A double-blind, placebo-controlled single ascending dose study to evaluate the safety, tolerability, pharmacokinetics, and pharmacodynamics of HM15136, a novel long-acting glucagon analogue, in healthy subjects

Hanmi

KY Huh*,[1], W Shin*,[2], E Baek[2], S Seo[2], W Lee[2], S Baek[2], H Lee[1,3]

[1] Department of Clinical Pharmacology and Therapeutics, Seoul National University College of Medicine and Hospital, Seoul, Korea. [2] Hanmi Pharm. Co., Ltd, Seoul, Korea. [3] Department of Molecular Medicine and Biopharmaceutical Sciences, Graduate School of Convergence Science and Technology, Seoul National University, Seoul, Korea.

ABSTRACT

HM15136 is a novel long-acting glucagon analogue with an extended half-life. In vivo efficacy studies of HM15136 in animal models showed its therapeutic potential in congenital hyperinsulinism, hypoglycaemia post-bariatric surgery, and obesity. We performed a randomized, double-blind, placebo-controlled first-in-human trial to assess the safety, pharmacokinetics, and pharmacodynamics of a single subcutaneous dose of HM15136 in healthy adults. Fifty-six subjects randomly received HM15136 or its matching placebo in a ratio of 6:2 in 7 cohorts (10, 20, 30, 50, 80, 100, and 120 μg/kg). All adverse events were transient and mild in severity. Neither serious adverse event nor discontinuation due to adverse event occurred during the study. The most frequent Treatment Emergent Adverse Event was nausea (7.2%, only in the 100 and 120 μg/kg groups). Injection site erythema, which was barely noticeable and spontaneously subsided without sequelae, was reported in 2 out of 42 subjects who received HM15136. HM15136 was slowly, but steadily absorbed with the peak serum concentration reaching 34.2-65.2 hours after dose. The terminal half-life ranged from 69.8 to 85.6 hours, which was significantly greater than that of glucagon (< 1 hour). HM15136, particularly at doses >50 μg/kg, increased mean serum glucose from baseline by up to 27.7 mg/dL, which was maintained until 17 days after dose. In conclusion, a single subcutaneous dose of HM15136 at 10-120 μg/kg was safe and well-tolerated. The long half-life of HM15136, coupled with increase in serum glucose for ~2 weeks, may warrant a weekly or longer dosing regimen.

BACKGROUND

HM15136 is a glucagon analogue, conjugated with a human IgG Fc fragment *via* a flexible PEG linker

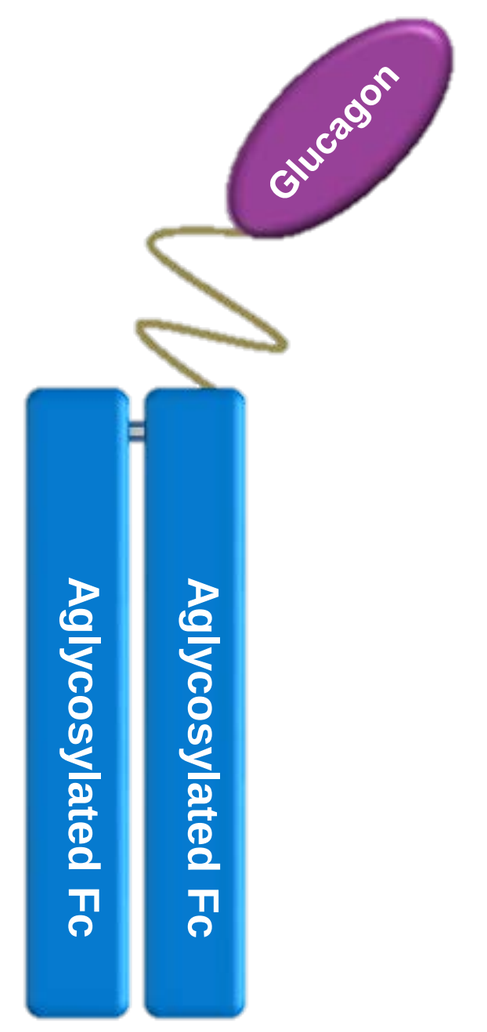
Glucagon
Aglycosylated Fc
Aglycosylated Fc

[General Profile of HM15136]

- **Although glucagon is considered as one of the most potent therapeutic options, its utilization is limited due to poor solubility, limited stability at physiological pH and short duration of action.**
- **To overcome these limitations, HM15136, a novel long-acting glucagon analogue with an extended half-life, is being developed.**
- **Stable and highly soluble weekly glucagon analog**
- **Sufficient PK profile for once a week administration**
- ***In vivo* efficacy studies of HM15136 in animal models showed its therapeutic potential in congenital hyperinsulinism, hypoglycaemia post-bariatric surgery, and obesity.**

