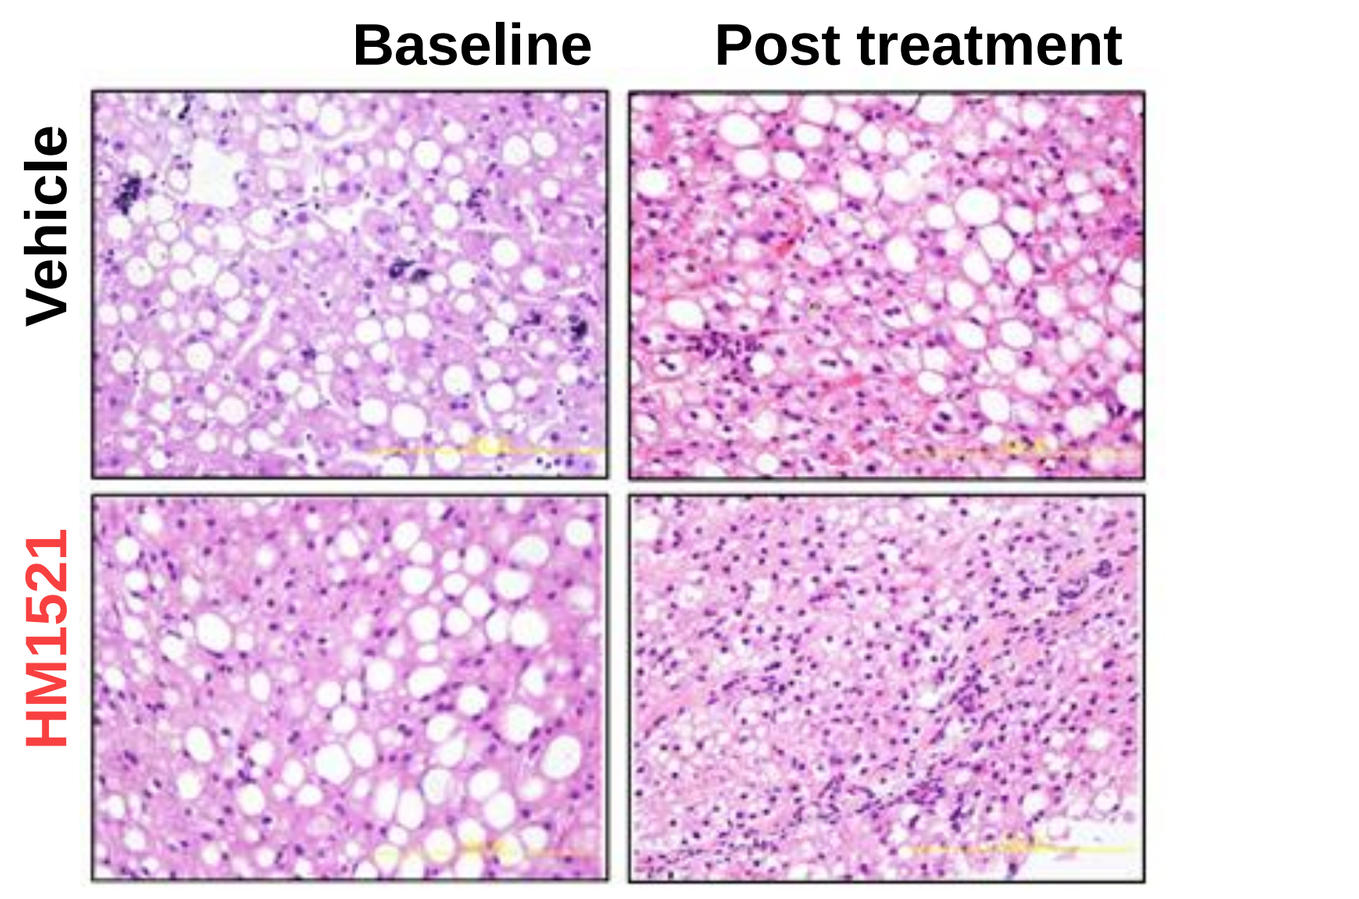
Baseline
Post treatment
Vehicle
HM1521

* ~ **p<0.0 5 ~0.01 *vs.* vehicle by un-paired t-test

➢ Relatively short-term treatment of HM15211 led to meaningful improvement in NAS + fibrosis score (*vs.* vehicle) in obese/NASH NHP

CONCLUSIONS

- **HM15211, a novel long-acting triple agonist, is designed to treat NASH and fibrosis by aiming multiple pathways involved in NASH and fibrosis progression**
- **In rodent NASH models, HM15211 reduces liver fat, inflammation marker expression, leading to NASH resolution**
- **In addition, HM15211 has potential to improve fibrosis in rodent NASH and fibrosis models regardless of fibrosis stage**
- **Beneficial effects of HM15211 on NASH and fibrosis improvement are well-translated in obese/NASH NHP**
- **Therefore, HM15211 might be a novel therapeutic option for NASH and fibrosis**

European Association for the Study of Diabetes (EASD) 55th Annual Meeting, Barcelona, Spain, 16 - 20 September 2019

Hanmi Pharm. Co., Ltd.