STUDY DESIGN

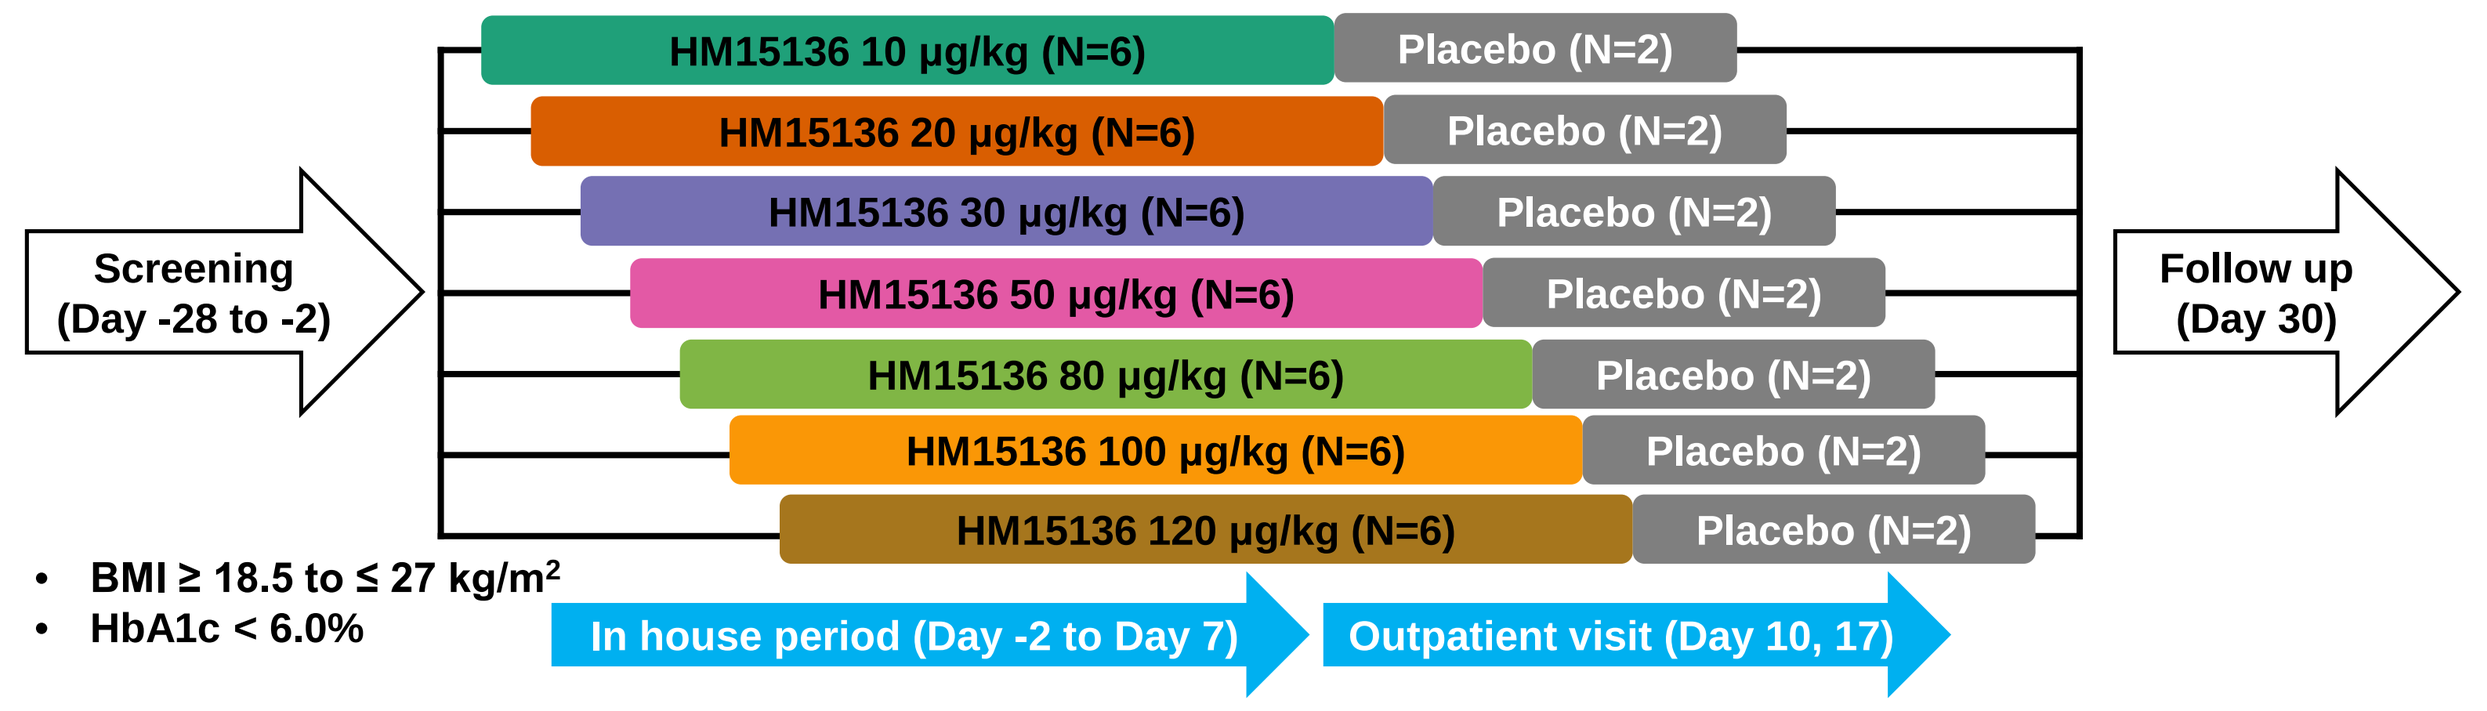
Screening
(Day -28 to -2)
HM15136 10 μg/kg (N=6)
Placebo (N=2)
HM15136 20 μg/kg (N=6)
Placebo (N=2)
HM15136 30 μg/kg (N=6)
Placebo (N=2)
HM15136 50 μg/kg (N=6)
Placebo (N=2)
HM15136 80 μg/kg (N=6)
Placebo (N=2)
HM15136 100 μg/kg (N=6)
Placebo (N=2)
HM15136 120 μg/kg (N=6)
Placebo (N=2)
Follow up
(Day 30)
BMI ≥ 18.5 to ≤ 27 kg/m²
HbA1c < 6.0%
In house period (Day -2 to Day 7)
Outpatient visit (Day 10, 17)

- **Primary objective:** To assess safety and tolerability of HM15136 after single subcutaneous (sc) doses
- **Secondary objective:** To assess pharmacokinetic profile of HM15136 after single sc doses
- **Exploratory objective:** To assess pharmacodynamics properties of HM15136 after single sc doses

RESULTS

- **Demographics and baseline characteristics**

	HM15136 dose (μg/kg)							
	10 (N=6)	20 (N=6)	30 (N=6)	50 (N=6)	80 (N=6)	100 (N=6)	120 (N=6)	Placebo (N=14)
Male sex	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	14 (100.0)
Age (years)	31.7 ± 5.8	35.2 ± 6.5	31.2 ± 5.3	27.8 ± 2.7	32.3 ± 6.3	32.2 ± 12.4	26.2 ± 7.9	31.4 ± 6.9
Weight (kg)	67.4 ± 7.1	65.7 ± 7.2	69.7 ± 7.5	75.2 ± 7.9	71.1 ± 9.0	69.3 ± 11.1	63.7 ± 6.4	70.7 ± 6.3
Height (cm)	172.4 ± 4.5	173.0 ± 4.0	172.2 ± 3.0	174.0 ± 3.7	172.6 ± 7.2	172.6 ± 6.8	172.0 ± 6.4	174.3 ± 5.6
BMI (kg/m²)	22.6 ± 1.9	21.9 ± 2.5	23.5 ± 2.3	24.8 ± 1.6	23.8 ± 1.7	23.2 ± 3.0	21.6 ± 3.1	23.3 ± 2.0
HbA1c (%)	5.2 ± 0.3	5.2 ± 0.3	5.3 ± 0.3	5.2 ± 0.2	5.3 ± 0.2	5.2 ± 0.4	5.2 ± 0.3	5.2 ± 0.3

* Mean ± standard deviation was presented except for male sex, where number of subjects (percentage of subjects) was presented.

- **Safety and tolerability**

a. All adverse events were mild in severity and transient. Neither serious adverse event nor discontinuation due to adverse event occurred during the study. The most frequent Treatment Emergent Adverse Event was nausea (7.2%, only in the 100 and 120 μg/kg groups).

Type of TEAEs, n (%)	HM15136 dose (μg/kg)							
	10 (N=6)	20 (N=6)	30 (N=6)	50 (N=6)	80 (N=6)	100 (N=6)	120 (N=6)	Placebo (N=14)
Any TEAE	1 (16.7)	3 (50.0)	4 (66.7)	1 (16.7)	3 (50.0)	3 (50.0)	5 (83.3)	3 (21.4)
Maximum Severity of TEAE								
Mild	1 (16.7)	3 (50.0)	4 (66.7)	1 (16.7)	3 (50.0)	3 (50.0)	5 (83.3)	3 (21.4)
Moderate	0	0	0	0	0	0	0	0
Severe	0	0	0	0	0	0	0	0
Any TEAE related to IMP	0	2 (33.3)	2 (33.3)	1 (16.7)	1 (16.7)	2 (33.3)	4 (66.7)	1 (7.1)
Maximum Severity of TEAE related to IMP								
Mild	0	2 (33.3)	2 (33.3)	1 (16.7)	1 (16.7)	2 (33.3)	4 (66.7)	1 (7.1)
Moderate	0	0	0	0	0	0	0	0
Severe	0	0	0	0	0	0	0	0
Any TEAE resulting in death	0	0	0	0	0	0	0	0
Any TEAE leading to withdrawal	0	0	0	0	0	0	0	0
Any serious TEAE	0	0	0	0	0	0	0	0

b. Injection site erythema, which was barely noticeable and spontaneously subsided without sequelae, was reported in 2 out of 42 subjects who received HM15136.

c. Overall, no anti-drug antibodies and anti-polyethylene glycol antibodies were detected in the study. There were no subjects had confirmed antibodies, so neutralizing antibodies were not analyzed.

Time point	Tier	Category, n (%)	HM15136 dose (μg/kg)							
			10 (N=6)	20 (N=6)	30 (N=6)	50 (N=6)	80 (N=6)	100 (N=6)	120 (N=6)	Placebo (N=14)
Baseline	Screen	Negative	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	13 (92.9)
		Positive	0	0	0	0	0	0	0	1 (7.1)
	Confirm	N/A	6 (100.0)	6 (100.0)	6 (100.0)	5 (83.3)	6 (100.0)	6 (100.0)	6 (100.0)	13 (92.9)
		Negative	0	0	0	1 (16.7)	0	0	0	1 (7.1)
Day 17	Screen	Negative	5 (83.3)	6 (100.0)	5 (83.3)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	13 (92.9)
		Positive	1 (16.7)	0	1 (16.7)	0	0	0	0	1 (7.1)
	Confirm	N/A	5 (83.3)	6 (100.0)	5 (83.3)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	13 (92.9)
		Negative	1 (16.7)	0	1 (16.7)	0	0	0	0	1 (7.1)
Day 30	Screen	Negative	5 (83.3)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	5 (83.3)	14 (100.0)
		Positive	1 (16.7)	0	0	0	0	0	0	0
	Confirm	N/A	5 (83.3)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	5 (83.3)	14 (100.0)
		Negative	1 (16.7)	0	0	0	0	0	0	0

* Number of subjects (percentage of subjects)

- **Pharmacokinetics**

a. Mean serum HM15136 concentration versus time

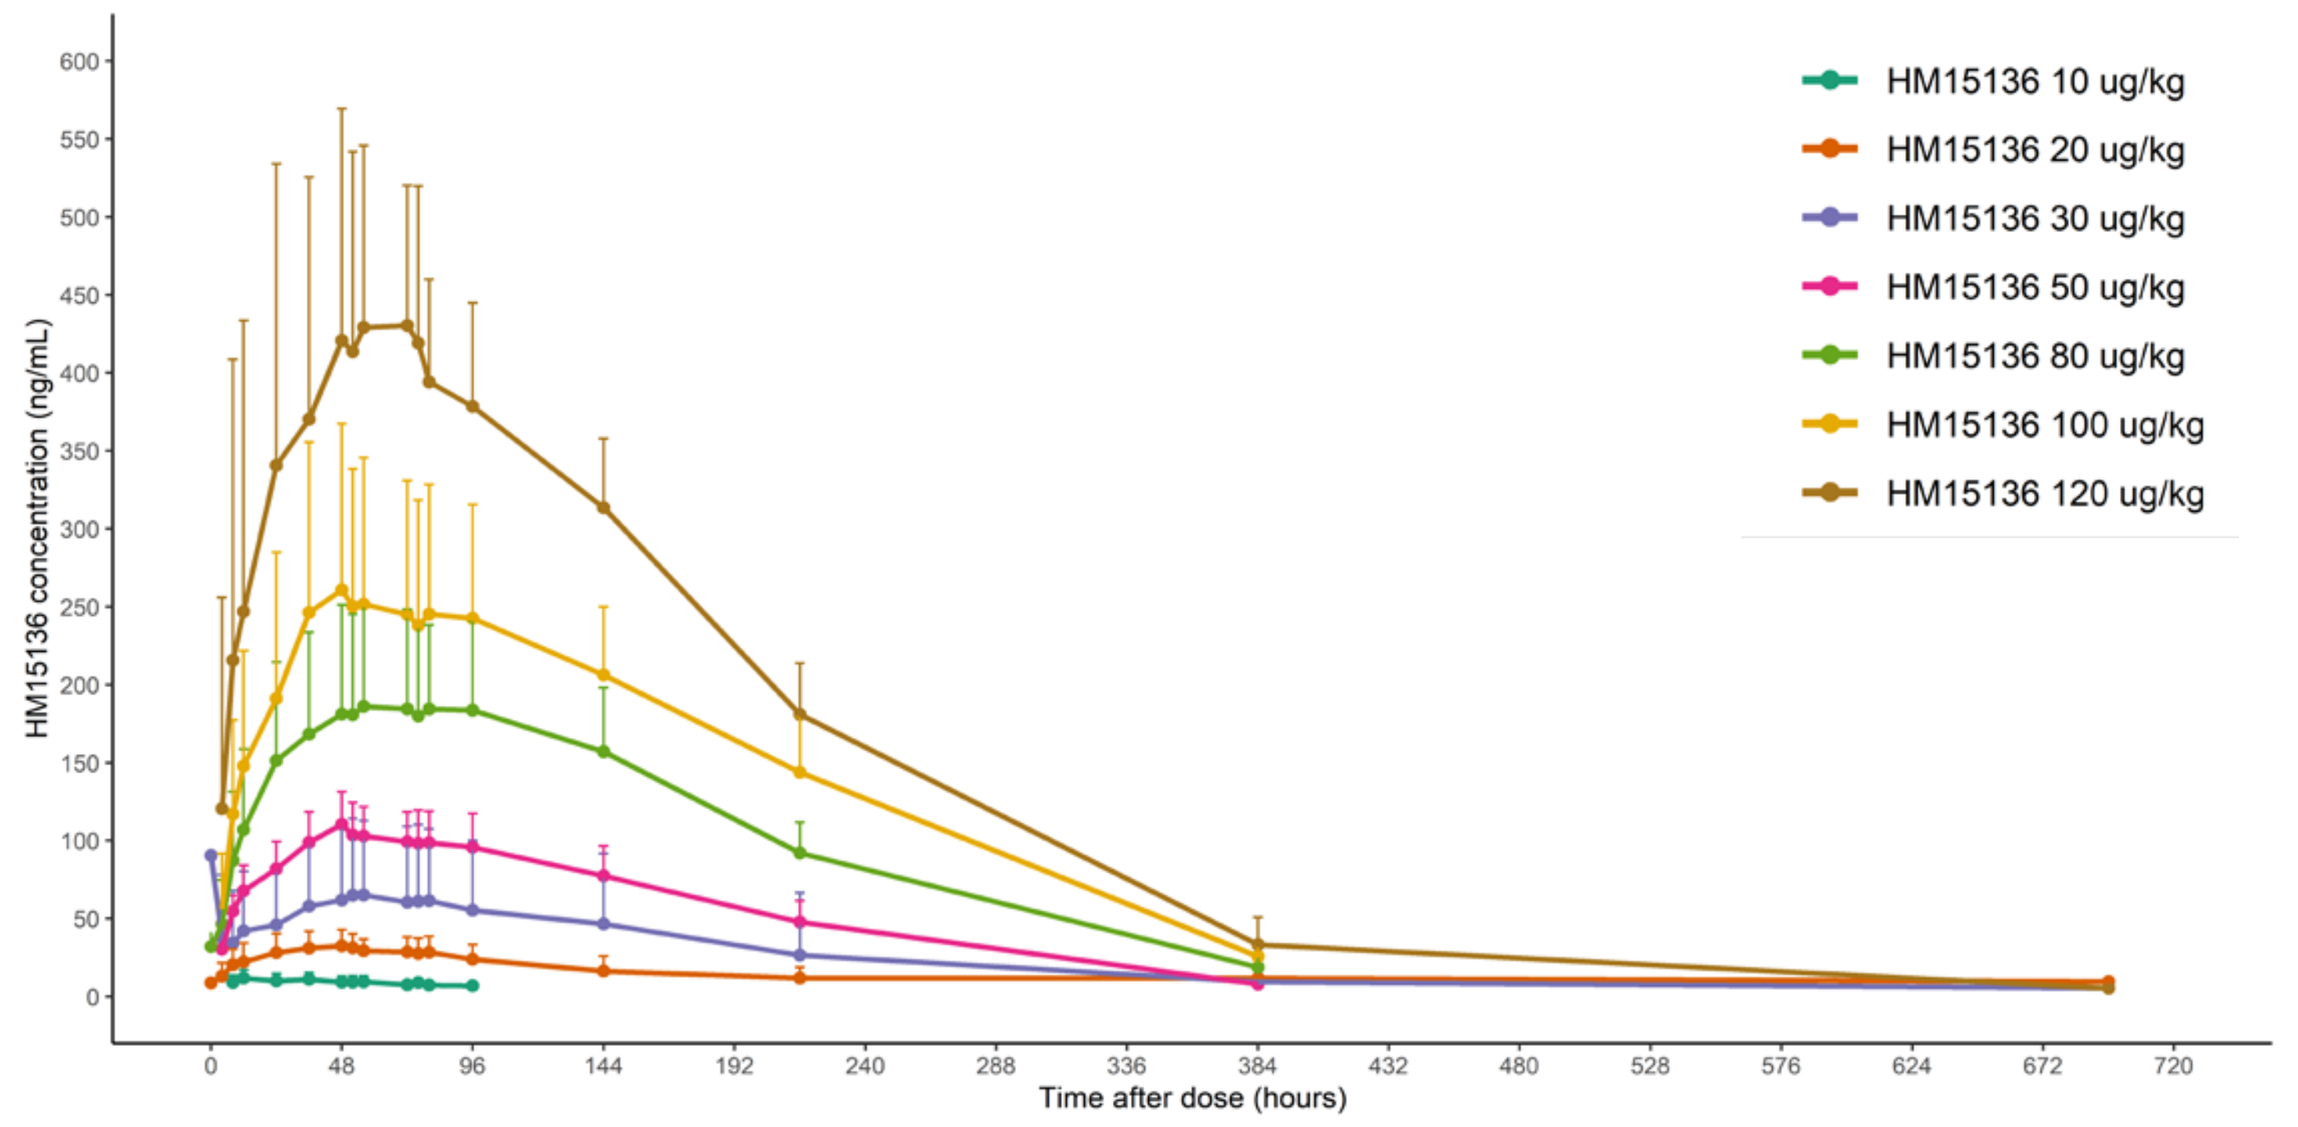
HM15136 concentration (ng/mL)
Time after dose (hours)
HM15136 10 ug/kg
HM15136 20 ug/kg
HM15136 30 ug/kg
HM15136 50 ug/kg
HM15136 80 ug/kg
HM15136 100 ug/kg
HM15136 120 ug/kg

* Error bars denoted standard deviation.

b. Summary PK parameters of HM15136

	HM15136 dose (μg/kg)						
	10 (N=4)	20 (N=6)	30 (N=6)	50 (N=6)	80 (N=6)	100 (N=6)	120 (N=6)
C_{max} (ng/mL)	9.9 ± 4.6	33.1 ± 9.8	66.9 ± 48.5	110.5 ± 20.8	199.2 ± 61.7	273.7 ± 93.9	469.5 ± 134.8
T_{max} (hr)	46.0 [12.1-56.0]	48.0 [36.0-56.0]	56.0 [35.6-80.0]	48.0 [48.0-48.0]	68.0 [48.0-80.0]	48.0 [36.0-96.0]	60.0 [24.0-80.0]
AUC_{0-t} (hr*ng/mL)	668.1 ± 399.2	5,068.4 ± 4,308.2	11,926.2 ± 12,860.7	19,575.9 ± 5,196.2	38,429.5 ± 11,281.1	53,516.0 ± 11,828.7	83,623.1 ± 11,367.1
AUC_{inf} (hr*ng/mL)	1,472.2 ± 159.6	6,499.6 ± 5,398.4	13,084.3 ± 12,749.7	22,146.8 ± 4,095.3	42,217.2 ± 11,155.6	56,625.9 ± 10,607.4	87,613.8 ± 10,135.5
$t_{1/2}$ (hr)	77.1 ± 41.0	101.1 ± 77.1	84.3 ± 26.2	84.0 ± 21.4	79.2 ± 24.6	79.7 ± 19.0	81.5 ± 16.5

* Mean ± standard deviation was presented except for T_{max}, where median [minimum-maximum] was presented.

- **Pharmacodynamics**

a. Mean change from baseline of fasting blood sugar level versus time

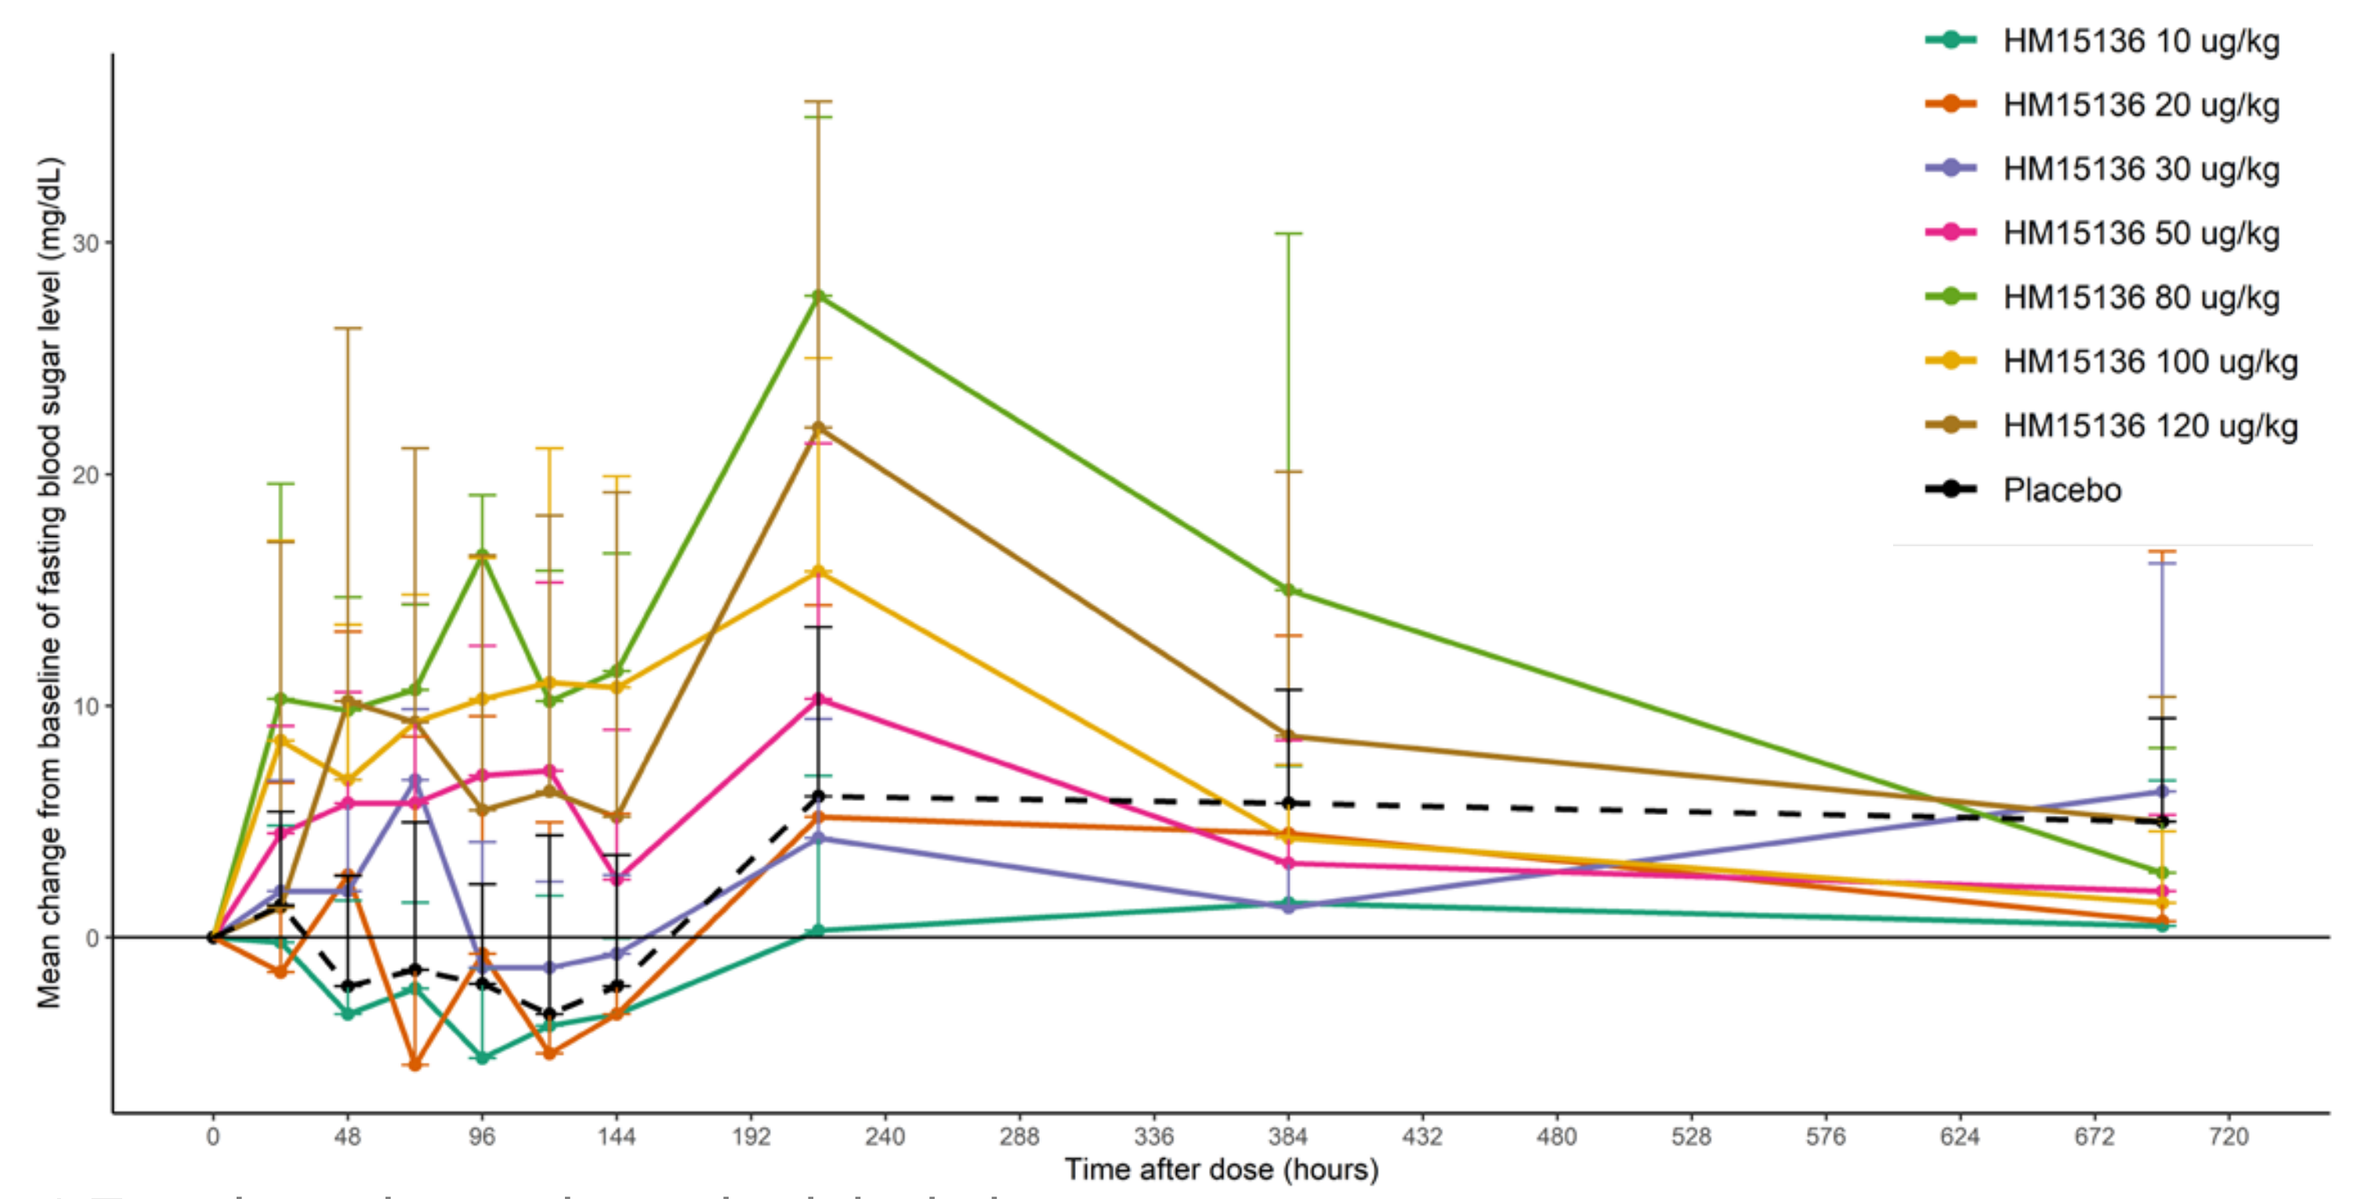
Mean change from baseline of fasting blood sugar level (mg/dL)
Time after dose (hours)
HM15136 10 ug/kg
HM15136 20 ug/kg
HM15136 30 ug/kg
HM15136 50 ug/kg
HM15136 80 ug/kg
HM15136 100 ug/kg
HM15136 120 ug/kg
Placebo

* Error bars denoted standard deviation.

CONCLUSIONS

A single subcutaneous dose of HM15136 at 10-120 μg/kg was safe and well-tolerated.

The long half-life of HM15136, coupled with increase in serum glucose, may warrant a weekly or longer dosing regimen.

American Diabetes Association's (ADA) 80th Scientific Sessions, Virtual Session; June 12-16, 2020.

Hanmi Pharm. Co., Ltd.